XTX202-01, Phase 1/2 First-in Human Study of XTX202, a Masked, Tumor-Activated IL-2βγ, in Patients with Advanced Solid Tumors

611

Diana Hanna, MD[1], Yousef Zakharia, MD[2], Sanjay Goel, MD[3], Randy Hurley, MD[4], Meredith Pelster, MD[5], Anthony El-Khoueiry, MD[1], Jacob Thomas, MD[1], John Powderly, MD, PhD[6], David Miller, MD, PhD[7], Richard Wu, MD, PhD[8], Bartosz Chmielowski, MD, PhD[9], Suthee Rapisuwon, MD[10], Anurag Gupta, PhD[11], Meghan Duncan[11], Aika Siu[11], Ekta Patel, PhD[11], Damiano Fantini[11], PhD, David Crowe[11], Sattanathan Paramasivan, PhD[11], George Lee[11], Katarina Luptakova, MD[11] and Meredith McKean, MD[5]

(1) USC Norris Comprehensive Cancer Center, Los Angeles, CA; (2) University of Iowa Hospitals and Clinics, Iowa City, IA; (3) Rutgers Cancer Institute of New Jersey, New Brunswick, NJ; (4) HealthPartners Cancer Center at Regions Hospital, Saint Paul, MN; (5) Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN; (6) Carolina BioOncology Institute, Huntersville, NC; (7) Massachusetts General Hospital, Boston, MA; (8) The Ohio State University Wexner Medical Center James Cancer Hospital and Solove Research Institute, Columbus, OH; (9) University of California Los Angeles, Jonsson Comprehensive Cancer Center, Los Angeles, CA; (10) MedStar Georgetown University Hospital, Washington, DC; (11) Xilio Therapeutics, Inc., Waltham, MA.

Background

- Historically, rhIL-2 (aldesleukin) has been one of the few treatments for adults with stage IV solid tumors that could result in CRs that were often durable for decades without further therapy.
- Aldesleukin has demonstrated responses in ~15% of RCC and melanoma patients, including durable CRs.
- However, clinical use of aldesleukin has been confined to specialized centers able to provide intensive support, as patients experienced severe and potentially life-threatening adverse events related to systemic exposure to high dose IL-2, including vascular leak syndrome (VLS).
- Low dose rhIL-2 is generally tolerated but predominantly targets α-receptor and thus activates regulatory T cells, leading to immunosuppression.
- The critical challenge in the development of IL-2 therapies is to maximize the efficacy achieved with high dose while improving patient tolerability.

Patient population in pivotal aldesleukin studies were primarily younger patients naïve to systemic therapy with the following characteristics:

- median age 42 (RCC[1]) and 52 (melanoma[2])
- 65% (RCC) and 71% (melanoma) of patients had ECOG PS 0
- only 3% (RCC) and 46% (melanoma) of patients had prior systemic therapy.

Of the multiple prognostic factors analyzed,[1,2] only ECOG PS and prior systemic therapy were associated with response:

- ORR doubled for ECOG PS 0 vs. 1:
 - ORR was 17% and 19% in RCC and melanoma patients with ECOG PS of 0, while it was only 9% in both RCC and melanoma with ECOG PS of 1.
- ORR doubled for patients with no prior systemic therapy:
 - In melanoma, patients who have not received prior systemic therapy had ORR of 21% vs. 10% among patient who received prior systemic therapy.

XTX202, a Masked Tumor-Activated IL-2βγ Designed to Overcome the Limitations of Systemically Active Molecules

- XTX202 is an investigational tumor-activated IL-2βγ designed to be inactive in the periphery and activated by matrix metalloproteases (MMP) in the tumor microenvironment
- XTX202 designed to retain Fc post-activation enabling high, sustained tumor exposure, cross-presentation and receptor binding
- A Phase 1/2 First-In Human clinical trial of XTX202 in advanced solid tumors is ongoing (NCT05052268)

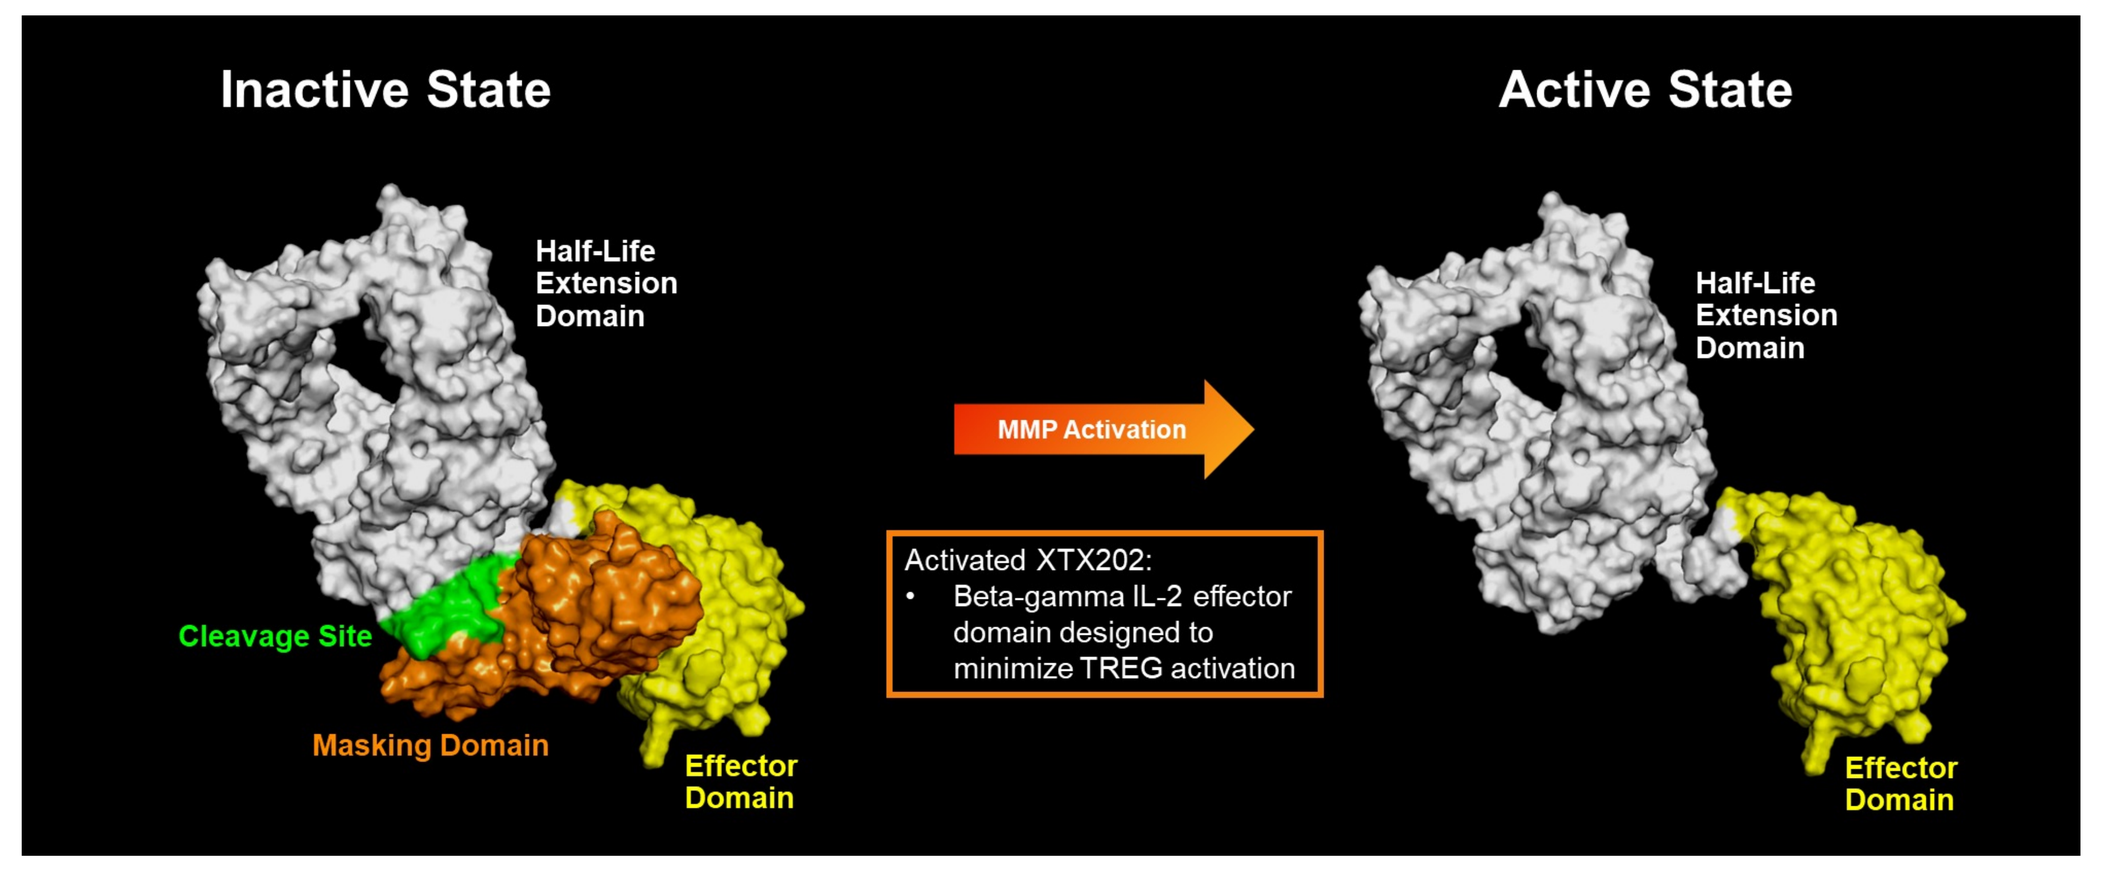
Inactive State
Active State
Half-Life Extension Domain
Cleavage Site
Masking Domain
Effector Domain
MMP Activation
Activated XTX202:
Beta-gamma IL-2 effector domain designed to minimize TREG activation
Half-Life Extension Domain
Effector Domain

Study Design – Enrollment Ongoing

- XTX202 is administered in outpatient setting with a Q3W schedule
- Premedication is not mandatory and was not allowed in first cycle in Phase 1A

Current dose level: 4.0 mg/kg

Phase 1A
Monotherapy Dose Escalation
Advanced Solid Tumors

Phase 1B
Monotherapy
Pharmacodynamic Cohort

Dose Level 1: 1.4 mg/kg
Dose Level 2: 4.0 mg/kg*

Phase 2, Cohort A
Monotherapy Expansion
RCC Cohort

Phase 2, Cohort B
Monotherapy Expansion
Melanoma Cohort

* Evaluating more than 1 dose level in Phase 2 in line with FDA Project Optimus guidance

Results

Baseline Patient Demographics and Disease Characteristics

Phase 1: 54 Patients Enrolled with a Wide Range of Advanced & IO-Treatment Refractory Solid Tumors

Patient Characteristics	Total (N=54)
Demographics	
Age, median (range)	67 (25, 82)
Female	20 (37%)
ECOG PS 0	20 (37%)
ECOG PS 1	34 (63%)
Prior Lines of Anti-Cancer Treatment	**Median 4 (1-14)**
1	5 (9%)
2	9 (17%)
3	7 (13%)
4	13 (24%)
5	9 (17%)
6 or more	11 (20%)
Prior Treatment with IO	
≥1	37 (69%)
Time since initial diagnosis (months)	**Median 29 (4-147)**

74% of patients had 3 or more prior lines of treatment

Tumor Types	Total (N=54)*
Colorectal ca	8
NSCLC	7
Melanoma	6
Sarcoma	5
Pancreatic ca	4
RCC	4
Prostate cancer	3
Endometrial ca	2
Cervical cancer	1
Esophageal ca	1
Ovarian cancer	1
Other	13

Disposition	Total (N=54)
Continuing on Treatment	**15**
Discontinued Treatment	**39**
Progressive Disease	30
Adverse Events (not treatment related)	1
Consent Withdrawal (hospice)	1
Death Due to Progressive Disease	4
Other	3

- One patient at 2.8 mg/kg dose in Part 1A had 2 primary cancer diagnoses of pancreatic cancer and prostate cancer and is therefore included in both tumor types in the table.

Phase 2: 8 Patients Enrolled; All Progressed on Prior IO Therapy

Patient Characteristics	Total (N=8)
Demographics	
Age, median (range)	62 (33, 74)
Female	2 (25%)
ECOG PS 0	4 (50%)
ECOG PS 1	4 (50%)
Prior Lines of Anti-Cancer Treatment	**Median 3.5 (1-12)**
1	3 (37.5%)
2	0
3	1 (12.5%)
4	1 (12.5%)
5	0
6 and more	3 (37.5%)
Prior Treatment with IO	
≥1	8 (100%)
Time since initial diagnosis (months)	**Median 50 (12-198)**

Tumor Types	Total (N=8)
Melanoma	6
RCC	2

Disposition	Total (N=8)
Continuing on Treatment	**5**
Discontinued Treatment	**3**
Progressive Disease	3

Phase 1: MTD Has Not Been Reached

No signs or symptoms of VLS reported through the 4 mg/kg dose level

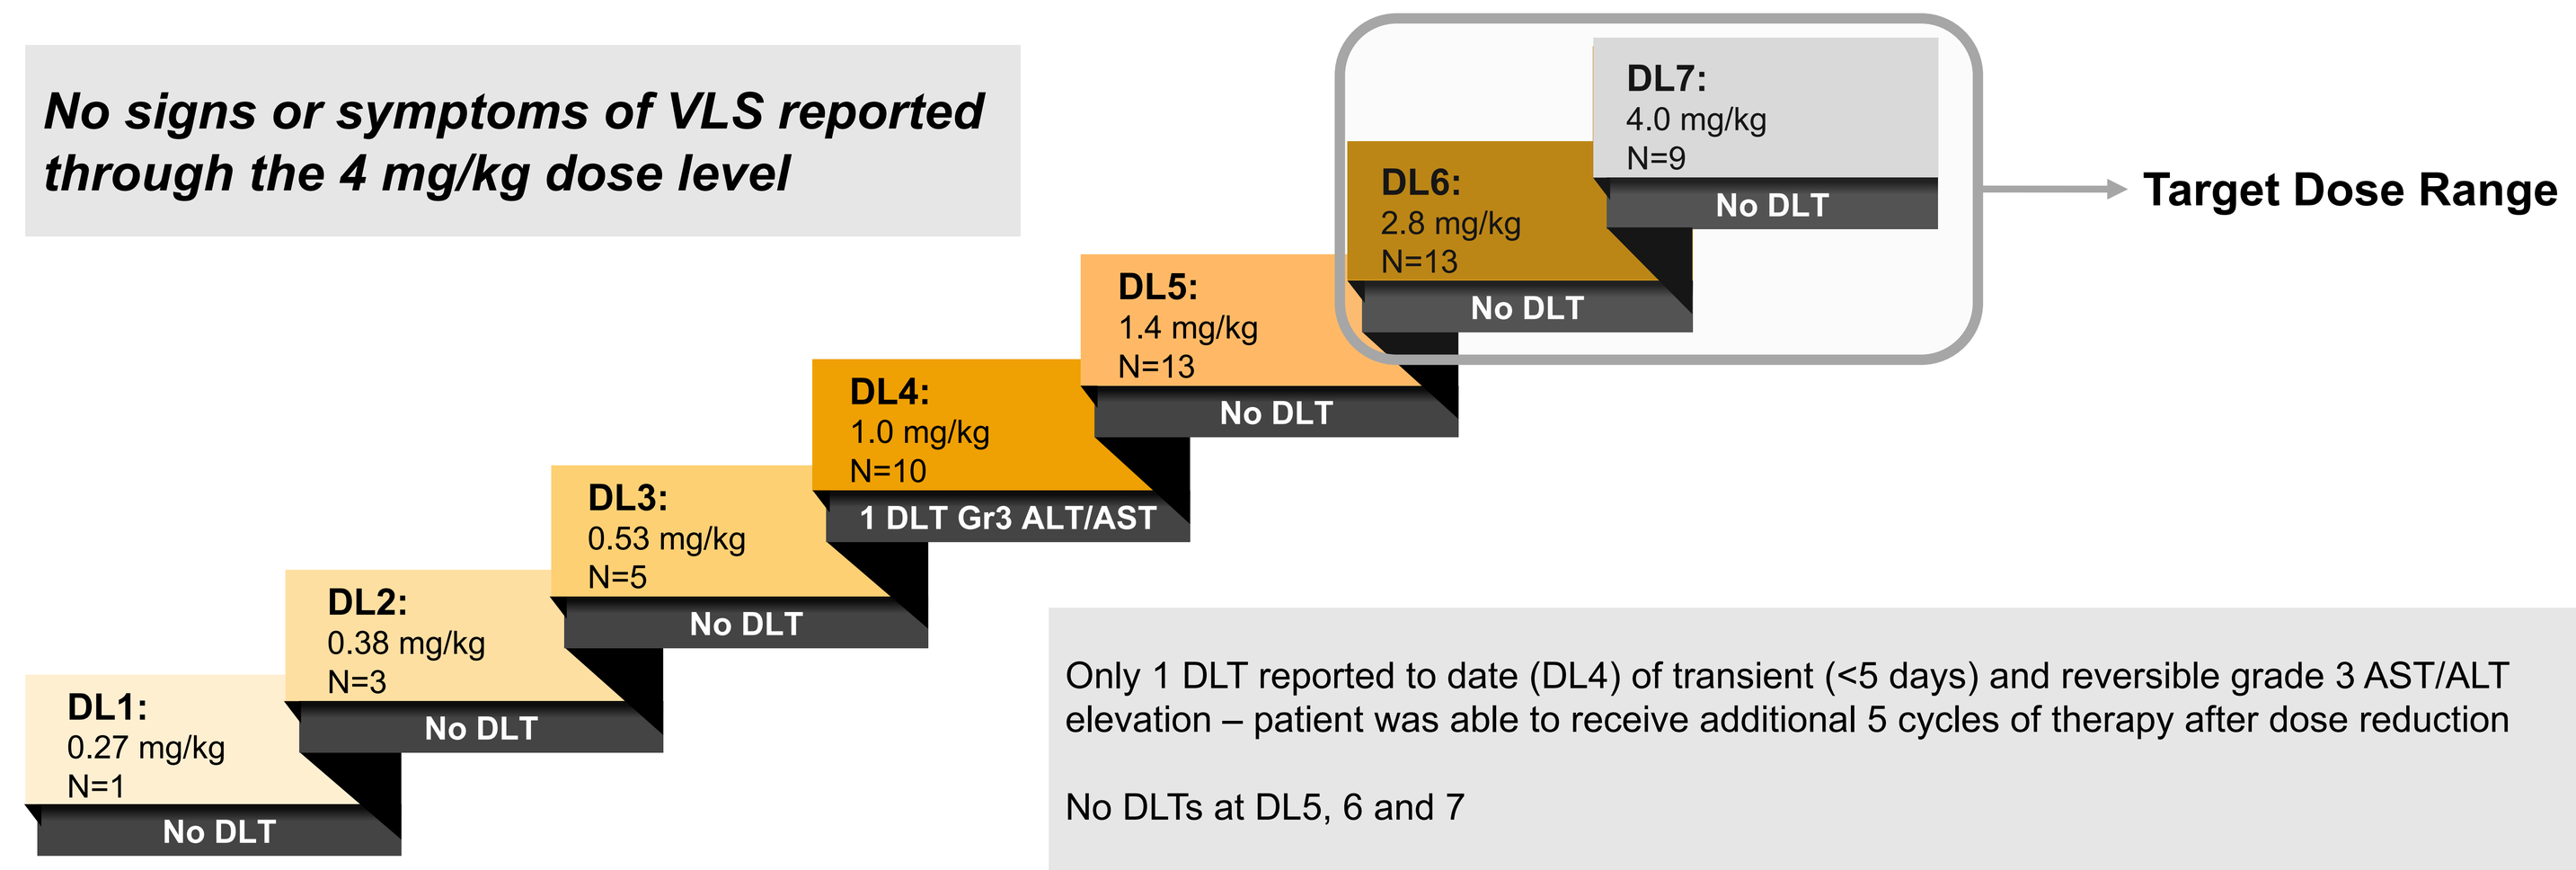
DL7:
4.0 mg/kg
N=9
No DLT
DL6:
2.8 mg/kg
N=13
No DLT
DL5:
1.4 mg/kg
N=13
No DLT
DL4:
1.0 mg/kg
N=10
1 DLT Gr3 ALT/AST
DL3:
0.53 mg/kg
N=5
No DLT
DL2:
0.38 mg/kg
N=3
No DLT
DL1:
0.27 mg/kg
N=1
No DLT
Target Dose Range

Only 1 DLT reported to date (DL4) of transient (<5 days) and reversible grade 3 AST/ALT elevation – patient was able to receive additional 5 cycles of therapy after dose reduction

No DLTs at DL5, 6 and 7

All dose levels are Q3W outpatient administration.

XTX202 Safety: TRAEs Primarily Grade 1-2

AE Category / Term *TRAEs with ≥10% incidence (any grade)*	All Patients Phase 1 and Phase 2 All dose levels *(n=62)*		All patients Phase 1 and Phase 2 1.4 mg/kg or higher dose level *(n=43)*	
	Any Grade	Grade ≥3	Any Grade	Grade ≥3
Fatigue	19% (n=12)	0	16% (n=7)	0
Pyrexia	18% (n=11)	0	23% (n=10)	0
Chills	16% (n=10)	2% (n=1)	23% (n=10)	2% (n=1)
Lymphocyte count decreased	15% (n=9)	8% (n=5)	14% (n=6)	9% (n=4)
Dose reduction due to TRAE	3% (n=2)		2% (n=1)	
Treatment discontinuation due to TRAE	0		0	

- No Grade 5 TRAEs
- Only Grade 4 TRAEs included 2 events of Grade 4 lymphocyte count decreased /lymphopenia, both transient (< 3 days)
- Grade 3 TRAEs not included above (n=1 each): diarrhea; colitis; myalgia; hypoxia; lymphopenia; AST/ALT increased
 The events of Grade 3 diarrhea and Grade 3 colitis were reported in the same patient treated at 1.4 mg/kg. The patient was treated with 40 mg prednisone for 1 week followed by a taper and the AEs resolved.

No treatment discontinuation due to TRAEs and minimal dose reduction due to TRAEs
No signs/symptoms of Vascular Leak Syndrome (VLS) observed at any dose level

XTX202 Demonstrated Dose Proportional PK

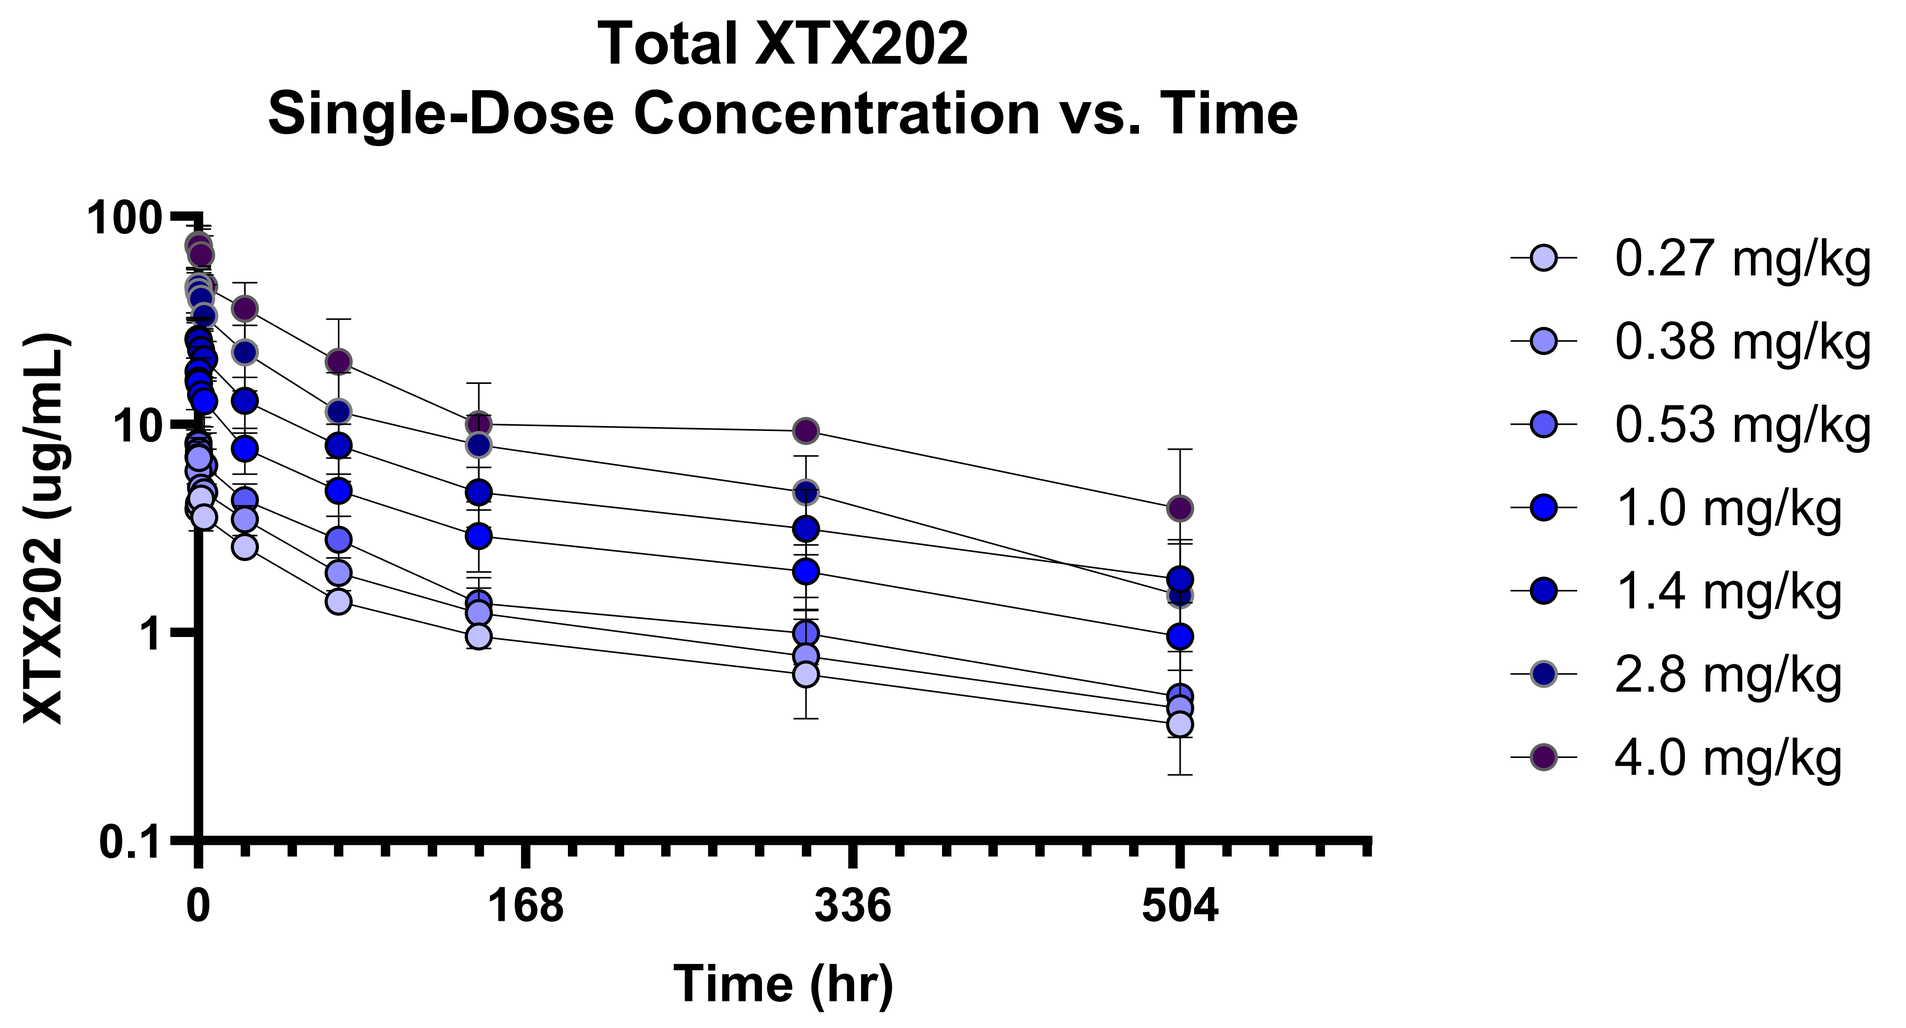
Total XTX202
Single-Dose Concentration vs. Time
XTX202 (ug/mL)
100
10
1
0.1
0
168
336
504
Time (hr)
0.27 mg/kg
0.38 mg/kg
0.53 mg/kg
1.0 mg/kg
1.4 mg/kg
2.8 mg/kg
4.0 mg/kg

XTX202 On-Treatment Biopsy Demonstrated ~15% Activated Molecule in Tumor vs < 1% Activated Molecule in Plasma

CLINICAL DATA

- Patient with leiomyosarcoma treated with XTX202 at 2.8 mg/kg Q3W; tumor specimen collected cycle 2, day 2 (C2D2)
- >40-fold increase of active XTX202 in tumor relative to plasma for patients at 2.8 mg/kg dose level
- Well above minimum design criteria and consistent with range that enabled T cell and NK cell stimulation in preclinical models

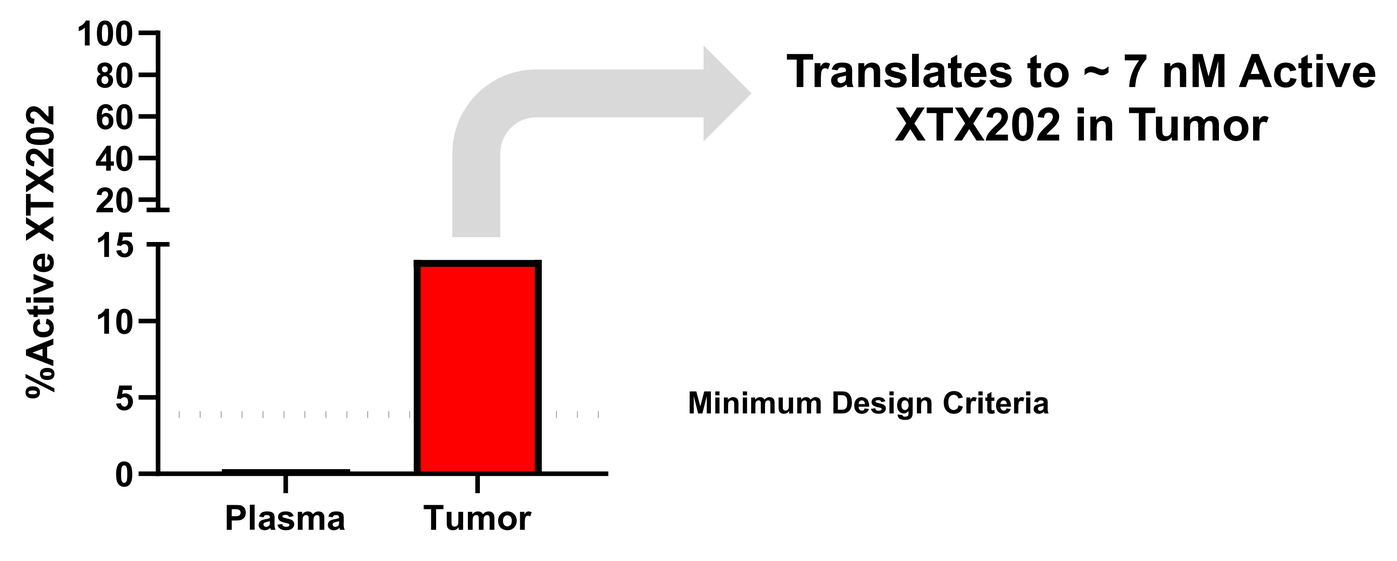
%Active XTX202
100
80
60
40
20
15
10
5
0
Plasma
Tumor
Translates to ~ 7 nM Active XTX202 in Tumor
Minimum Design Criteria

PRECLINICAL DATA

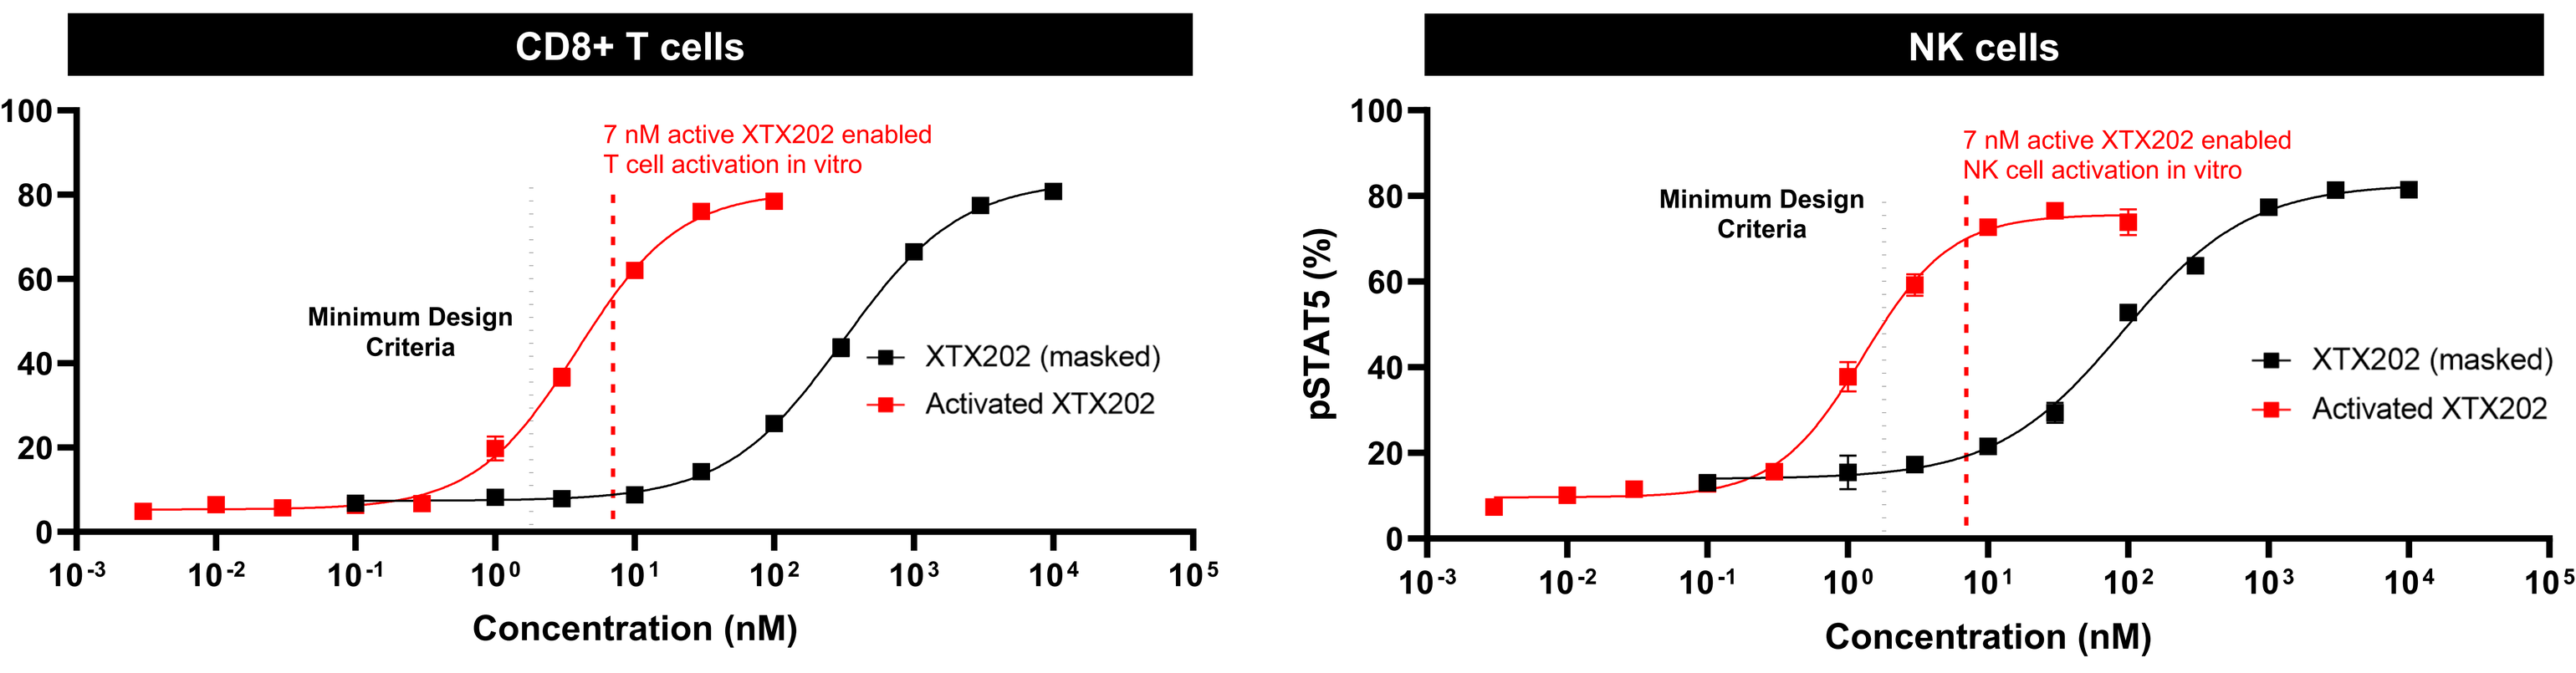
CD8+ T cells
pSTAT5 (%)
7 nM active XTX202 enabled T cell activation in vitro
Minimum Design Criteria
XTX202 (masked)
Activated XTX202
Concentration (nM)
NK cells
7 nM active XTX202 enabled NK cell activation in vitro
Minimum Design Criteria
XTX202 (masked)
Activated XTX202
Concentration (nM)

Data suggest 2.8 mg/kg or higher monotherapy doses are approaching optimal range to activate CD8+ effector T cells and NK cells in the tumor

Top: Patient biopsy was the only one available for XTX202 bioanalytical analysis. Percent activated XTX202 in tumor was calculated using raw liquid chromatography/ mass spectrometry data. Tumor biopsy specimen was collected C2D2. Percent activated molecule in plasma represents the average for area under the curve (AUC) for Cycle 1 for patients treated at 2.8 mg/kg dose level.

Bottom: Primary human peripheral blood mononuclear cells were treated with a dose-titration of activated XTX202 (unmasked, red) or XTX202 (masked, black) and pSTAT5 positivity was assessed by flow cytometry. The concentration of active XTX202 detected in the human biopsy (7 nM) is overlayed as a red vertical dashed line.

Dose-Dependent Increase in CD8+ T and NK Cells Observed in Response to XTX202 Treatment, Consistent with IL-2βγ Biology

XTX202 Induced T and NK Cell Proliferation in a Dose-Dependent Manner

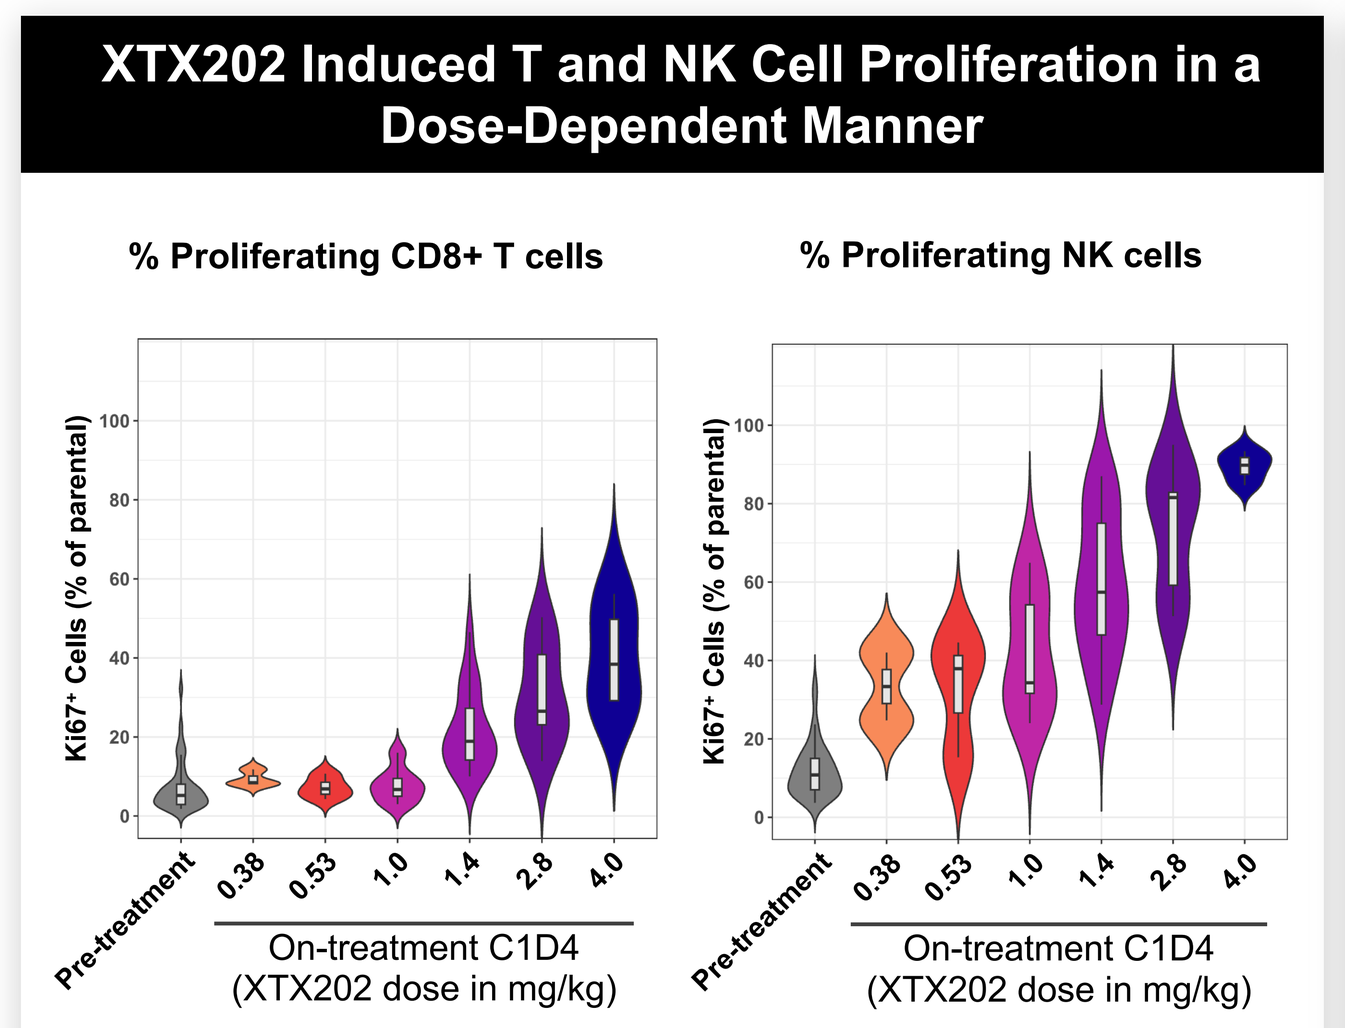
% Proliferating CD8+ T cells
% Proliferating NK cells
Ki67+ Cells (% of parental)
Pre-treatment
0.38
0.53
1.0
1.4
2.8
4.0
On-treatment C1D4
(XTX202 dose in mg/kg)

XTX202 Treatment Resulted in Dose-Dependent Upregulation of Key T and NK Cell Markers

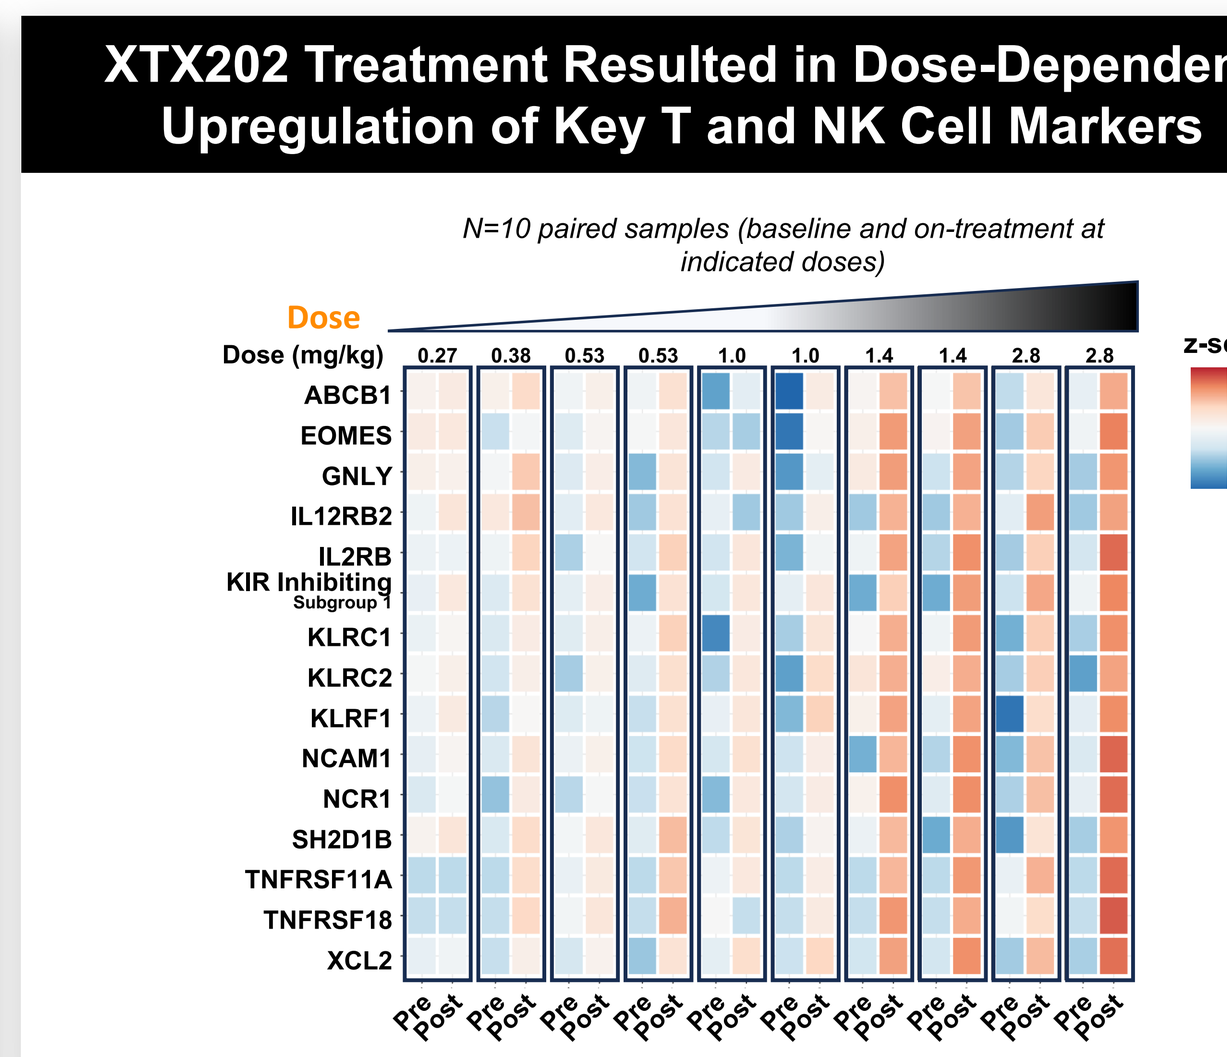
N=10 paired samples (baseline and on-treatment at indicated doses)
Dose
Dose (mg/kg)
0.27
0.38
0.53
0.53
1.0
1.0
1.4
1.4
2.8
2.8
ABCB1
EOMES
GNLY
IL12RB2
IL2RB
KIR Inhibiting Subgroup 1
KLRC1
KLRC2
KLRF1
NCAM1
NCR1
SH2D1B
TNFRSF11A
TNFRSF18
XCL2
z-score
+2
0
-2
Pre
Post

Left Panel: %Ki67⁺ cell populations determined by flow cytometry of peripheral blood mononuclear cells at pre-dose and at cycle 1 day 4 (C1D4).
Right Panel: Heatmap showing standardized expression (z-score-transformed log2-nanoString counts) of top differentially expressed genes (y-axis), separated pre vs. post treatment across all paired XTX202 samples.

No Observed Increase in Regulatory T Cells, Consistent with IL-2βγ Biology

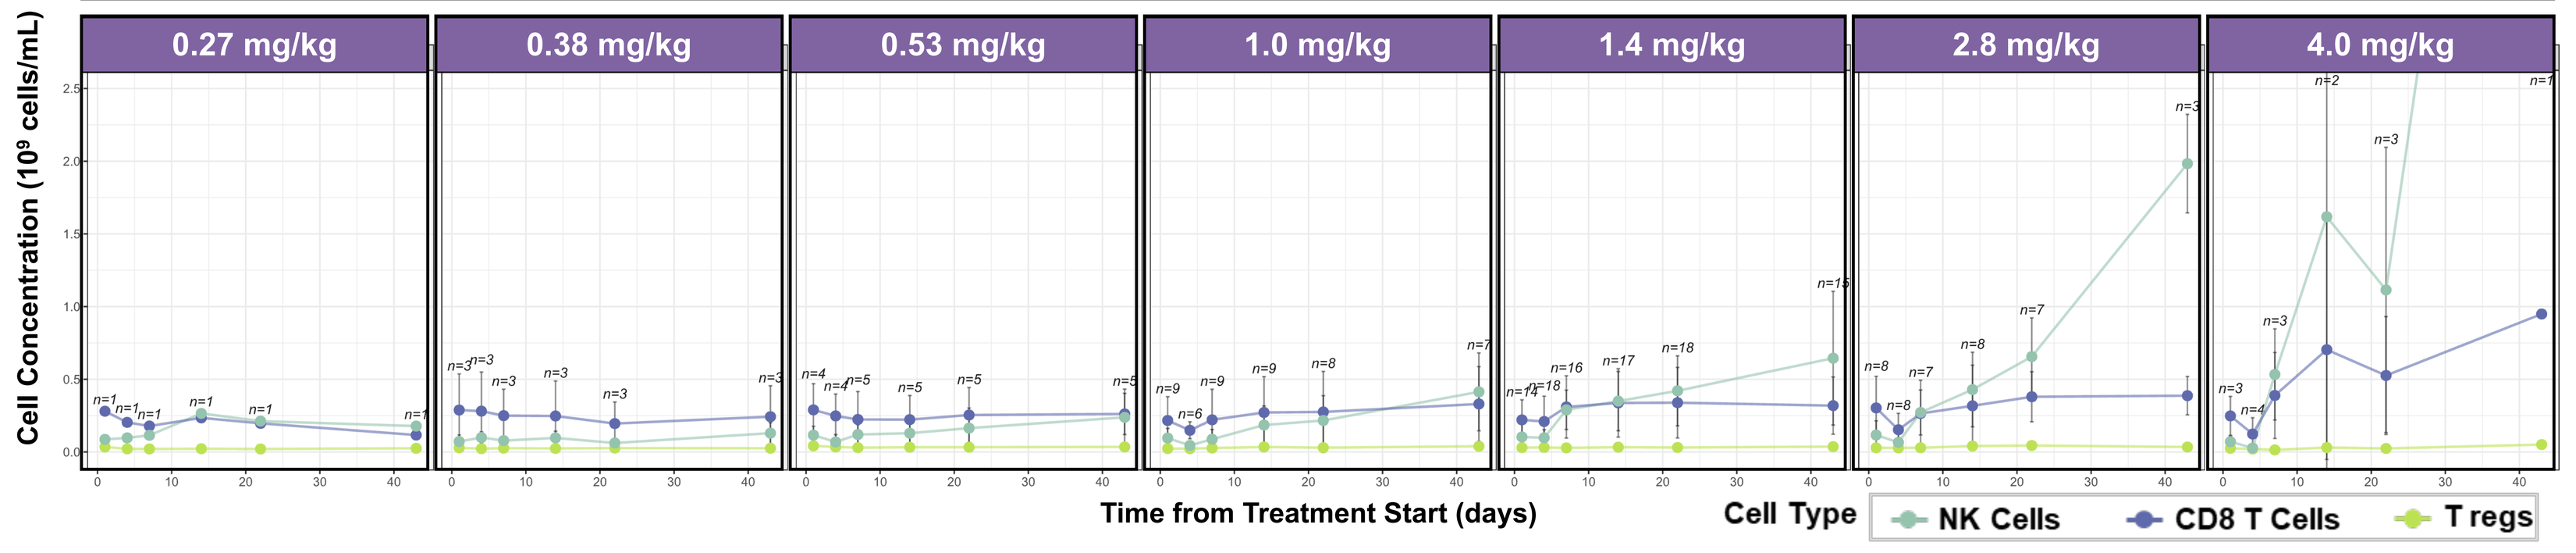
0.27 mg/kg
0.38 mg/kg
0.53 mg/kg
1.0 mg/kg
1.4 mg/kg
2.8 mg/kg
4.0 mg/kg
Cell Concentration (10^9 cells/mL)
Time from Treatment Start (days)
Cell Type
NK Cells
CD8 T Cells
T regs

Absolute Lymphocytes Count: Spaghetti graph showing the average concentration of different cell types in the periphery (y-axis, concentration expressed in 10^9 cells / L) at different timepoints (x-axis) for each treatment dose group (columns). Colors indicate cell types. Points and error bars indicate average +/- sd. The sample size is annotated for each group and timepoint.

50% DCR at Doses ≥2.8 mg/kg, 31% DCR Across All Dose Levels

Dose Level (mg/kg)*	# Patients Treated (Phase 1 and Phase 2)	# EOT Without Response Assessment	# Ongoing Before 1st Response Assessment	# Response Evaluable	# SD for 9+ Weeks as BOR	DCR † (% of evaluable)
<1	7	0	0	7	1	14%
1	9	1	0	8	1	13%
1.4	24	1	2	21	8	38%
≥2.8	22	6	10	6	3	50%
All	62	8	12	42	13	31%

† DCR defined as SD or PR at 9+ weeks
* Patients who had dose increase (n=3) are categorized under the highest received dose level

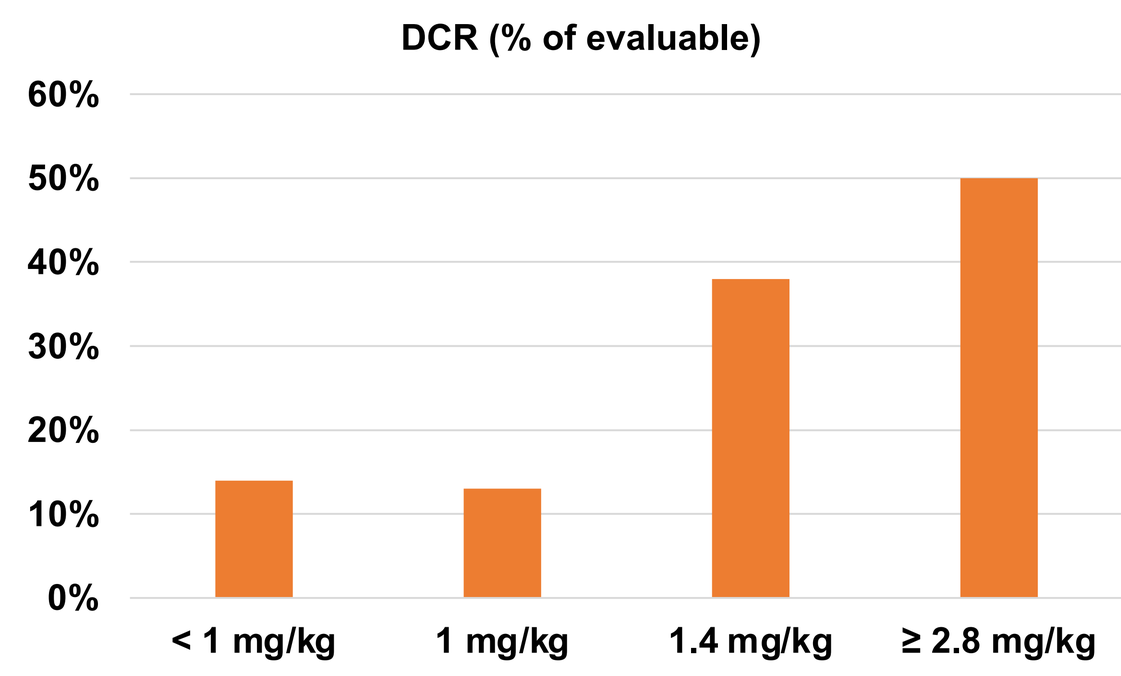
DCR (% of evaluable)
60%
50%
40%
30%
20%
10%
0%
< 1 mg/kg
1 mg/kg
1.4 mg/kg
≥ 2.8 mg/kg

Of 42 response-evaluable patients:

- only 6 patients were treated at a dose level of 2.8 mg/kg or higher as of the data cutoff date.
- among these 6 patients, disease control rate was 50%.

2 patients, including a treatment-refractory MSS CRC patient and a RCC patient, are ongoing on treatment for over 20+ cycles (14 months), suggesting XTX202 is well-tolerated for long-term therapy.

Tumor-Selective Increases in CD8+ Effector T Cells Observed with XTX202 Across Dose Levels in Multiple Indications

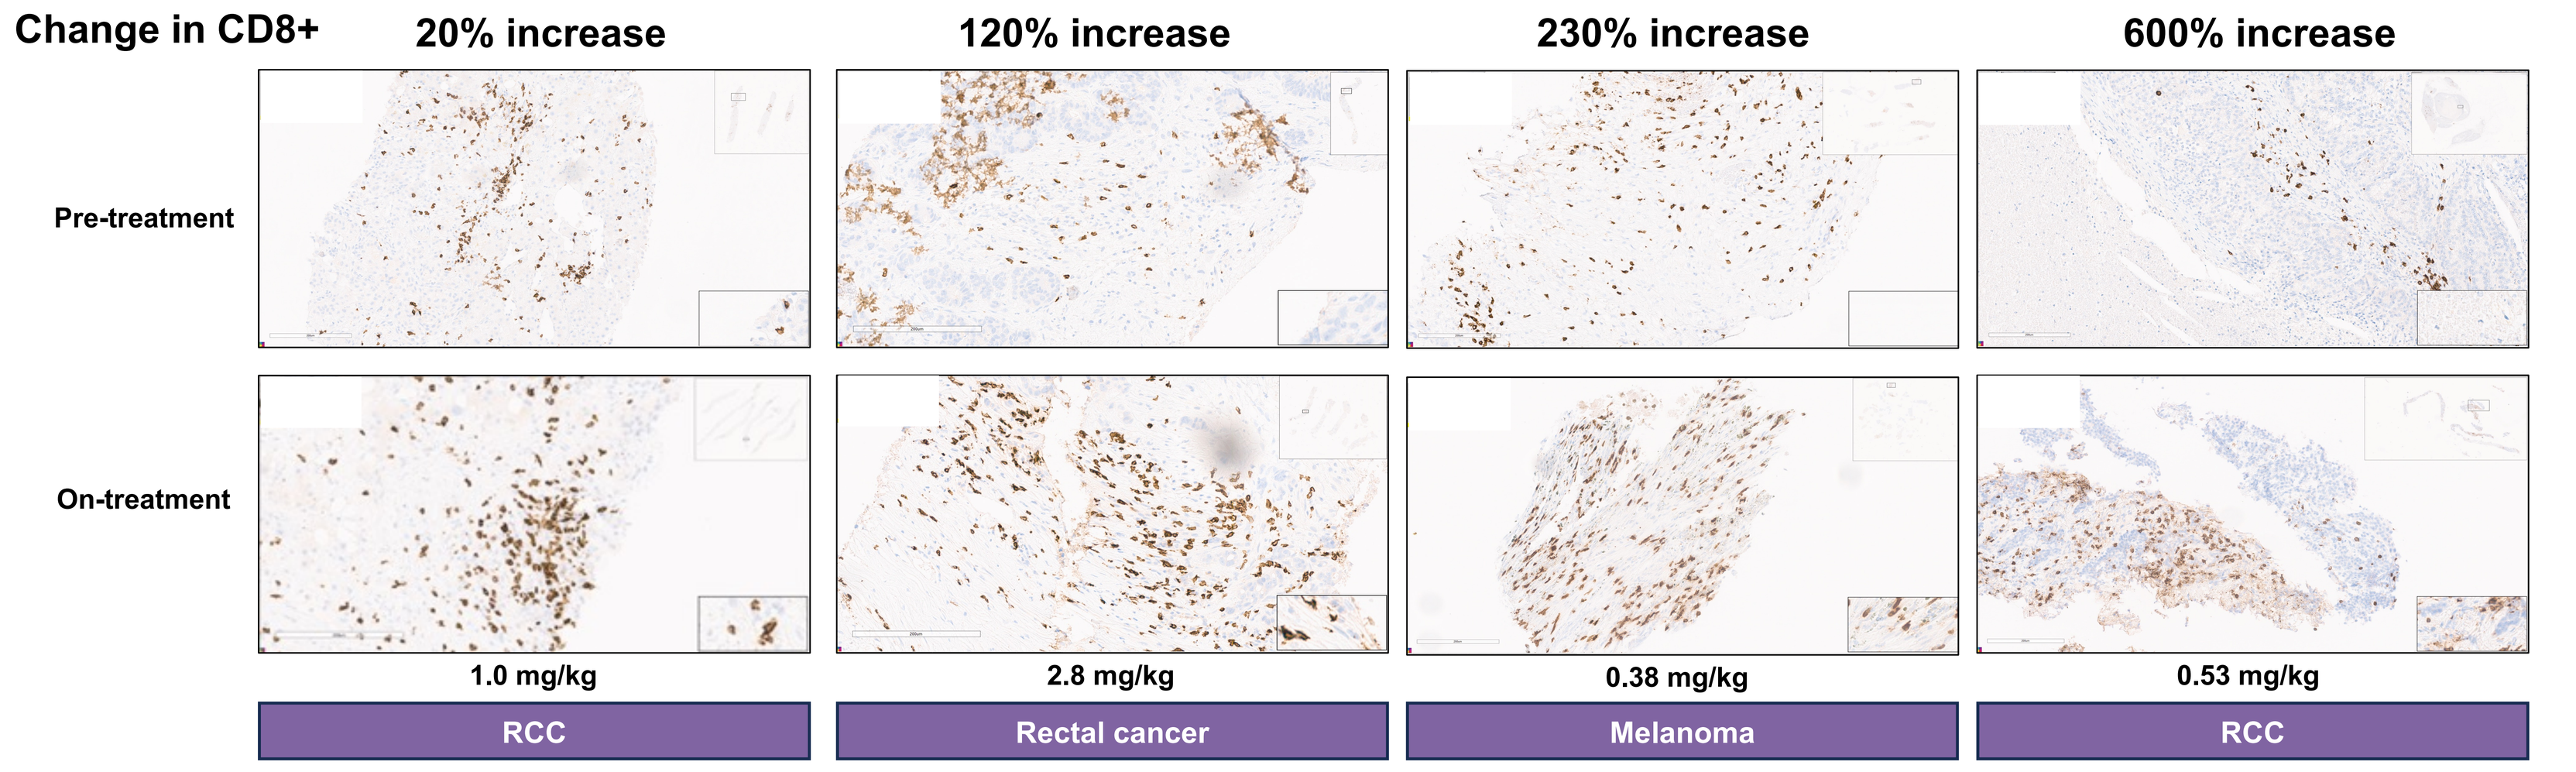
Change in CD8+
20% increase
120% increase
230% increase
600% increase
Pre-treatment
On-treatment
1.0 mg/kg
2.8 mg/kg
0.38 mg/kg
0.53 mg/kg
RCC
Rectal cancer
Melanoma
RCC

Immunohistochemistry for CD8 was performed on available pre- and on-treatment (cycle 2 day 2-21) tumor biopsies. The percentage of tumor-infiltrating immune cells was scored by a certified pathologist by assessing the percent positivity in the intra-tumoral stroma as a fraction of all immune cells in the intra-tumoral stroma. Change in CD8+ cells in tumor takes into account changes in intra-tumoral immune cells as assessed by H&E stain and CD8+ IHC.

Conclusions for XTX202, a Tumor-Activated Engineered IL-2βγ

- **The patient population enrolled in the Phase 1/2 trial for XTX202 comprised older, heavily pre-treated patients with a range of advanced solid tumors and lower performance status in contrast to the younger RCC and melanoma patient population with good performance status that was included in pivotal aldesleukin trials**
- **XTX202 was administered with doses up to 4 mg/kg Q3W as an outpatient regimen and was generally well-tolerated with TRAEs primarily Grade 1-2, supporting effective masking of XTX202:**
 - **No grade 5 TRAEs and no signs or symptoms of VLS were observed through 4.0 mg/kg dose level**
 - **Higher grade TRAEs were generally asymptomatic laboratory abnormalities**
 - **There were no treatment discontinuations due to TRAEs**
- **Evidence of tumor-selective XTX202 activation resulting in tumor concentrations of the active molecule in the range capable of T cell and NK cell stimulation was observed at 2.8 mg/kg dose level**
 - **Suggests 2.8 mg/kg or higher monotherapy doses are approaching optimal range to activate CD8+ effector T cells and NK cells in the tumor**
- **Peripheral PK data support Q3W dosing schedule and demonstrated limited XTX202 activation in peripheral circulation**
- **Tumor-selective increases in CD8+ effector T cells were observed in patient tumor samples following XTX202 treatment**
- **Dose-dependent increase in CD8+ T and NK cells observed in response to XTX202 treatment, consistent with IL-2βγ biology**
- **Data suggest dose dependent increase in DCR, with 50% DCR at doses ≥ 2.8 mg/kg**
- **The totality of preliminary data supports plans to evaluate XTX202 in Phase 2 study in patients with RCC and melanoma at a higher dose level of 4.0 mg/kg Q3W**
- **XTX202 was generally well-tolerated with repeated administration (including >1 year) and favorable profile for combination therapy**

Data cutoff date: October 26, 2023

Abbreviations: AE: adverse event; ALT: alanine aminotransferase; AST: aspartate aminotransferase; BOR: best overall response; Ca: cancer; CR: complete response; DCR: disease control rate; DL: dose level; DLT: dose limiting toxicity; ECOG PS: ECOG Performance Status; EOT: end of treatment; IHC: immunohistochemistry; IO: immuno-oncology; MMP: matrix metalloprotease; MSS CRC: microsatellite stable colorectal cancer; MTD: maximum tolerated dose; NK: natural killer; NSCLC: non-small cell lung cancer; ORR: objective response rate; PK: pharmacokinetic; PR: partial response; Q3W: every 3 weeks; RCC: renal cell carcinoma; rhIL-2: recombinant human interleukin 2; SD: stable disease; TIL: tumor-infiltrating lymphocyte; TRAE: treatment related adverse event; Treg: regulatory T cells; VLS: vascular leak syndrome.

References: 1. Journal of Clinical Oncology, Vol 13, No 3 (March), 1995: pp 688-696; 2. Journal of Clinical Oncology, Vol 17, No 7 (July), 1999: pp 2105-2116.

Contact information: medicalaffairs@xiliotx.com