XTX301, A Protein-engineered IL-12, Exhibits Tumor-selective Activity In Mice Without Peripheral Toxicities And Is Well Tolerated In Non-Human Primates

Xilio Therapeutics
Waltham, MA, USA

Ekta Patel, Natalia Malkova, Sallyan Vu, Rebekah O'Donnell, Manoussa Fanny, Wilson Guzman, Parker Johnson, Megan McLaughlin, Oleg Yerov, Kurt Jenkins, Hanumantha Rao Madala, Caitlin O'Toole, Magali Pederzoli-Ribeil, Jia Chen, Benjamin Nicholson, Huawei Qiu, Jennifer O'Neil

Introduction

Interleukin-12 (IL-12) is a proinflammatory cytokine which bridges innate and adaptive immunity via induction of T helper 1 differentiation, promoting cytolytic activity of natural killer and T cells. IL-12 has demonstrated potent anti-tumor activity in syngeneic mouse models and promising anti-tumor efficacy in humans. However, clinical development of IL-12 has been limited by severe systemic toxicities. To overcome toxicity and improve the therapeutic index of IL-12, we employed our geographically precise solutions, or GPS, platform to develop XTX301, to achieve systemic delivery of a tumor-selective half-life extended (HLE) and masked IL-12.

Design and predicted activity of XTX301

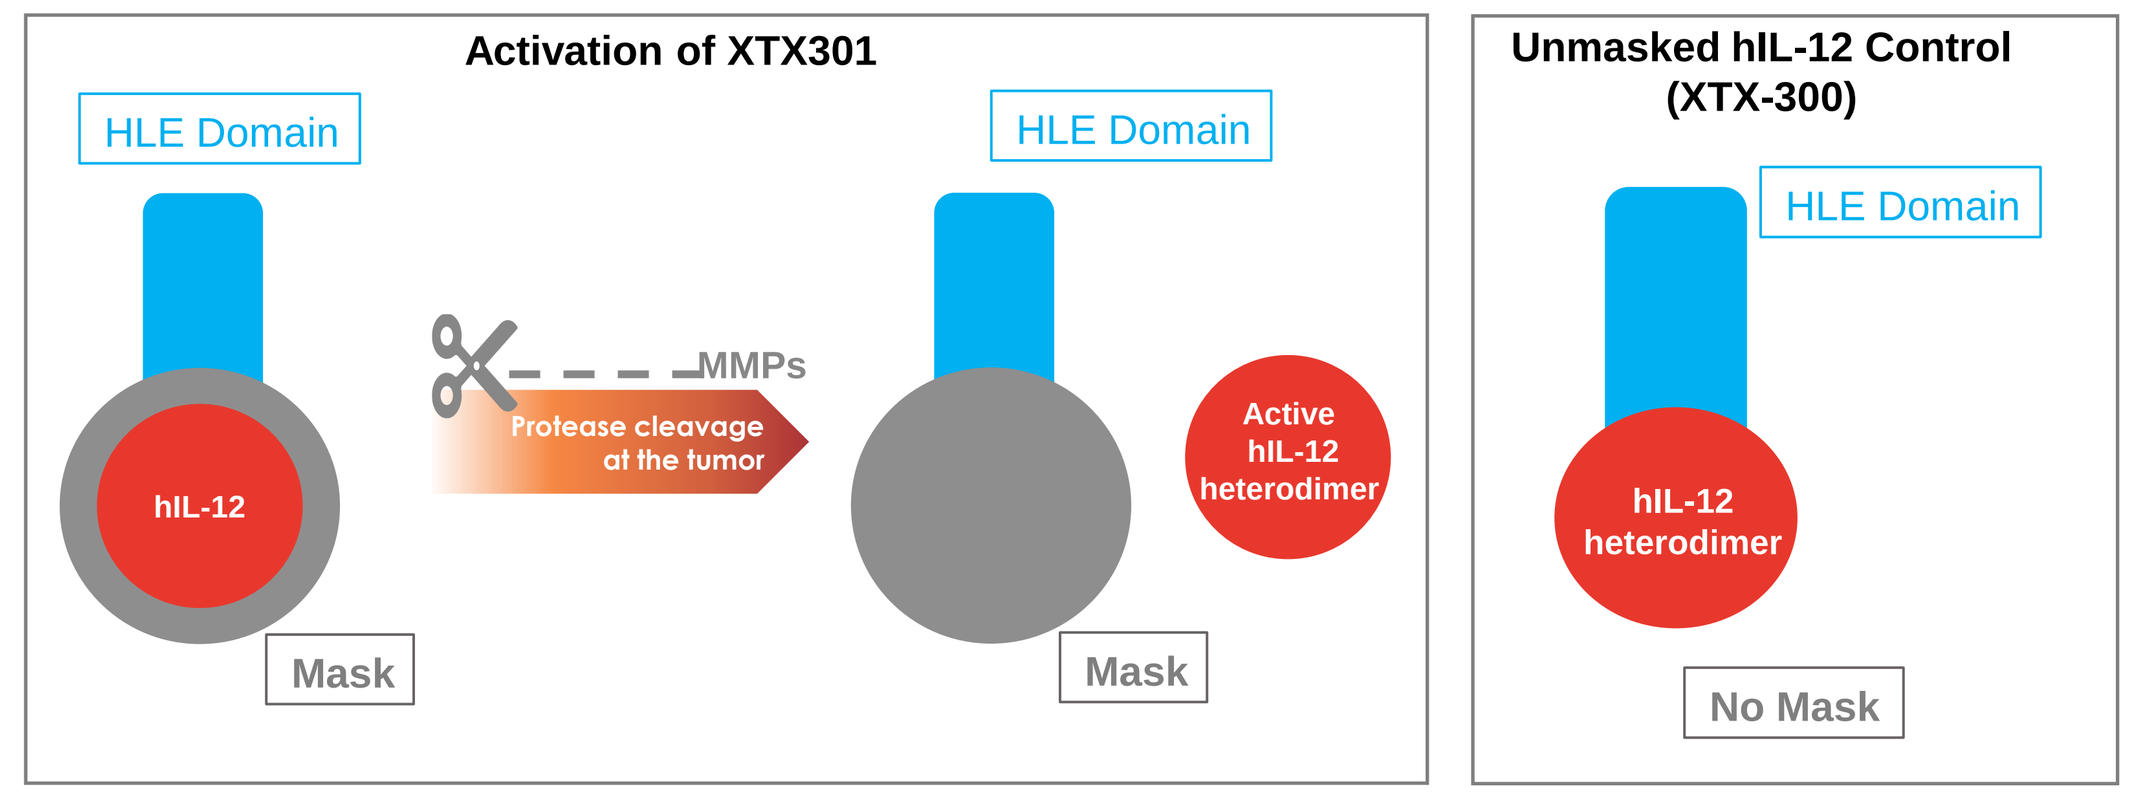

Activation of XTX301
HLE Domain
hIL-12
Mask
MMPs
Protease cleavage at the tumor
HLE Domain
Active hIL-12 heterodimer
Mask
Unmasked hIL-12 Control (XTX-300)
HLE Domain
hIL-12 heterodimer
No Mask

Figure 1: The masking domain of XTX301 is designed to pharmacologically inactivate IL-12 systemically and render an active IL-12 moiety upon cleavage by proteases that are enriched in the tumor microenvironment. XTX300 was designed to serve as an unmasked control molecule. Human IL-12 does not cross react with mouse IL-12 receptors; hence a murine surrogate (mXTX301) was created for in vivo anti-tumor efficacy evaluation.

Masking and recovery of *in vitro* activity with XTX301

XTX301 binds to IL12RB2 when proteolytically cleaved

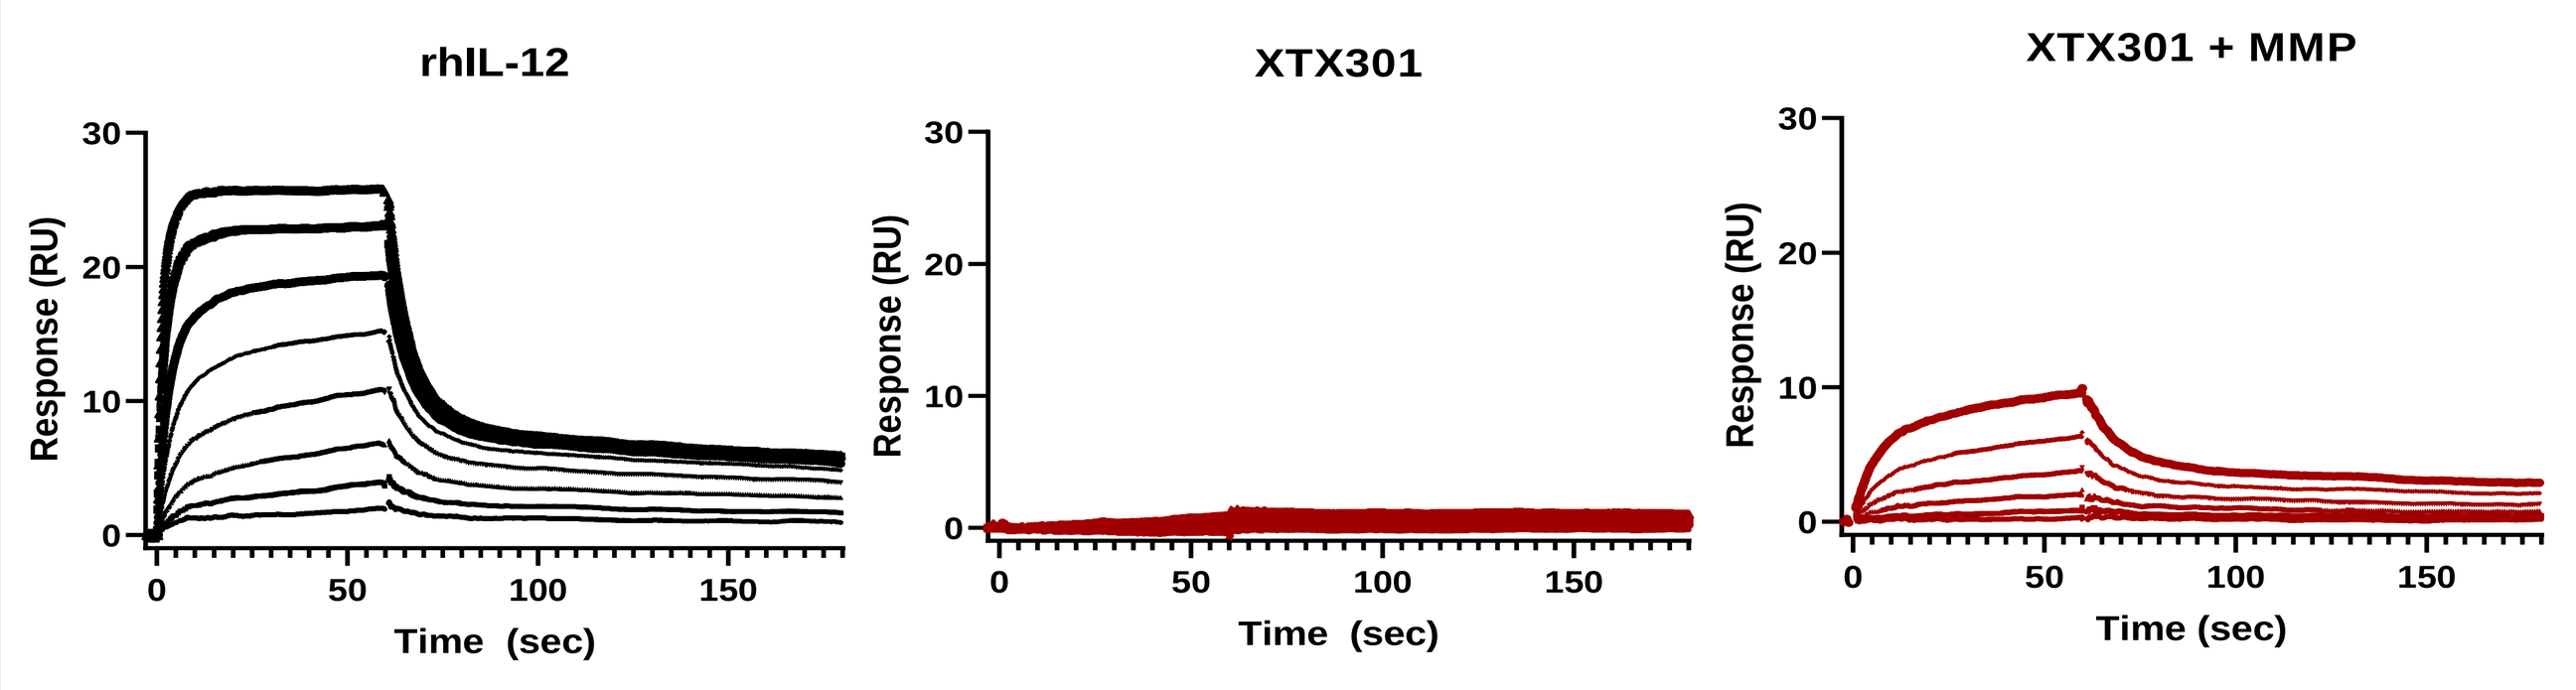

rhIL-12
XTX301
XTX301 + MMP
Response (RU)
Time (sec)

Ligand	Sample	ka (1/Ms)	kd (1/s)	KD (M)
huIL12Rb2	huIL-12	2.05E+06	1.64E-02	7.97E-09
huIL12Rb2	XTX301		-	
huIL12Rb2	XTX301+MMP	8.87E+04	8.27E-03	9.33E-08

Figure 2: Surface plasmon resonance (SPR) was used to measure binding kinetics of (A) XTX301, proteolytically cleaved XTX301, and recombinant human IL-12 to human IL-12 receptor Beta2. IL12 receptor Beta2 was immobilized to a sensor chip, with XTX301, proteolytically cleaved XTX301, and IL-12 flowed over at concentrations of 3.125 nM ~ 400 nM with 2-fold dilution. The concentrations decrease from top to bottom in panels.

Masking and recovery of in vitro activity with XTX301

In vitro potency of proteolytically activated XTX301 was equivalent to unmasked XTX300

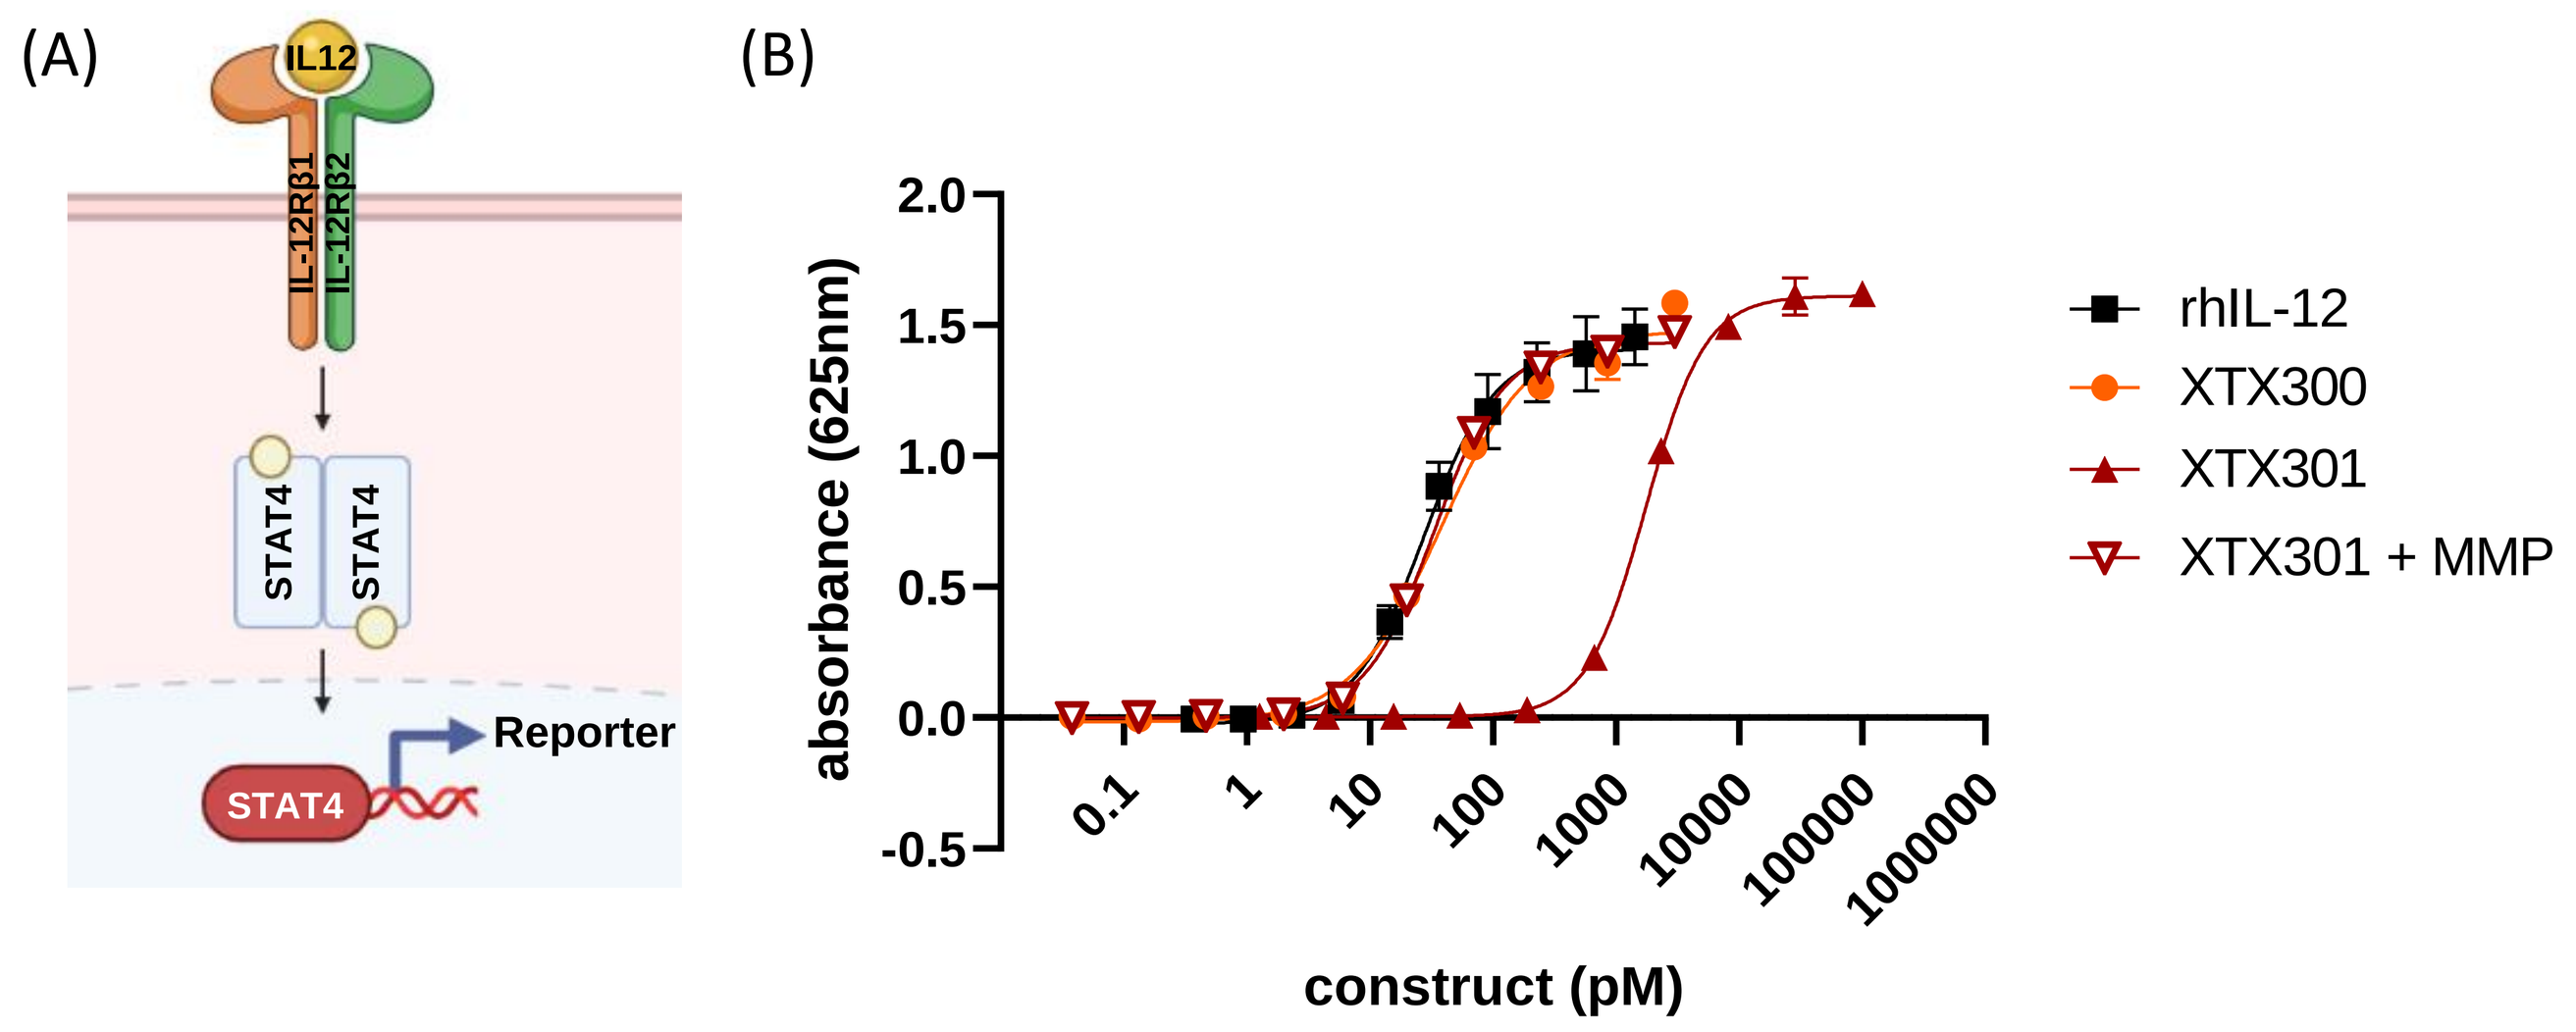

(A)
IL12
IL-12Rβ1
IL-12Rβ2
STAT4
STAT4
Reporter
STAT4
(B)
absorbance (625nm)
construct (pM)
rhIL-12
XTX300
XTX301
XTX301 + MMP

Table 1	rhIL-12	XTX300	XTX301	XTX301 + MMP
EC50 ± SD (pM)	25.5 ± 2.8	29.5 ± 12.3	1584 ± 246	30.5 ± 4.5
Fold-masking (vs. XTX300)			54	

Figure 3: (A) IL-12 reporter gene assay, which is engineered to express a reporter gene upon STAT4 activation downstream of the IL-12 receptor. XTX301 is intended to engage the IL-12 receptor and promote STAT4-mediated transcriptional activity. (B) HEK Blue IL-12 cells were incubated with serial dilutions of test article at 37°C. After 24 h, cell supernatant was collected and measured for secreted embryonic alkaline phosphatase. Each data series was fit using nonlinear regression analysis, and the mean half maximal effective concentration (EC50) and standard deviation (SD) were calculated across 2 independent experiments. The average EC50 of XTX301 was 54-fold greater than the EC50 of the unmasked parental control, XTX300. After incubation with MMP, the EC50 of proteolytically cleaved XTX301 was comparable to XTX300, demonstrating a protease-dependent increase in potency.

XTX301 became pharmacologically active upon proteolytic cleavage in primary human PBMC

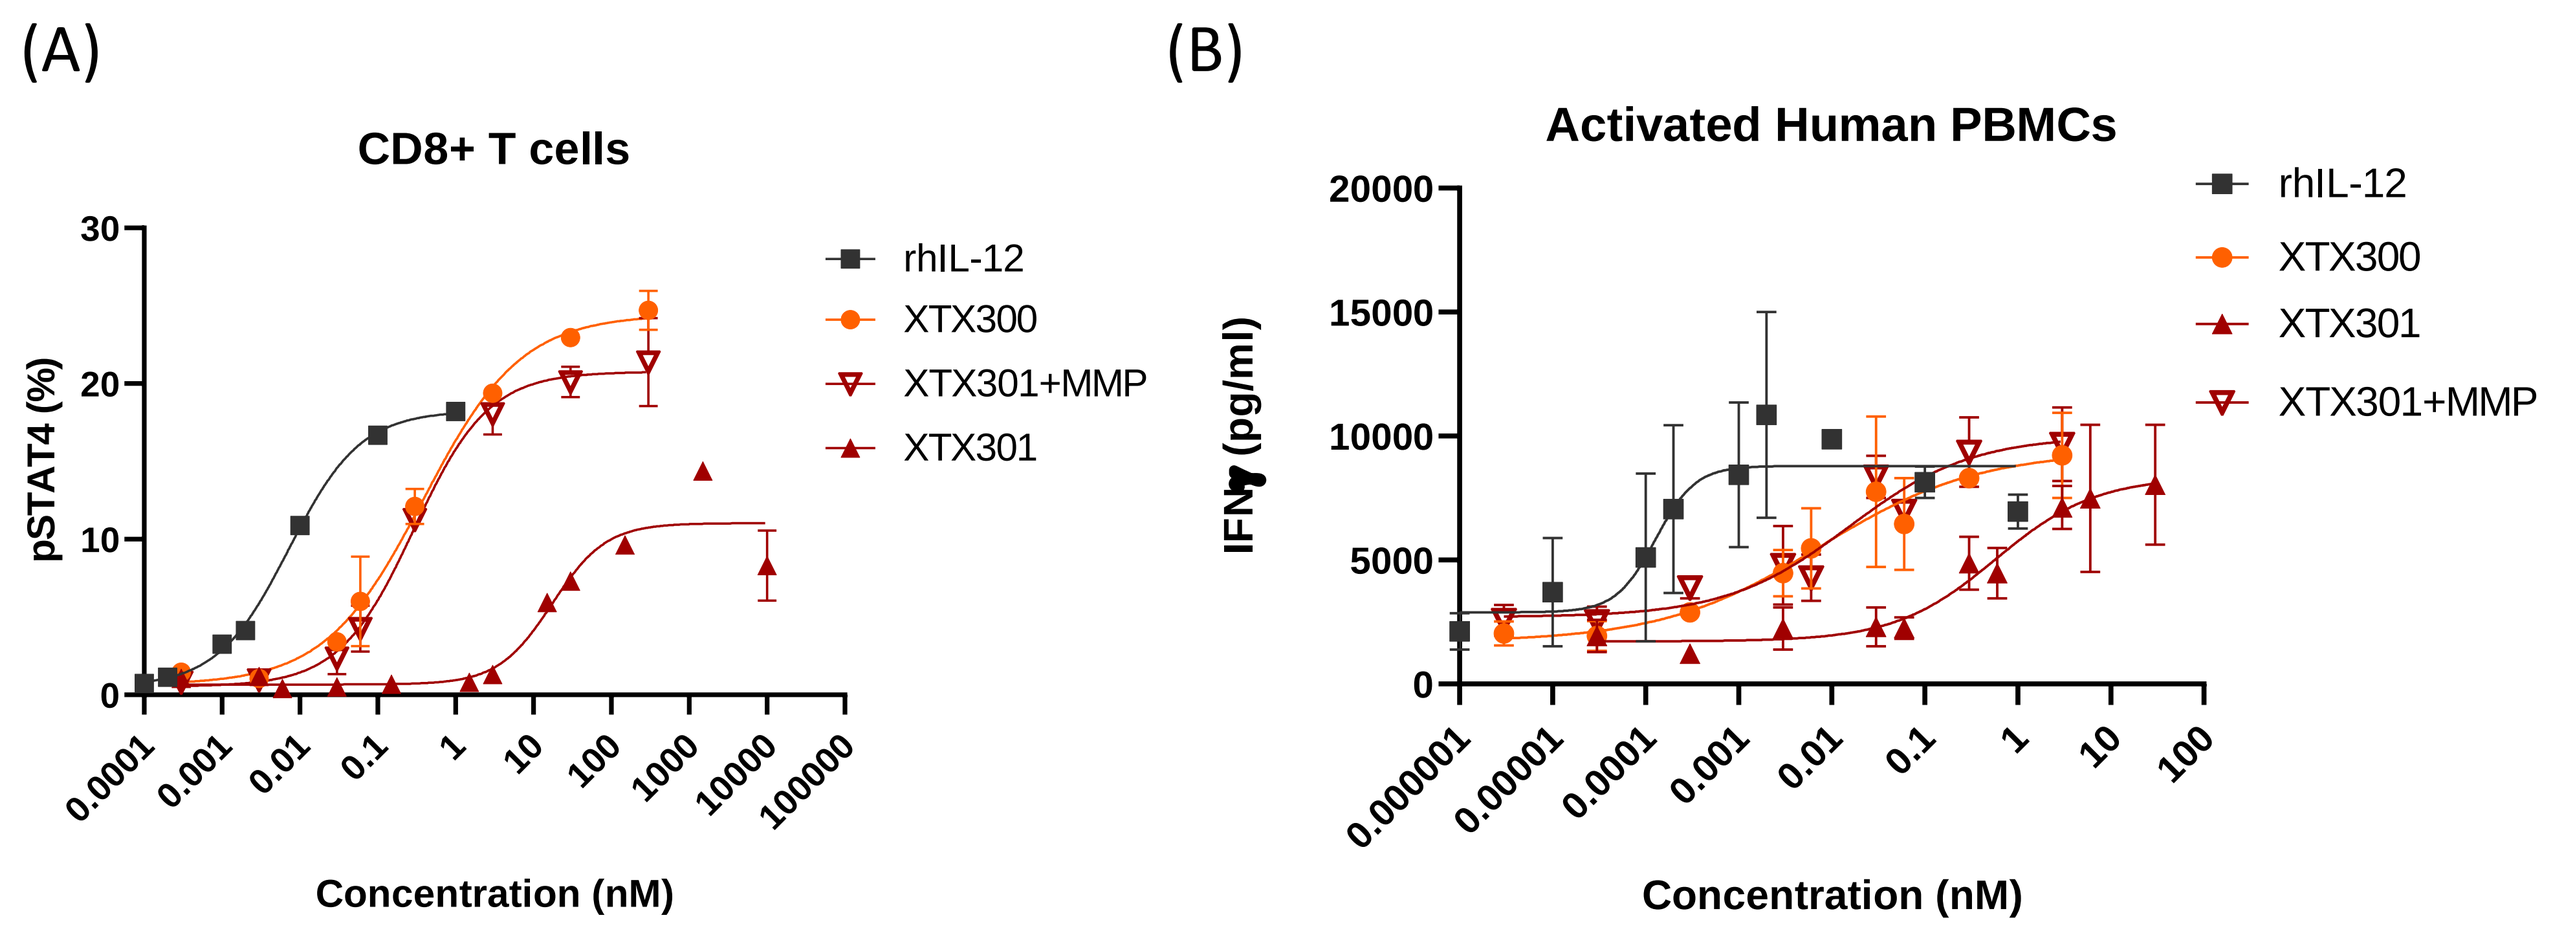

(A)
CD8+ T cells
pSTAT4 (%)
Concentration (nM)
rhIL-12
XTX300
XTX301+MMP
XTX301
(B)
Activated Human PBMCs
IFNγ (pg/ml)
Concentration (nM)
rhIL-12
XTX300
XTX301
XTX301+MMP

Figure 4: (A) Primary human peripheral blood mononuclear cells (PBMCs) were preactivated and evaluated for pSTAT4 phosphorylation by flow cytometry. (B) The ability of XTX301 to induce a functional effect was determined with and without proteolytic cleavage in primary human PBMCs. PBMCs were preactivated as in (A), then incubated with varying doses of rhIL-12 and test articles for 24 hours. XTX301 demonstrated attenuation of IFNγ secretion which was restored to unmasked XTX300 level upon activation by MMP activation.

Anti-tumor activity of XTX301 *in vivo*

mXTX301 demonstrated anti-tumor activity in MC38 (inflamed) and B16F10 (noninflamed) syngeneic tumor models without toxicity

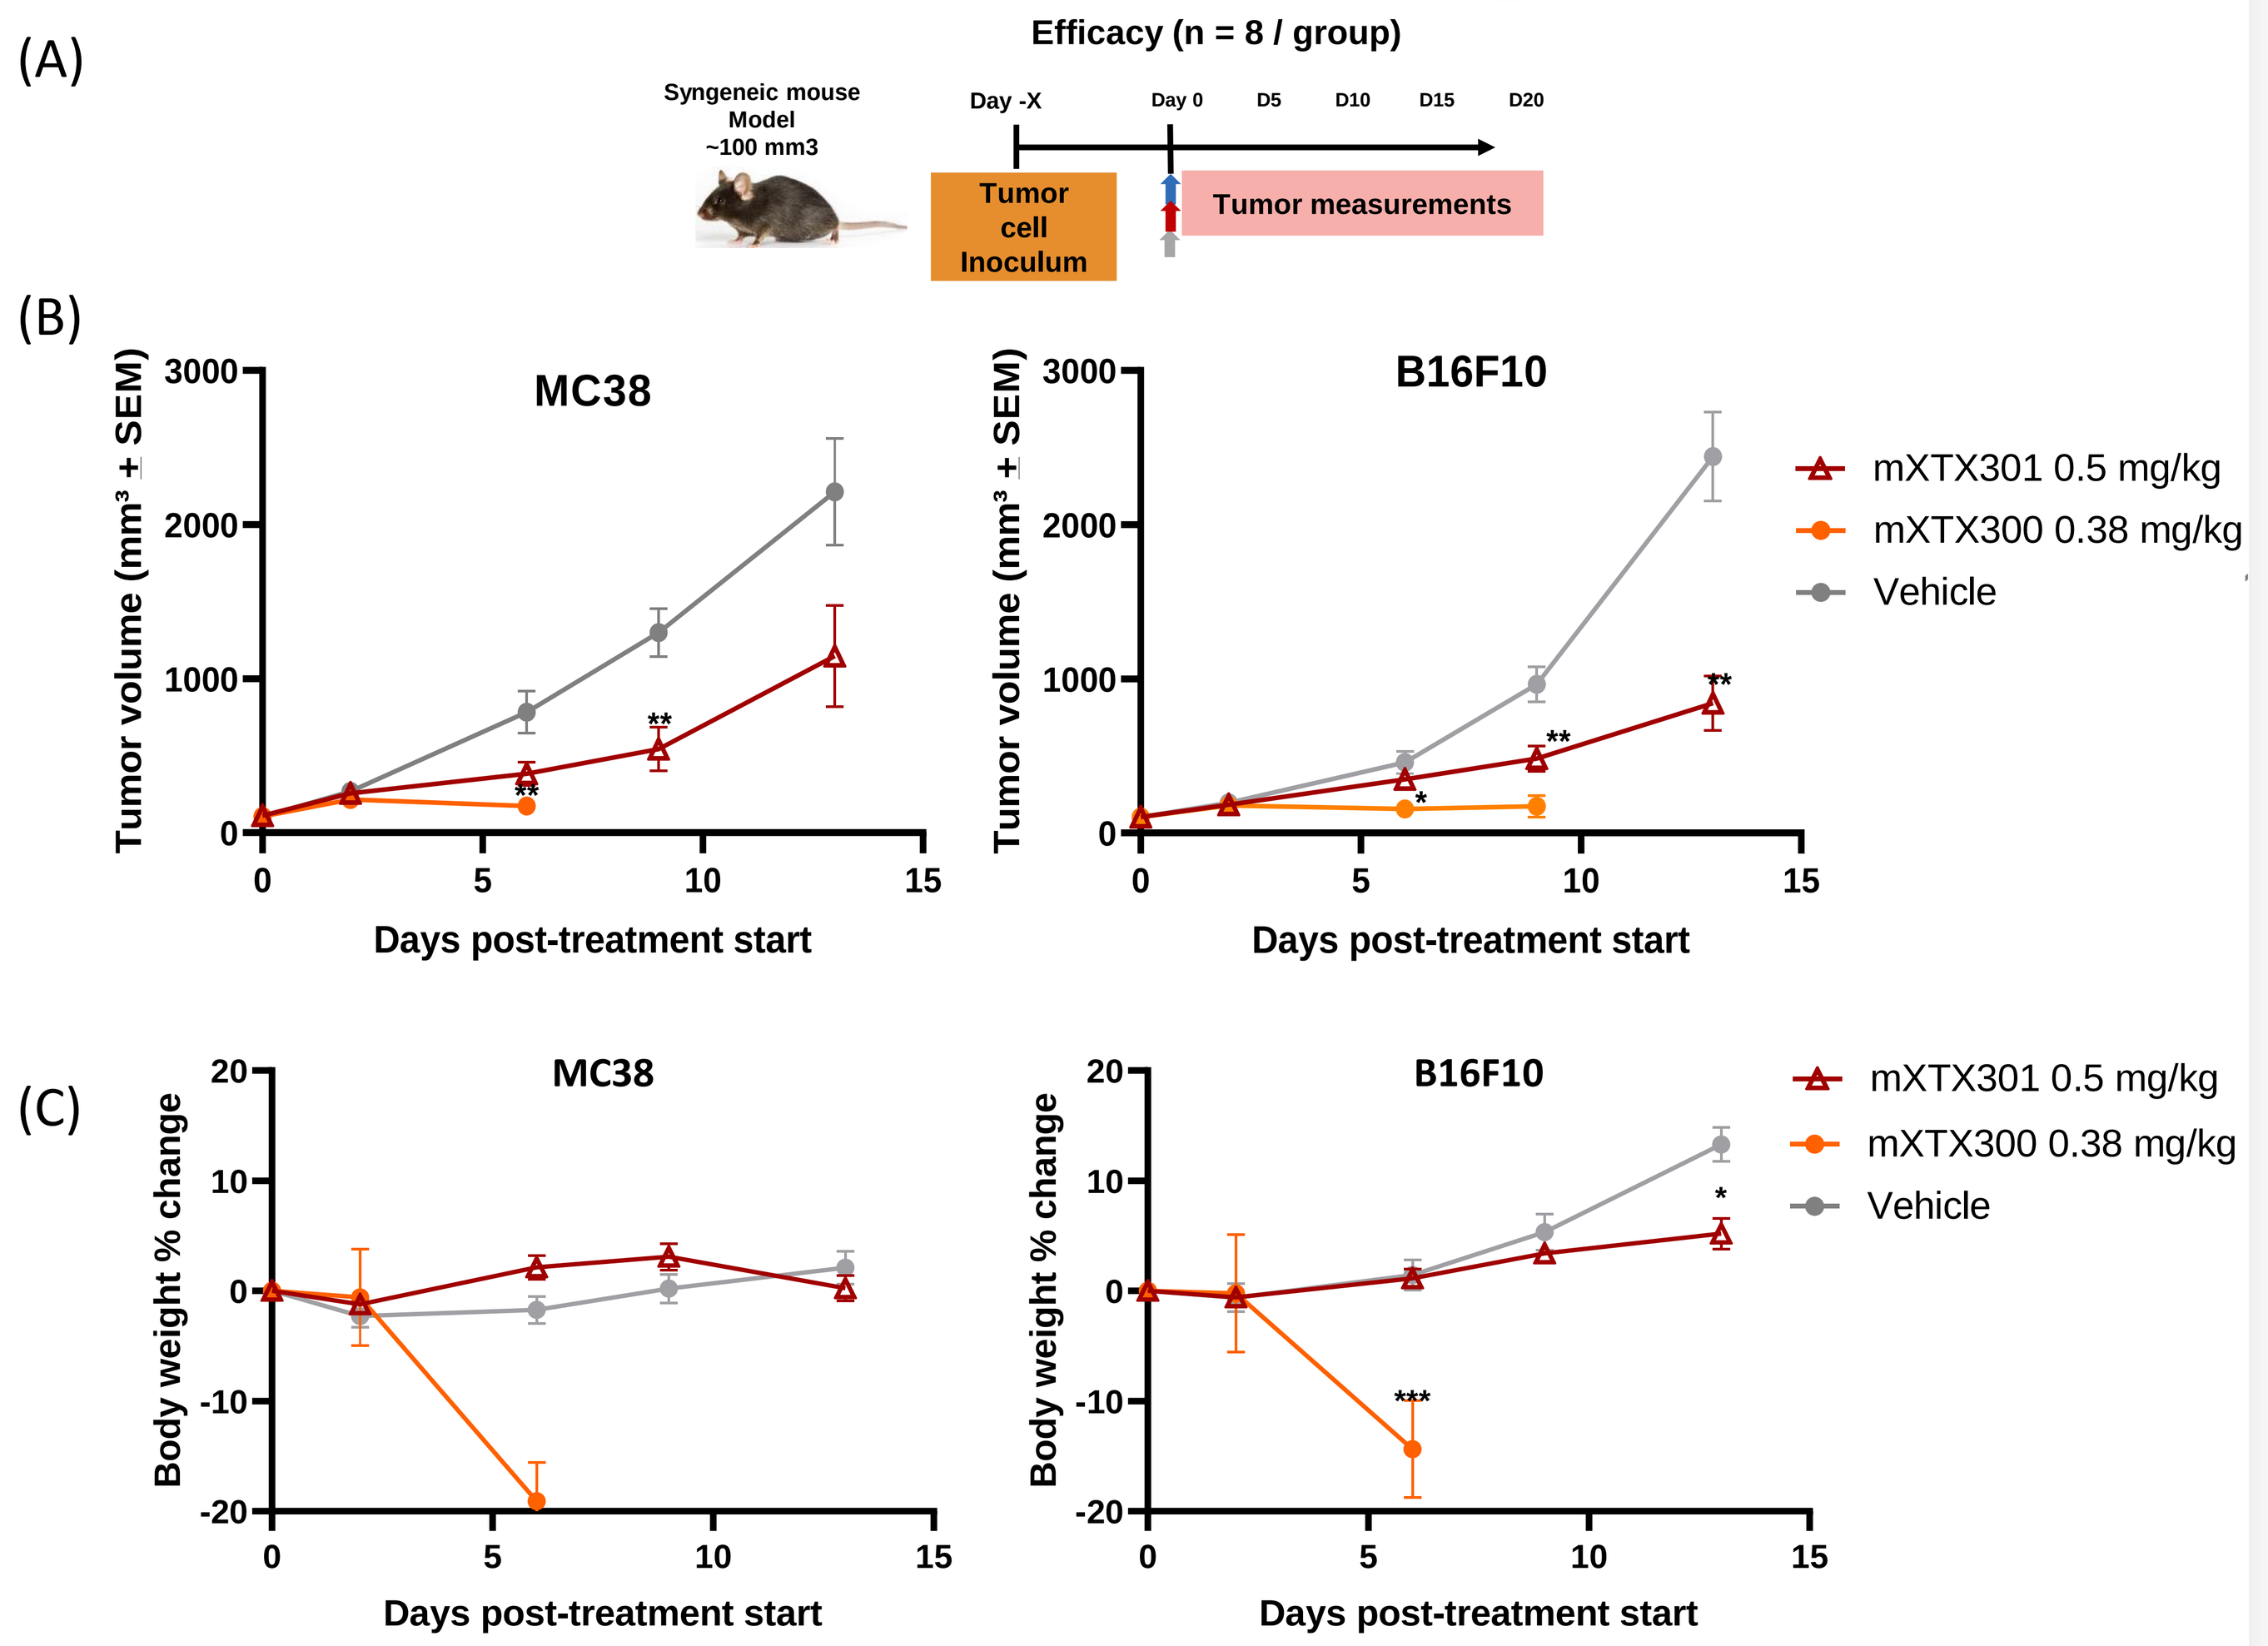

(A)
Efficacy (n = 8 / group)
Syngeneic mouse Model ~100 mm3
Day -X
Day 0
D5
D10
D15
D20
Tumor cell Inoculum
Tumor measurements
(B)
MC38
B16F10
Tumor volume (mm³ + SEM)
Days post-treatment start
mXTX301 0.5 mg/kg
mXTX300 0.38 mg/kg
Vehicle
(C)
MC38
B16F10
Body weight % change
Days post-treatment start
mXTX301 0.5 mg/kg
mXTX300 0.38 mg/kg
Vehicle

Figure 5: Evaluation of anti-tumor activity of mXTX301 in MC38 and B16F10 syngeneic tumor models. (A) Animals were implanted subcutaneously with tumor cells and received a single intravenous injection of mXTX300 at a dose 0.38 mg/kg or mXTX301 at 0.5 mg/kg. Tumor and body weight measurements were taken two/three times a week. (B) mXTX301 demonstrates strong anti-tumor activity in MC38 and B16F10 syngeneic tumor models. Tumor growth data are presented as mean for tumor volume ± SEM. Two-way ANOVA followed by Bonferroni post-hoc test (#p = 0.1; *p < 0.05; **p < 0.01; ***p < 0.0005). (C) mXTX300 therapy was not tolerated and resulted in 20% body weight loss in both MC38 and B16F10 mouse models. Compared to vehicle control, mXTX301 was well tolerated in MC38 model with no significant body weight loss. Body weight % change mean ± SEM was shown. Two-way ANOVA followed by Bonferroni post-hoc test (*p < 0.05; **p < 0.01; ***p < 0.0005).

mXTX301 demonstrated tumor PD and effective peripheral masking in mouse models

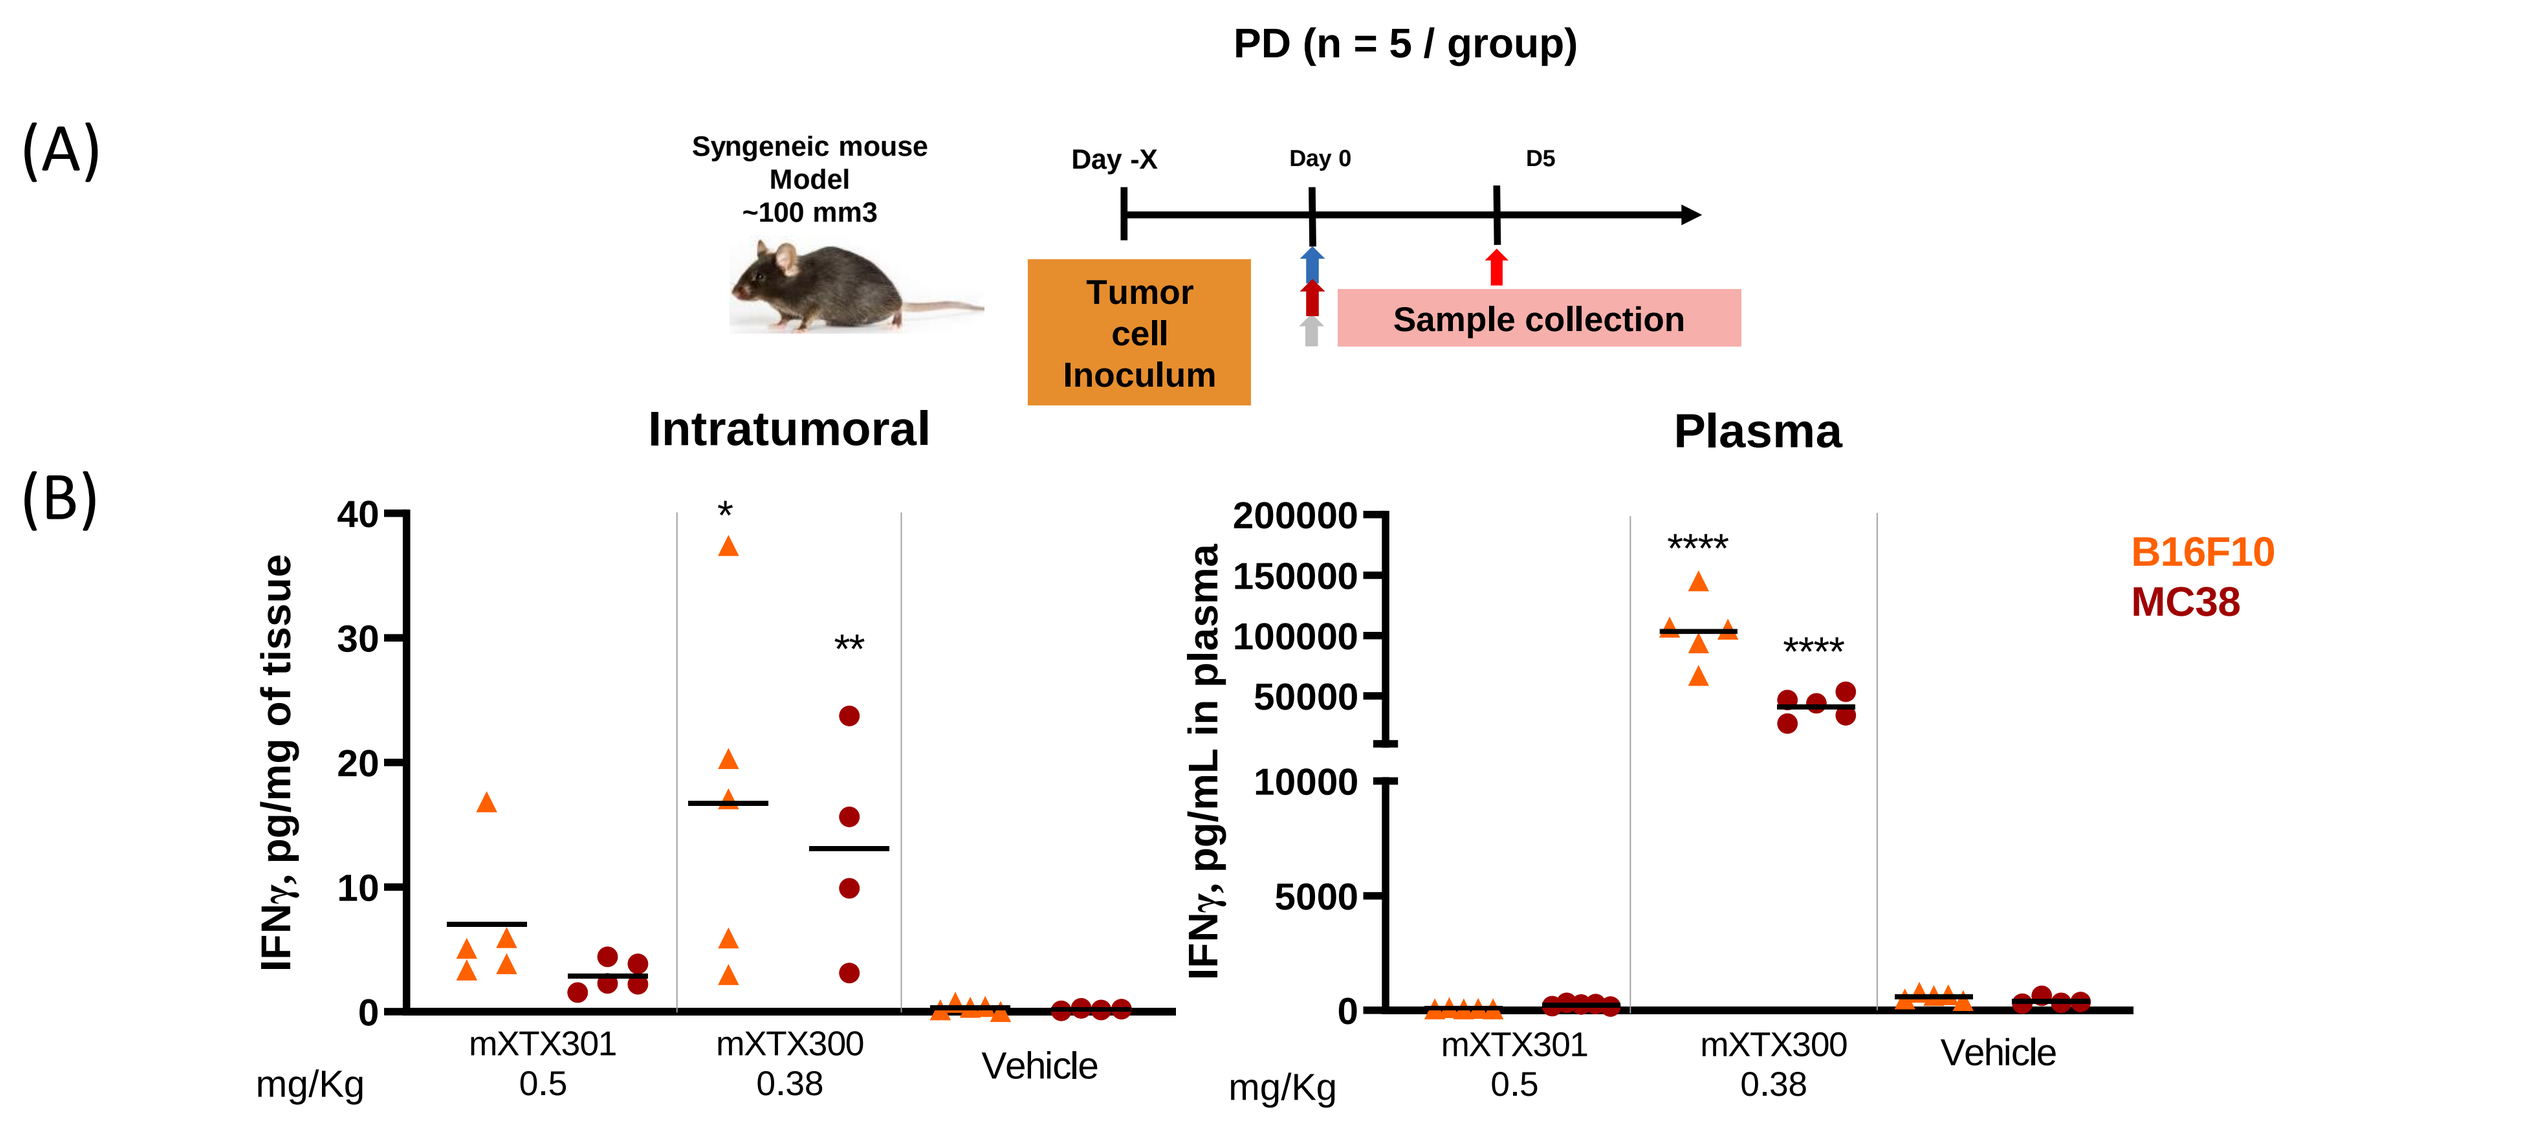

(A)
PD (n = 5 / group)
Syngeneic mouse Model ~100 mm3
Day -X
Day 0
D5
Tumor cell Inoculum
Sample collection
(B)
Intratumoral
Plasma
IFNγ, pg/mg of tissue
IFNγ, pg/mL in plasma
mg/Kg
mXTX301 0.5
mXTX300 0.38
Vehicle
B16F10
MC38

Figure 6: Pharmacodynamic assessment of mXTX301 in syngeneic tumor models. (A) Schematic of the pharmacodynamic study (B) mXTX301 therapy results in intra-tumoral induction of IFNγ in B16F10 and MC38 tumor bearing mice with low systemic exposure. IFNγ measurements in tumor and plasma were performed using MSD assay and displayed as means. One-way ANOVA followed by Bonferroni post-hoc test (#p = 0.17; *p < 0.05; **p < 0.005; ****p < 0.0001).

XTX301 safety assessment in cynomolgus monkeys

XTX301 was well tolerated in an exploratory repeat dose non-human primate study

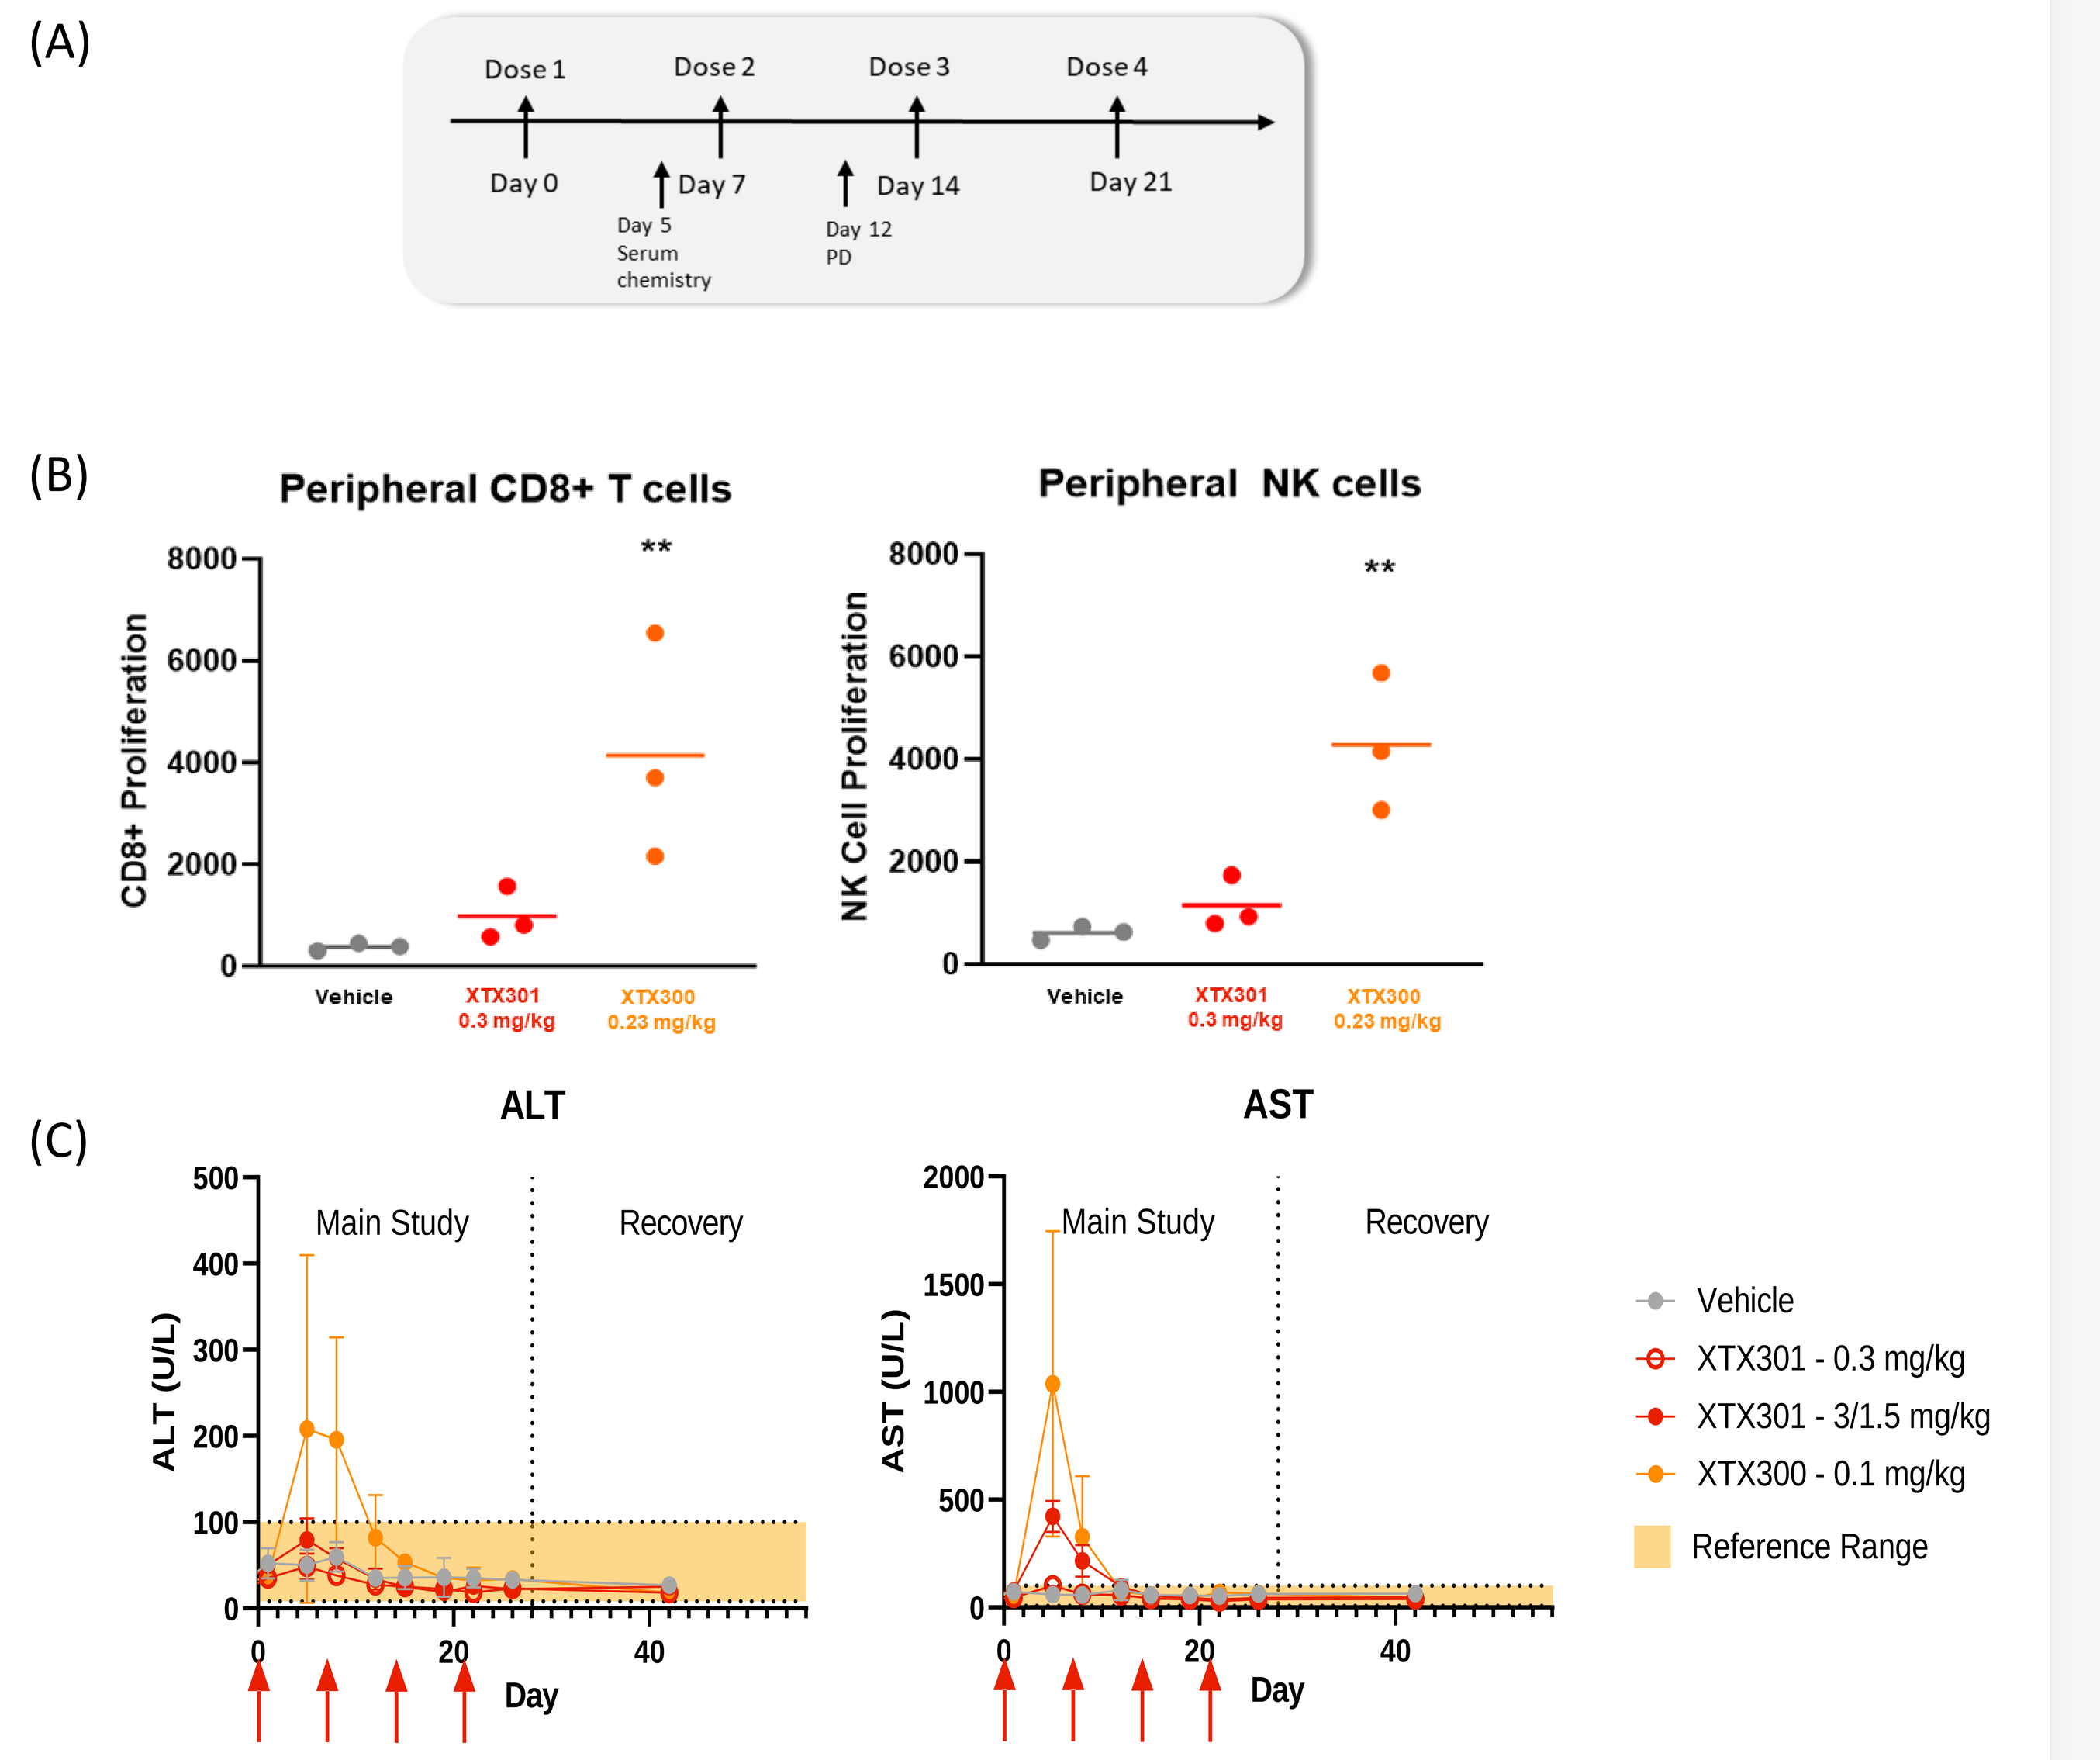

(A)
Dose 1
Dose 2
Dose 3
Dose 4
Day 0
Day 7
Day 14
Day 21
Day 5 Serum chemistry
Day 12 PD
(B)
Peripheral CD8+ T cells
Peripheral NK cells
CD8+ Proliferation
NK Cell Proliferation
Vehicle
XTX301 0.3 mg/kg
XTX300 0.23 mg/kg
(C)
ALT
AST
ALT (U/L)
AST (U/L)
Main Study
Recovery
Day
Vehicle
XTX301 - 0.3 mg/kg
XTX301 - 3/1.5 mg/kg
XTX300 - 0.1 mg/kg
Reference Range

Figure 7: Pharmacodynamics and tolerability of XTX301 and XTX300 (unmasked) were evaluated in cynomolgus monkeys. (A) Study design schematic, Non-masked XTX300 was poorly tolerated at all doses tested, including 0.03 mg/kg and required dose holidays on day 7 and 14 for all animals after the first dose. XTX301 was well tolerated at 0.3 mg/kg and none of the animals required dose holidays. XTX301 was dosed at 3 mg/kg for the first dose and then dropped to 1.5mg/kg for the subsequent three doses. The XTX301 dose was at least 50-fold higher than the MTD of XTX300, which was less than 0.03mg/kg (B) Peripheral activation of CD8+ T cells and NK cells was assessed on Day 12, XTX301 showed minimal peripheral T cell activation and was similar to vehicle control compared to XTX300, demonstrating effective masking of XTX301. One-way non-parametric ANOVA performed to determine the statistical significance of treatment vs vehicle (**P<0.005) (C) 0.1 mg/kg XTX300 (unmasked control) caused elevated AST, ALT, and bilirubin (not shown) on Day 5; 0.3 mg/kg XTX301 caused no transaminase or bilirubin elevations on Day 5, suggestive of effective masking of XTX301. 3.0 mg/kg XTX301 caused elevated AST and bilirubin but returned to normal range after a dose reduction to 1.5 mg/kg.

Conclusions

Our data demonstrated that XTX301 is pharmacologically inactive when in masked form and became activated upon proteolytic cleavage to exert bioactivity comparable to unmasked XTX300. Treatment with mXTX301 resulted in tumor growth inhibition and was well tolerated in syngeneic mouse models compared to unmasked mXTX300. XTX301 was well tolerated in a repeat dose non-human primate safety study.

XTX301 has potential for exerting anti-tumor activity with a favorable tolerability profile and hence enhancing the therapeutic index of IL-12.