Phase 1/2 Study of Vilastobart (formerly XTX101), a Tumor-Activated, Fc-enhanced Anti-CTLA-4 Monoclonal Antibody, in Combination with Atezolizumab in Patients with Advanced Solid Tumors

1455

Diwakar Davar[1], Ryan J. Sullivan[2], John George Knecht[3], Andrae Lavon Vandross[4], Cesar Augusto Perez[5], Alberto Bessudo[6], Anurag Gupta[7], Ekta Patel[7], Damiano Fantini[7], Sattanathan Paramasivan[7], David Crowe[7], Meghan Duncan[7], Scott McConnell[7], Katarina Luptakova[7], Aparna Parikh[2]

(1) UPMC Hillman Cancer Center, Pittsburgh, PA; (2) Massachusetts General Hospital, Boston, MA (3) Clear Lake Oncology, Webster, TX; (4) NEXT Oncology, Austin, TX; (5) Sarah Cannon Research Institute at Florida Cancer Specialists - Lake Nona, Orlando, FL; (6) cCARE, Encinitas, CA; (7) Xilio Therapeutics, Inc., Waltham, MA.

BACKGROUND

Vilastobart (XTX101): Tumor-Activated, High Affinity Binding, Fc-Enhanced Anti-CTLA-4

- Vilastobart is an investigational tumor-activated, high affinity, Fc-enhanced anti-CTLA4 antibody designed to focus activity towards the tumor and minimize systemic adverse events
- Vilastobart incorporates protease-cleavable masking peptides designed to block CTLA-4 binding when in the inactive state and dissociate upon activation by tumor-associated proteases
- Vilastobart is designed to block CTLA-4 checkpoint and drive ADCC against regulatory T-cells (Tregs) that highly express CTLA-4
- Vilastobart was 10-fold more potent than ipilimumab in *in vivo* studies[1]
- Fc enhancement augments FcγR co-engagement on antigen presenting cells and has been linked with efficacy of anti-CTLA-4 combinations in patients with "cold tumors," including MSS CRC[2]

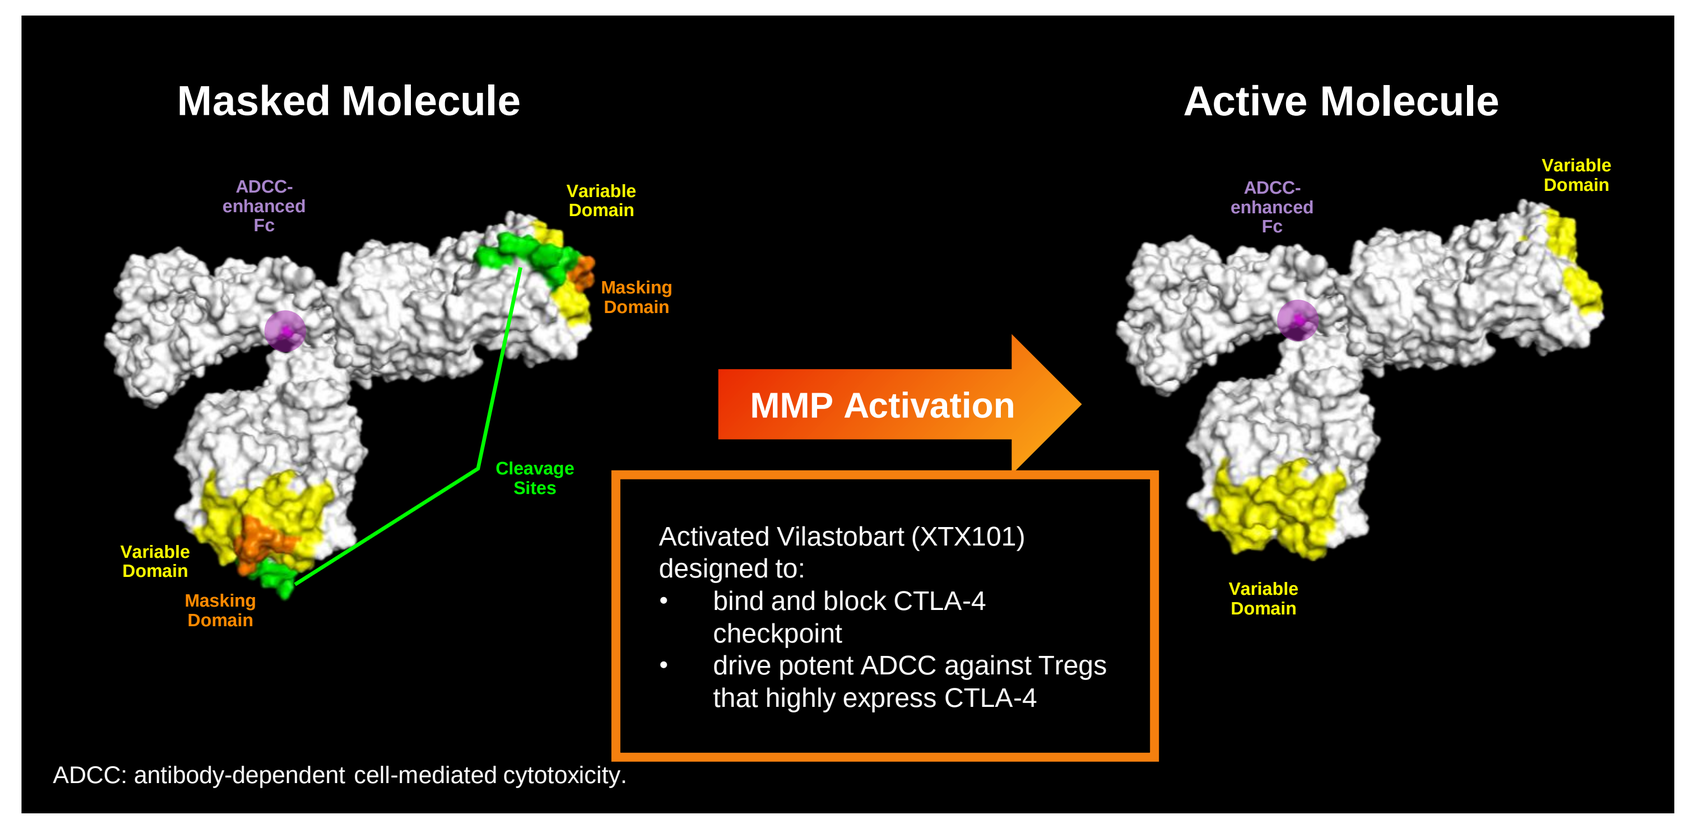
Masked Molecule
Active Molecule
ADCC-enhanced Fc
Variable Domain
Masking Domain
Cleavage Sites
MMP Activation
Activated Vilastobart (XTX101) designed to:
bind and block CTLA-4 checkpoint
drive potent ADCC against Tregs that highly express CTLA-4
ADCC: antibody-dependent cell-mediated cytotoxicity.

Study Design for First-In-Human Clinical Trial of Vilastobart (XTX101) in Advanced Solid Tumors

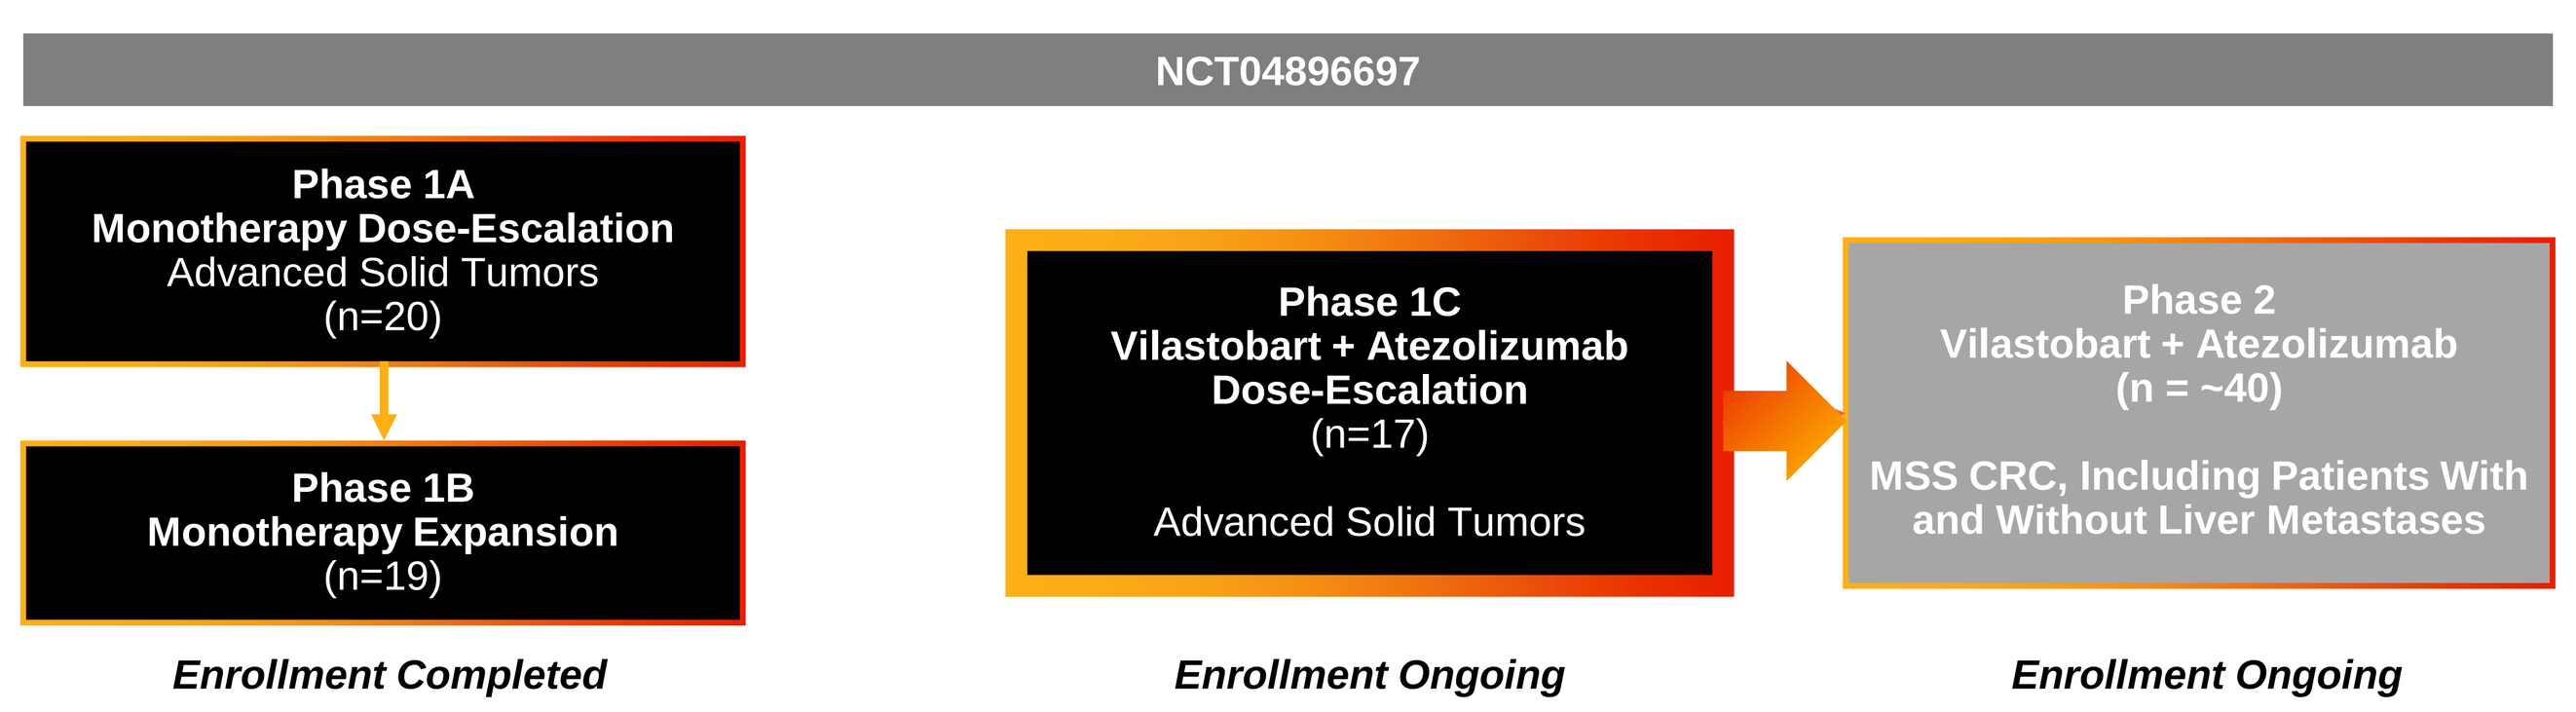
NCT04896697
Phase 1A
Monotherapy Dose-Escalation
Advanced Solid Tumors
(n=20)
Phase 1B
Monotherapy Expansion
(n=19)
Enrollment Completed
Phase 1C
Vilastobart + Atezolizumab
Dose-Escalation
(n=17)
Advanced Solid Tumors
Enrollment Ongoing
Phase 2
Vilastobart + Atezolizumab
(n = ~40)
MSS CRC, Including Patients With and Without Liver Metastases
Enrollment Ongoing

Vilastobart (XTX101) at the Monotherapy RP2D (150 mg Q6W) Was Generally Well-Tolerated and Demonstrated Evidence of Anti-Tumor Activity[3]

- Phase 1A and Phase 1B evaluated vilastobart monotherapy. As of October 7, 2024, 39 patients with median age 62 (43-80) and a variety of advanced solid tumors with median of 4 prior lines of therapy (1-12) were enrolled at dose levels ranging from 7 to 180 mg Q3W or 150 mg Q6W. Most common tumor types included CRC (n=12), NSCLC (n=5), melanoma (n=3), breast cancer (n=3) and pancreatic cancer (n=3).
- Evidence of tumor activation of vilastobart was observed (73-96% activation in n=2 on-treatment biopsies, including a biopsy from MSS CRC liver metastasis), compared to limited peripheral activation (~13%).
- Adjusted for 10-fold higher potency based on preclinical data, vilastobart dosing at 150 mg Q6W demonstrated peripheral exposures of activated molecule in concentrations estimated to be similar to 1.3 (AUC) or 2.7 mg/kg of ipilimumab (Cmax), and 73-96% activated molecule in the tumor estimated to lead to exposure comparable to approximately 15-20 mg/kg ipilimumab.
- 150 mg Q6W was established as the recommended Phase 2 dose (RP2D) for vilastobart monotherapy, and a total of 21 patients were treated with vilastobart monotherapy at this dose level.[3]

AE Category / Term *All TRAEs with ≥10% incidence in any category or any Grade 3 TRAE*	Patients at vilastobart 150mg Q6W (monotherapy RP2D, n = 21) Any	Grade 3
Diarrhea	3 (14%)	2 (10%)
Colitis	3 (14%)	2 (10%)
Infusion related reaction	2 (10%)	1 (5%)
Fatigue	3 (14%)	0
Lymphopenia	1 (5%)	1 (5%)
Dermatitis	1 (5%)	1 (5%)
Blood creatine phosphokinase increased	1 (5%)	1 (5%)
Dose reduction due to TRAE	2	
Treatment discontinuation due to TRAE	2	

- No Grade 4 or 5 TRAEs were reported with vilastobart at the monotherapy RP2D
- No endocrine irAEs and limited skin irAEs
- Colitis (any grade) only in 14% of patients at RP2D
- Safety data includes long-term administration > 1 year

Patients Treated in Phase 1 at Monotherapy RP2D

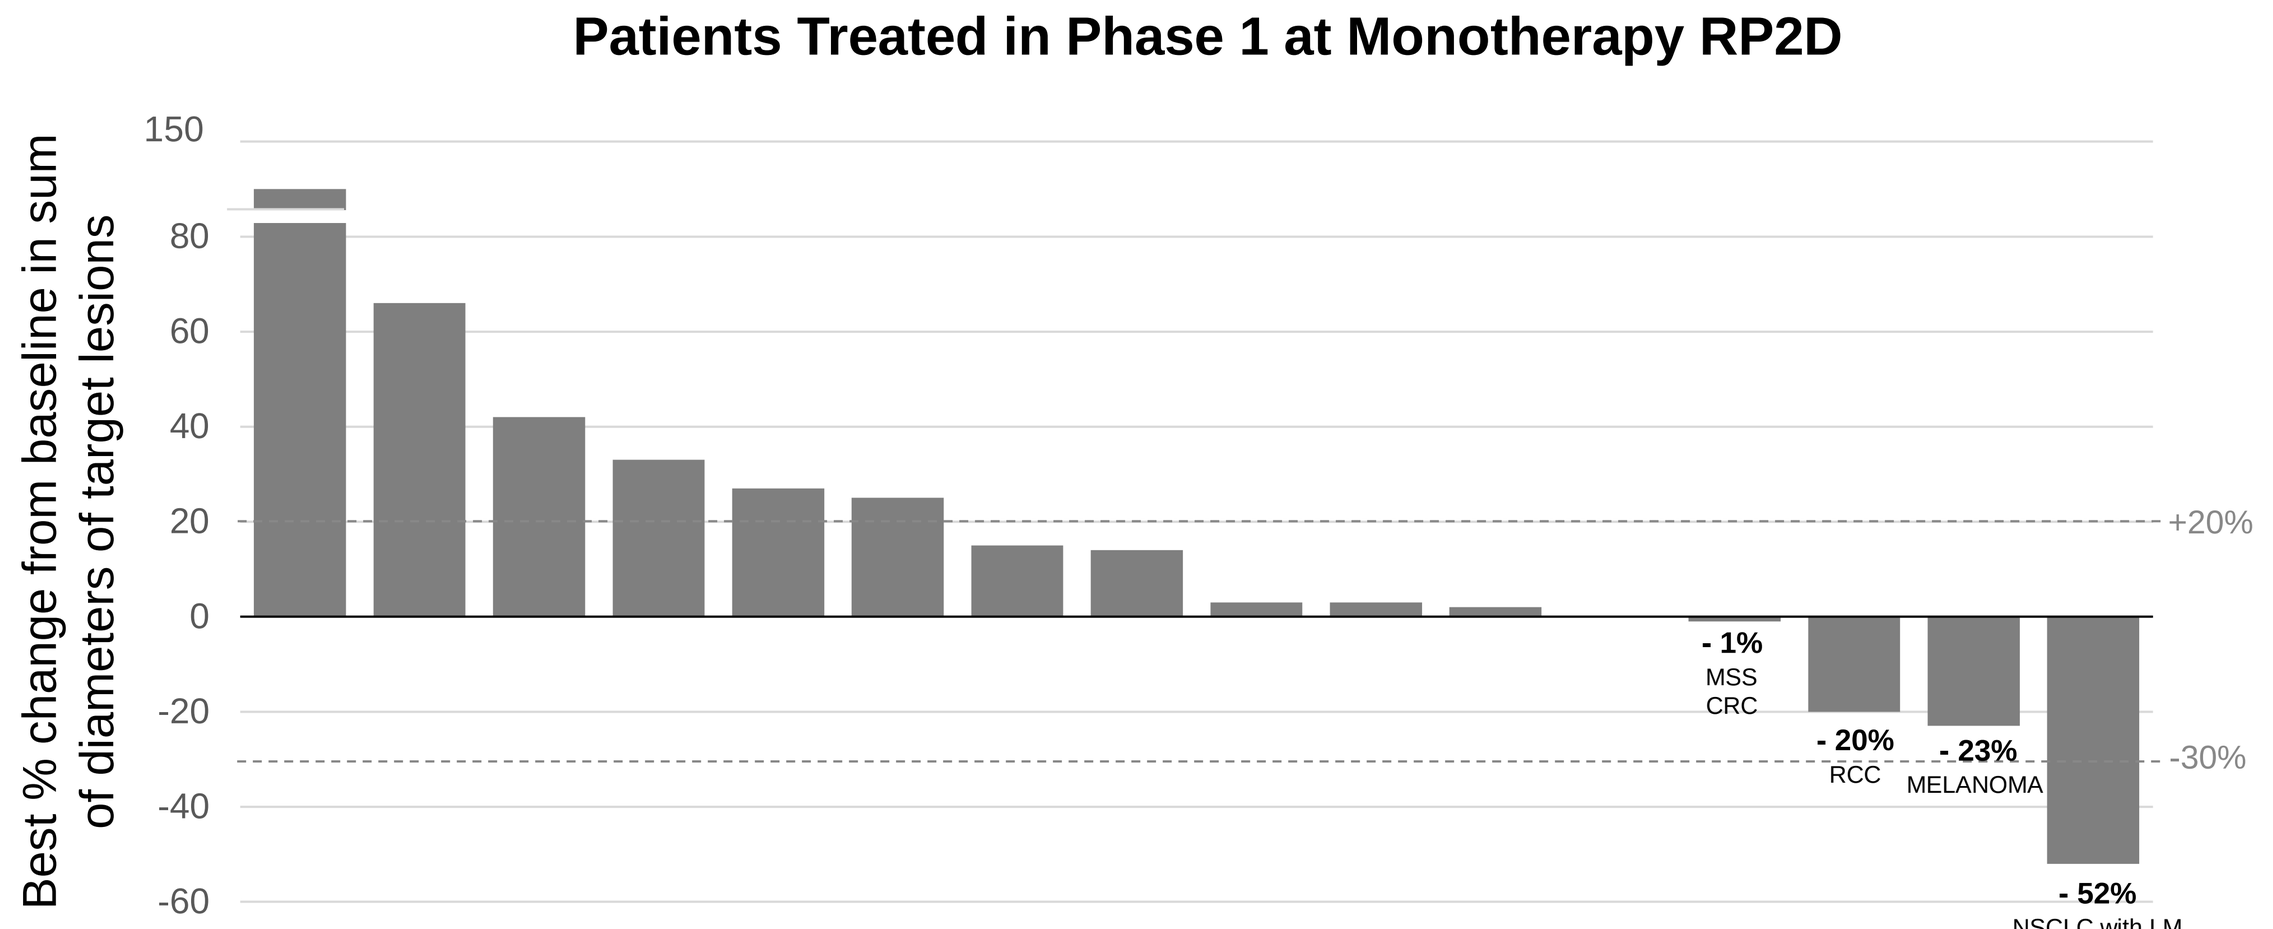
Best % change from baseline in sum of diameters of target lesions
150
80
60
40
20
0
-20
-40
-60
+20%
-30%
- 1%
MSS CRC
- 20%
RCC
- 23%
MELANOMA
- 52%
NSCLC with LM

Deep and durable confirmed PR in PD-L1 negative NSCLC, including resolution of liver metastases (LM)

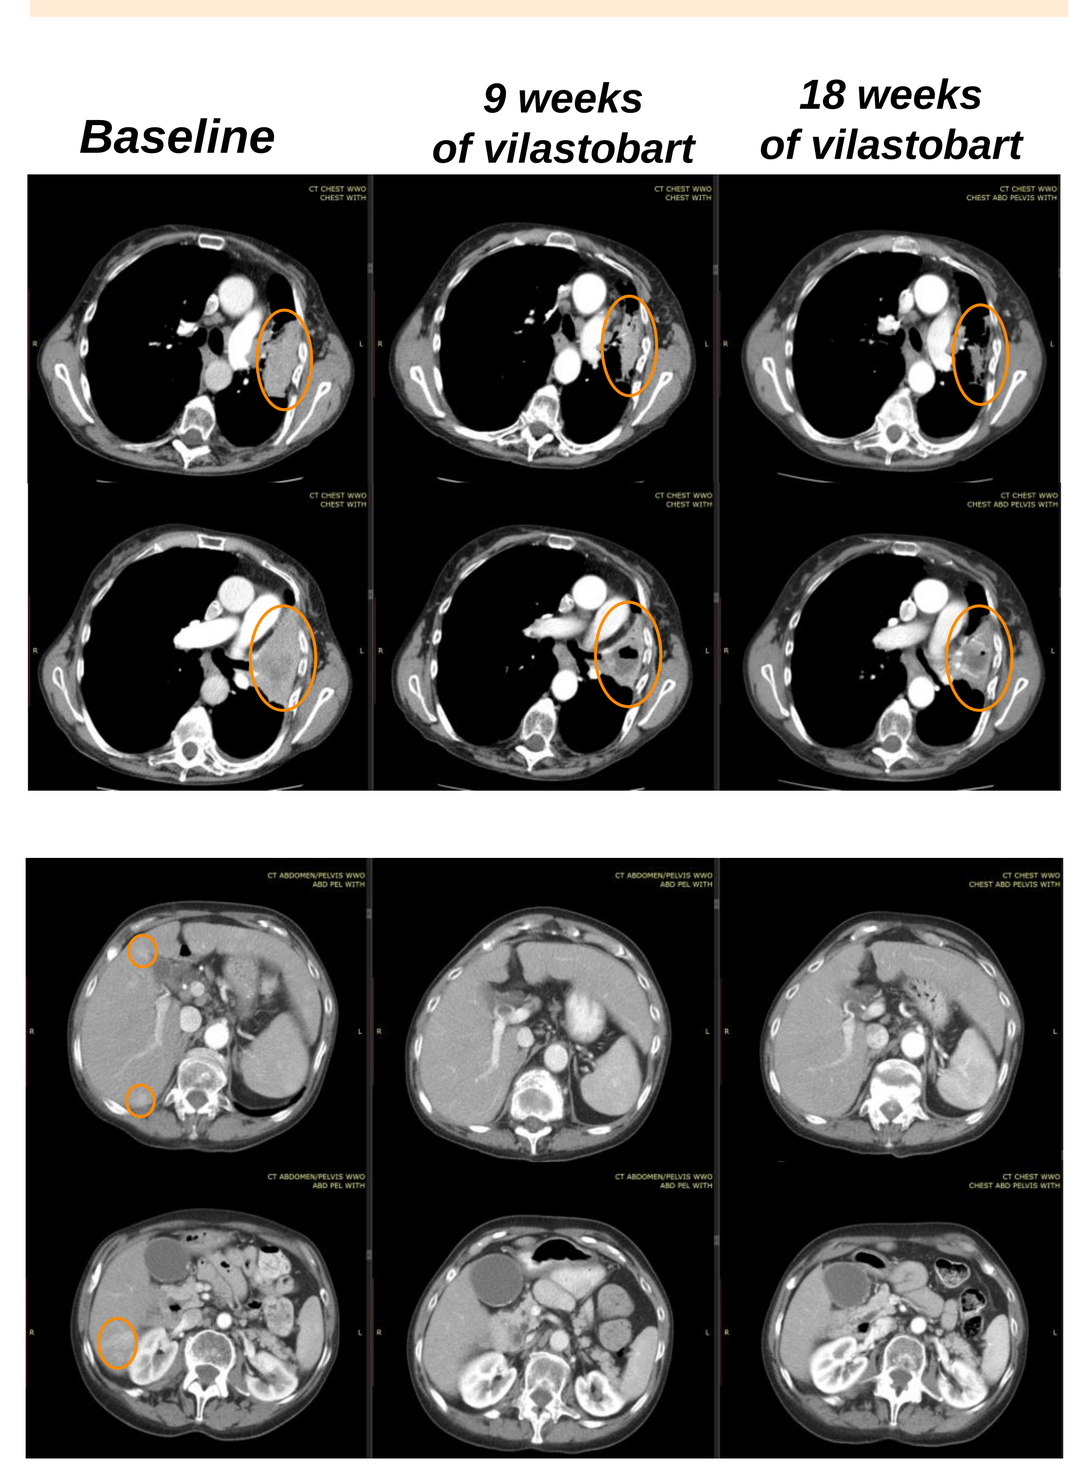
Baseline
9 weeks of vilastobart
18 weeks of vilastobart

Patients Treated in Phase 1 at Monotherapy RP2D

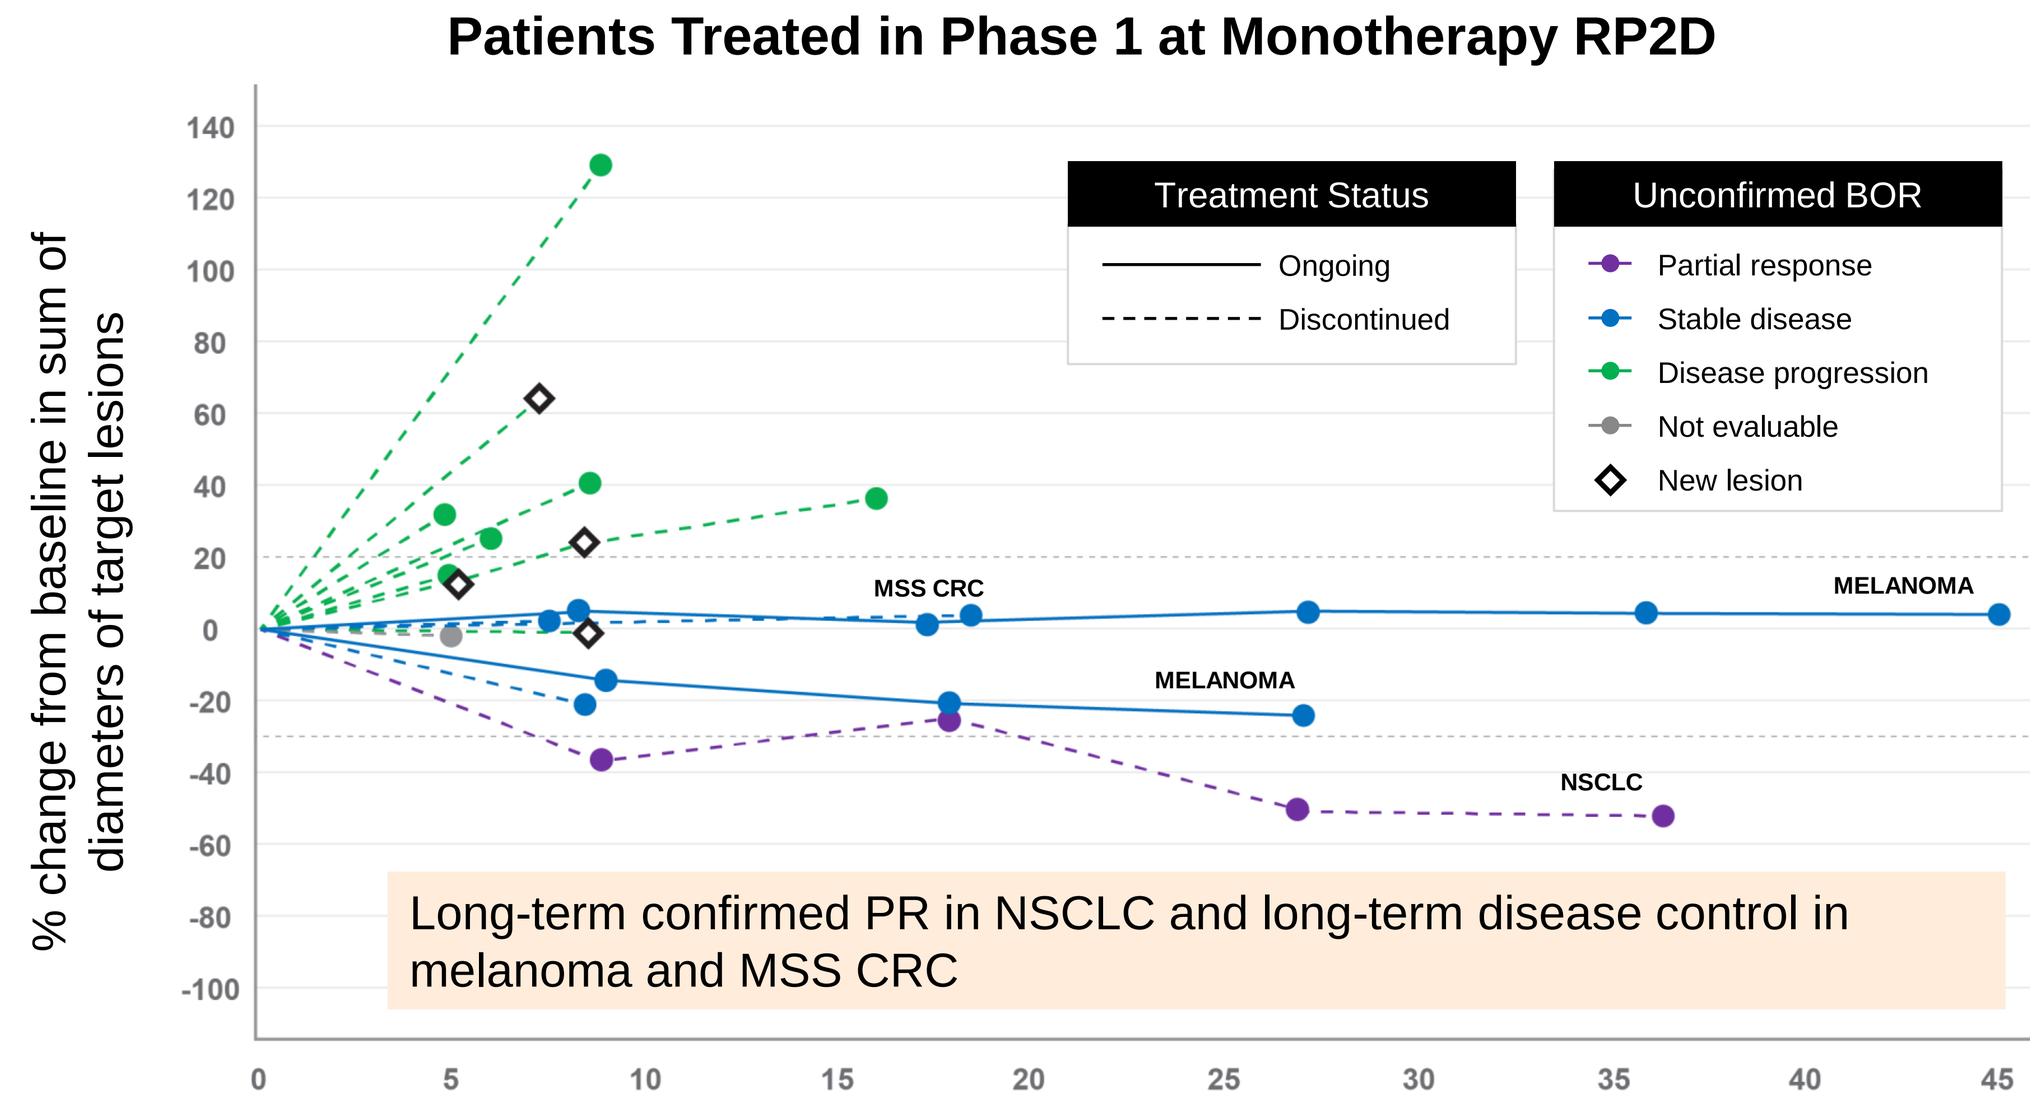
% change from baseline in sum of diameters of target lesions
Treatment Status
Ongoing
Discontinued
Unconfirmed BOR
Partial response
Stable disease
Disease progression
Not evaluable
New lesion
MSS CRC
MELANOMA
MELANOMA
NSCLC
+20%
-30%
Long-term confirmed PR in NSCLC and long-term disease control in melanoma and MSS CRC
Weeks from First Dose
Data cutoff date: Oct 7, 2024.
n=17 response-evaluable patients

RESULTS OF PHASE 1C

Combination Dose-Escalation for Vilastobart (XTX101) + Atezolizumab

Phase 1C Enrolled Heavily Pre-Treated Patients with Cold Tumors

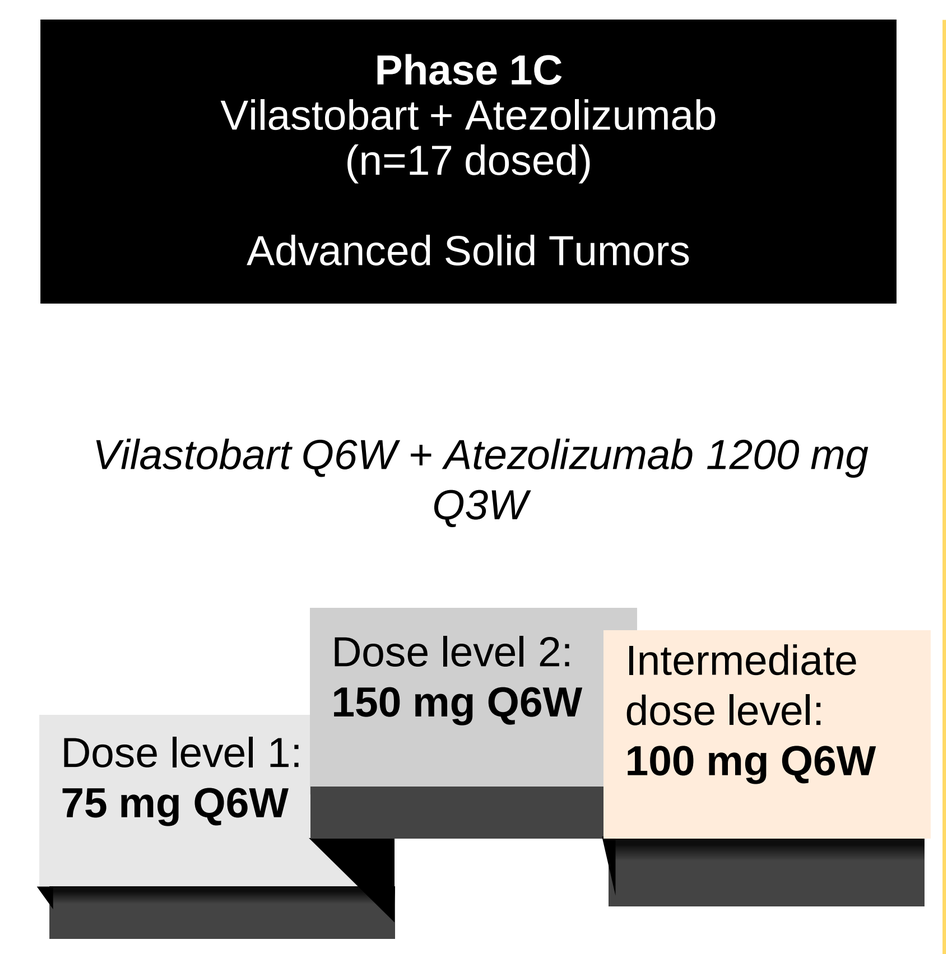
Phase 1C
Vilastobart + Atezolizumab
(n=17 dosed)
Advanced Solid Tumors
Vilastobart Q6W + Atezolizumab 1200 mg Q3W
Dose level 1:
75 mg Q6W
Dose level 2:
150 mg Q6W
Intermediate dose level:
100 mg Q6W

Patient Characteristics	Total (N=17)
Demographics	
Age, median (range)	69 (39, 77)
Female	6 (35%)
ECOG PS 0	7 (41%)
ECOG PS 1	10 (59%)
Prior Lines of Anti-Cancer Treatment	**Median 3 (1-12)**
1	2 (12%)
2	1 (6%)
3	6 (35%)
4	1 (6%)
5	3 (18%)
6 and more	4 (24%)
Progressed on Prior Treatment with IO	
≥1	4 (24%)

83% of patients had 3 or more prior lines of treatment

Tumor Types	Total (N=17)
MSS CRC	12
MSI-H CRC	1
NSCLC	1
Esophageal cancer	1
Ampullary carcinoma	1
Abdomen	1

Treatment Status	Total (N=17)
Continuing on Treatment	**7**
Discontinued Treatment	**10**
Progressive Disease	1
Adverse Events	2
Consent Withdrawal	4
Death	0
Investigator Decision	3

Patients Receiving the Combination of Vilastobart (XTX101) and Atezolizumab Experienced Minimal irAEs

AE Category / Term *All TRAEs with ≥10% incidence in any category or any Grade 3 TRAE*	All Phase 1C Patients (n = 17) Any	Grade 3
ALT increased	3 (18%)	2 (12%)
Blood ALP increased	2 (12%)	1 (6%)
Diarrhea	2 (12%)	1 (6%)
Colitis	1 (6%)	1 (6%)
Infusion related reactions (1)	10 (59%)	0
AST increased	3 (18%)	0
Lipase increased	3 (18%)	0
Fatigue	2 (12%)	0
Dose reduction due to AE	1	
Treatment discontinuation due to TRAE (2)	1	

1. Of the 10 patients with infusion related reactions, 4 experienced reactions related to vilastobart, 3 experienced reactions related to atezolizumab and 3 experienced reactions related to the combination.
2. Reflects discontinuation of both vilastobart and atezolizumab

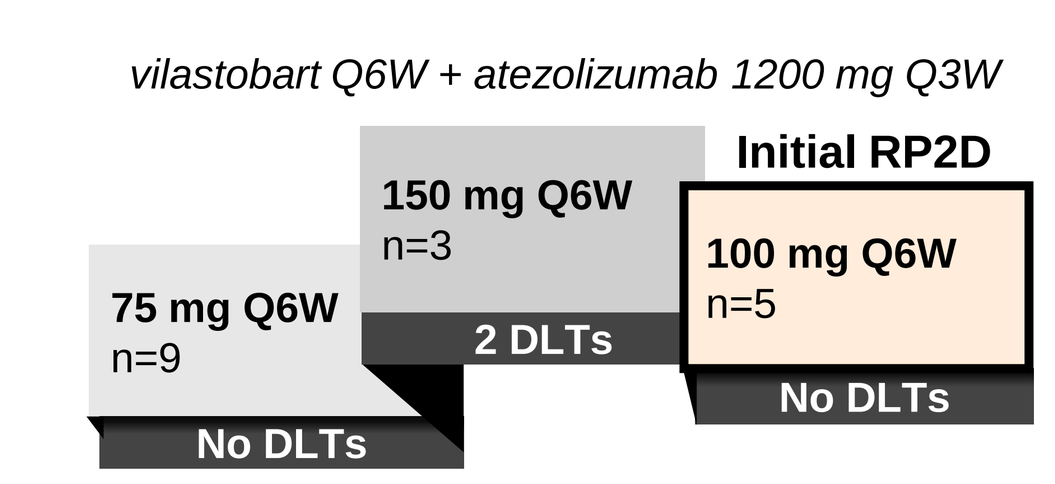
vilastobart Q6W + atezolizumab 1200 mg Q3W
Initial RP2D
75 mg Q6W
n=9
No DLTs
150 mg Q6W
n=3
2 DLTs
100 mg Q6W
n=5
No DLTs

- No Grade 4 or Grade 5 TRAEs at any dose level
- Only 3 patients experienced Grade 3 TRAEs, of these 2 experienced DLTs (both at 150 mg dose level):
 - One patient with Grade 3 colitis/diarrhea
 - One patient with Grade 3 ALT and ALP elevation
- Vilastobart 100 mg Q6W in combination with atezolizumab 1200 mg Q3W was established as initial RP2D

Combination of Vilastobart (XTX101) and Atezolizumab Demonstrated Anti-Tumor Activity in Cold Tumors with Sustained Tumor Reduction in a MSS CRC Patient with Liver Metastasis

Patients Treated with the Combination of Vilastobart and Atezolizumab in Phase 1C

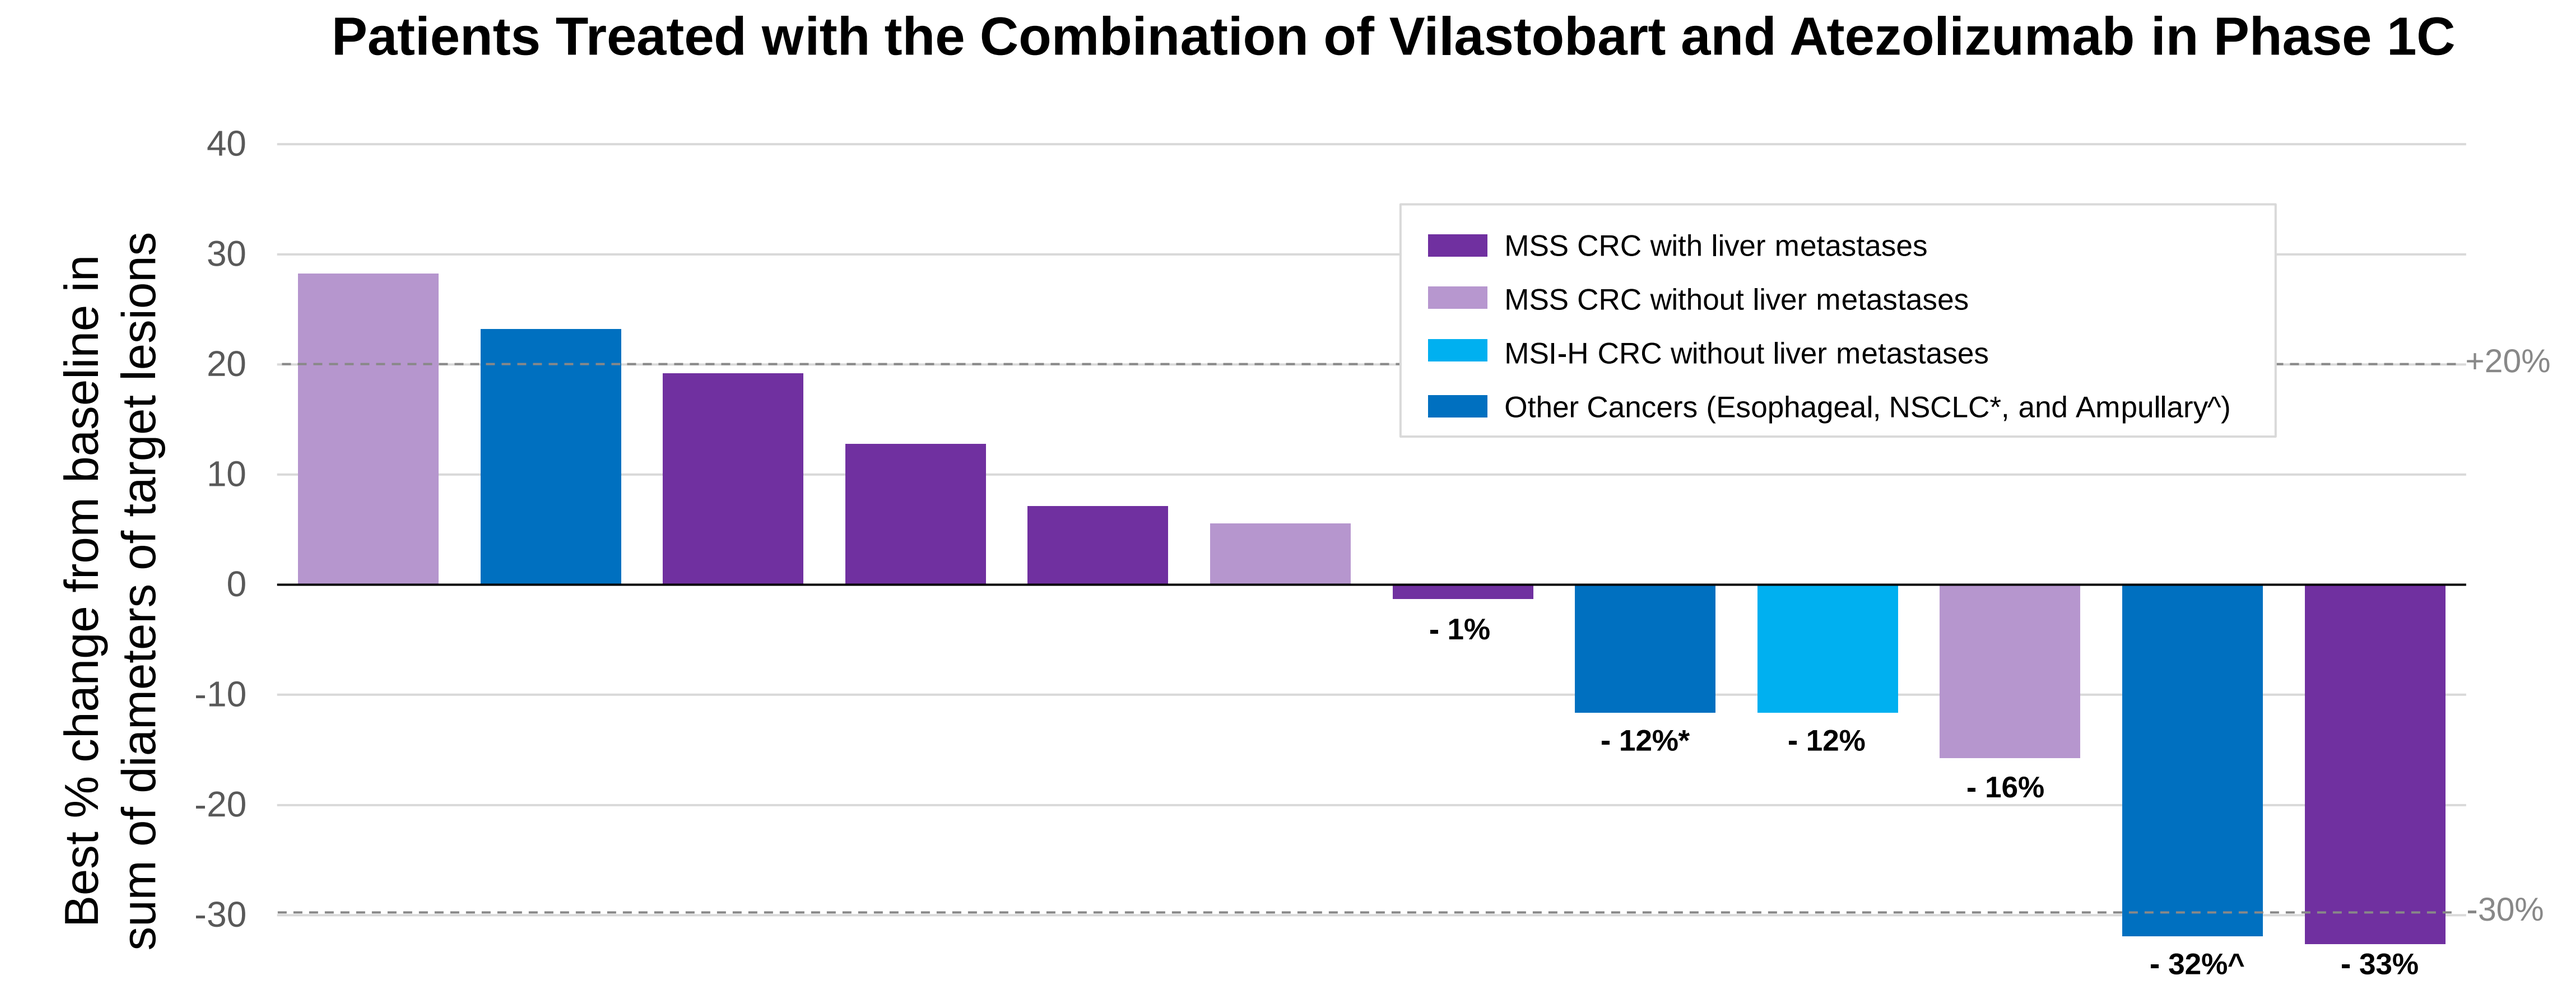
Best % change from baseline in sum of diameters of target lesions
MSS CRC with liver metastases
MSS CRC without liver metastases
MSI-H CRC without liver metastases
Other Cancers (Esophageal, NSCLC*, and Ampullary^)
+20%
-30%
- 1%
- 12%*
- 12%
- 16%
- 32%^
- 33%

Patients Treated with the Combination of Vilastobart and Atezolizumab in Phase 1C

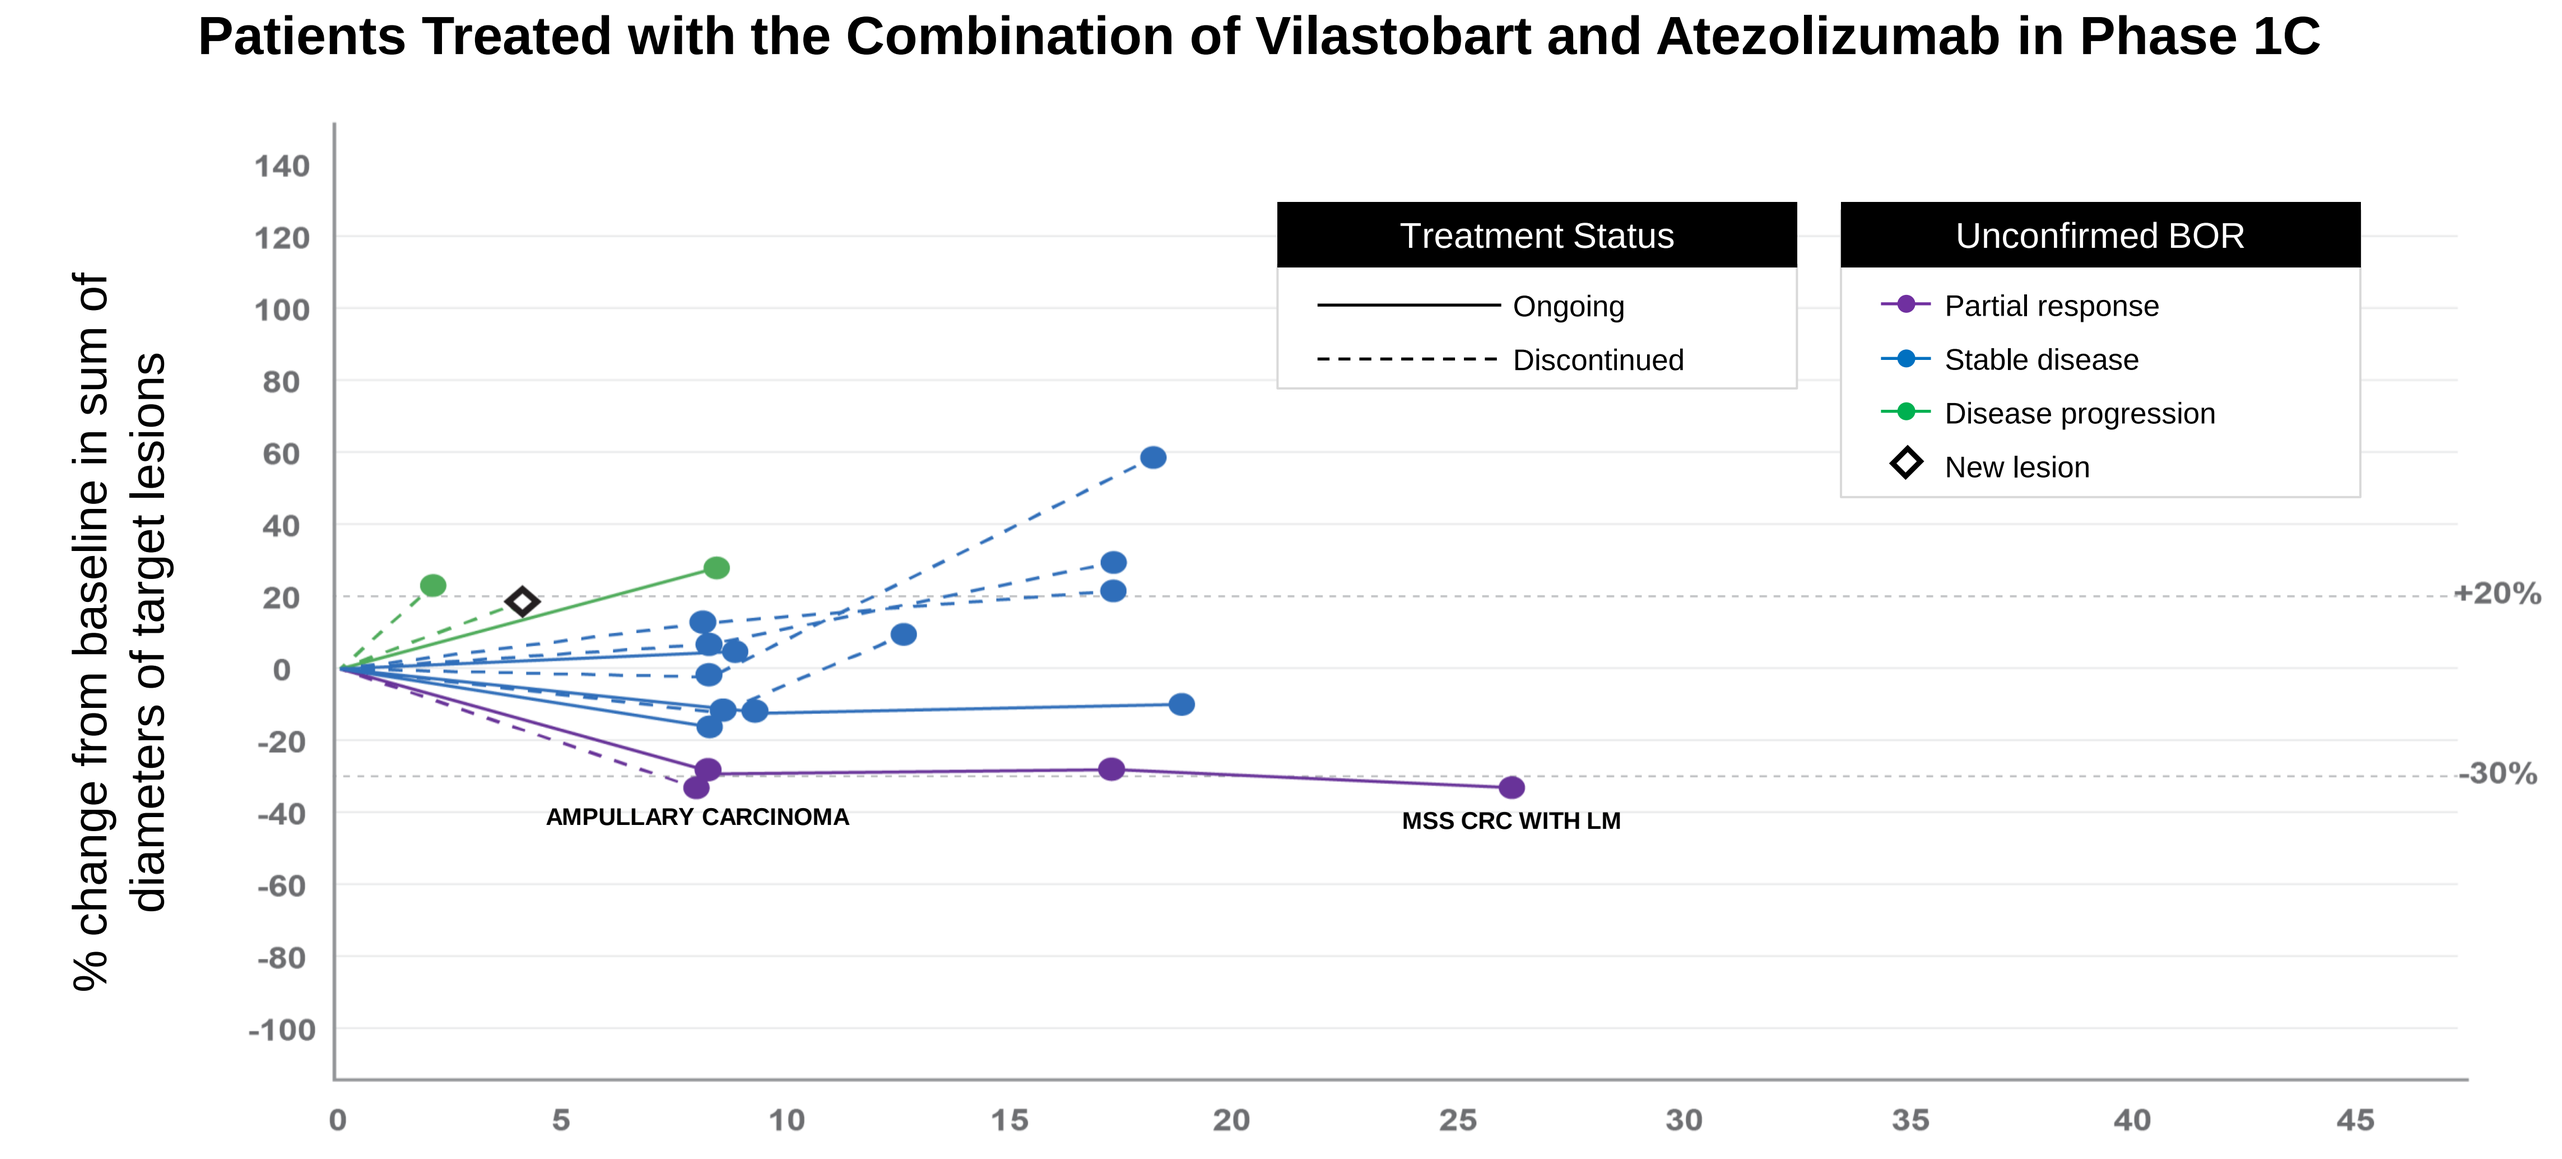
% change from baseline in sum of diameters of target lesions
Treatment Status
Ongoing
Discontinued
Unconfirmed BOR
Partial response
Stable disease
Disease progression
New lesion
+20%
-30%
AMPULLARY CARCINOMA
MSS CRC WITH LM
Weeks from First Dose

Data cutoff date: Oct 7, 2024.
*n=12 response-evaluable patients

Partial Responses Observed in 2 Patients with Cold Tumors, Including Resolution of Liver Metastasis in Patient with MSS CRC

76M with Malignant Neoplasm of Ampulla of Vater

- Diagnosed in January 2022
- 2 prior lines of therapy:
 - Gemcitabine + nab-Paclitaxel
 - 5FU + Irinotecan Liposome + Leucovorin
- Started vilastobart (150mg) + atezolizumab in March 2024
- PR (unconfirmed)* accompanied by a significant Ca19-9 decrease after a single cycle of the study treatment

** Patient withdrew consent prior to undergoing a confirmatory imaging scan*

	Screening		8 weeks after C1D1
Target lesion 1	37.8 mm		30.9 mm
Target lesion 2	11.3 mm		5.0 mm
Target lesion 3	11.4 mm		5.3 mm
Sum of diameters	60.5 mm		41.2 mm
Change			**- 32%**
Serum tumor marker	**Screening**	**C1D1**	**C3D1**
Ca 19-9 (U/mL)	**575**	**700.2**	**40.8**

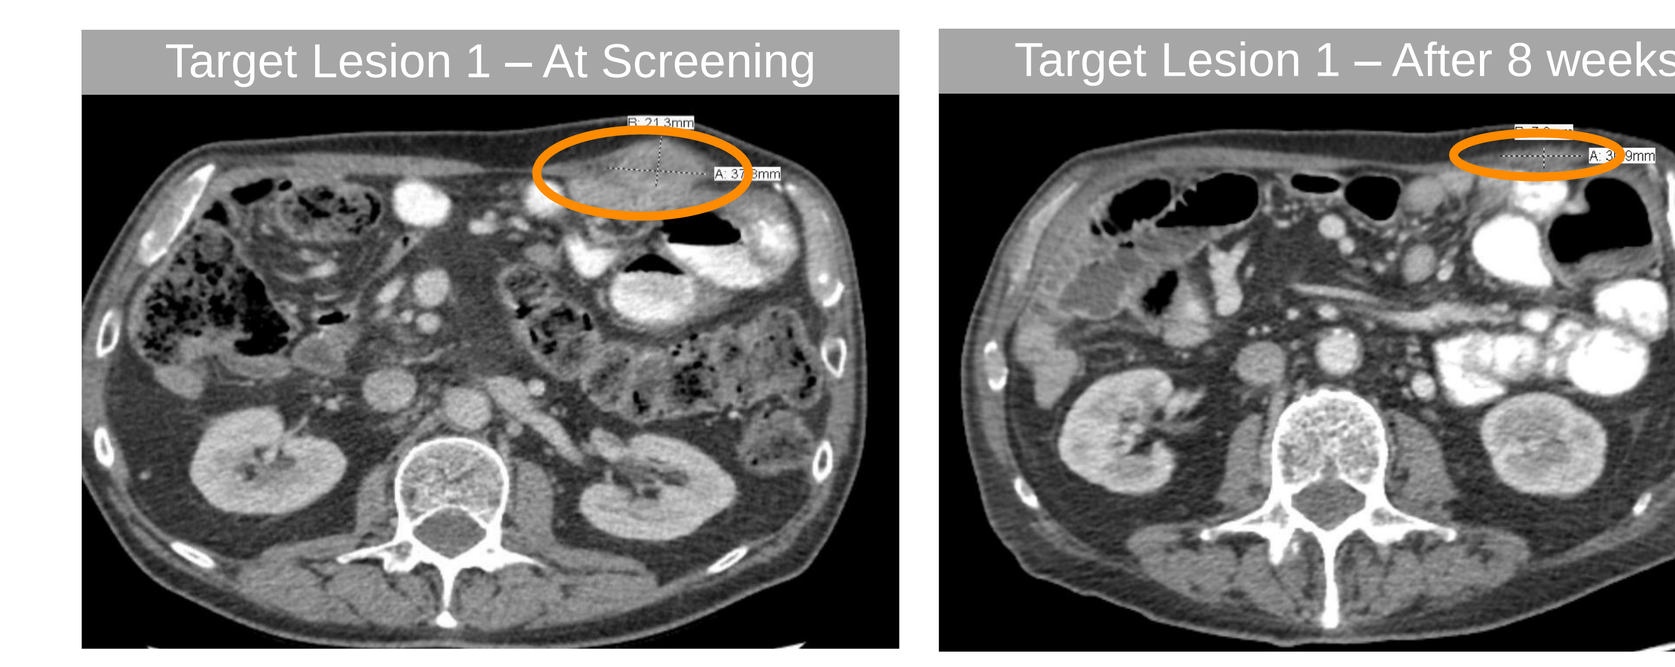
Target Lesion 1 – At Screening
Target Lesion 1 – After 8 weeks

69F with MSS Rectal Cancer and Liver Metastasis

- Diagnosed in August 2018
- 5 prior lines of therapy:
 - FOLFOX-Avastin
 - FOLFIRI-Avastin
 - Cetuximab
 - Lonsurf
 - FOLFIRI-Panitumamab
- Started vilastobart (150mg) + atezolizumab in March 2024
- PR (unconfirmed), awaiting confirmation
- Full resolution of the target lesion in the liver

	Screening	1st follow-up (9 weeks)	2nd follow-up (18 weeks)	3rd follow-up (27 weeks)
Target lesion 1	25.7 mm	23.1 mm	24.2 mm	26.2 mm
Target lesion 2	17.5 mm	17 mm	20.4 mm	20.9 mm
Target lesion 3 (Liver lesion)	15.6 mm	6.8 mm	5.8 mm	resolved
Target lesion 4	24.6 mm	15.1 mm	14.5 mm	13.4 mm
Target lesion 5	15 mm	8.5 mm	6.1 mm	5.8 mm
Sum of diameters	98.4 mm	70.5 mm	71 mm	66.3 mm
Change		**- 28%**	**- 28%**	**- 33%**

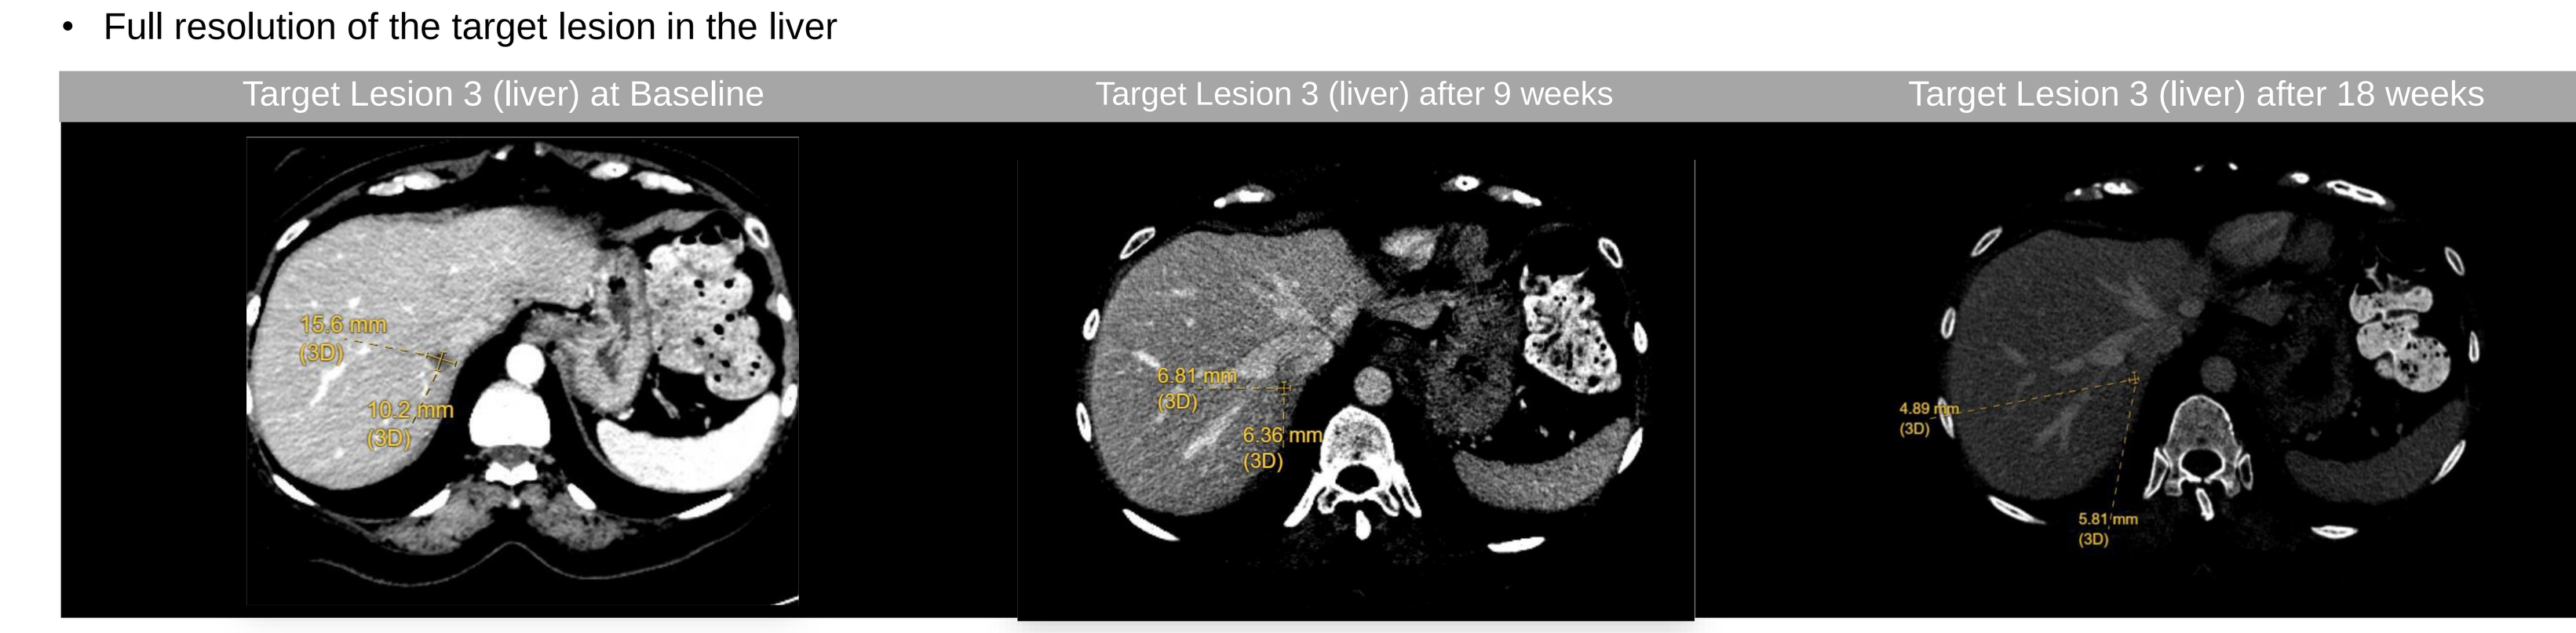
Target Lesion 3 (liver) at Baseline
Target Lesion 3 (liver) after 9 weeks
Target Lesion 3 (liver) after 18 weeks

Vilastobart (XTX101) Monotherapy and Combination With Atezolizumab Showed Dose-Dependent Increases in Ki67+ CD4 and CD8 T Cells, With Minimal Impact on Tregs in Periphery

Changes in immune cells in periphery after vilastobart monotherapy or combination with atezolizumab. Violin plots depicting fold changes from baseline (pre-treatment, day 1) in CD4+ Ki67+ cells (% of CD4 T cells, left panel), CD8+ Ki67+ cells (% of CD8 T cells, mid panel), and Tregs (% of CD4 T cells, right panel). Analysis performed on peripheral blood samples using flow cytometry at day 7±1 post-treatment initiation. Phase 1A/B data shown in grey; vilastobart 7mg (n=2); 20mg (n=1); 60mg (n=7); 150 mg (n=19); 180mg (n=5); Phase 1C data color-coded by dosage: 75 mg vilastobart + atezolizumab (n=9); 100 mg vilastobart + atezolizumab, (n=5); 150 mg vilastobart + atezolizumab (n=3).

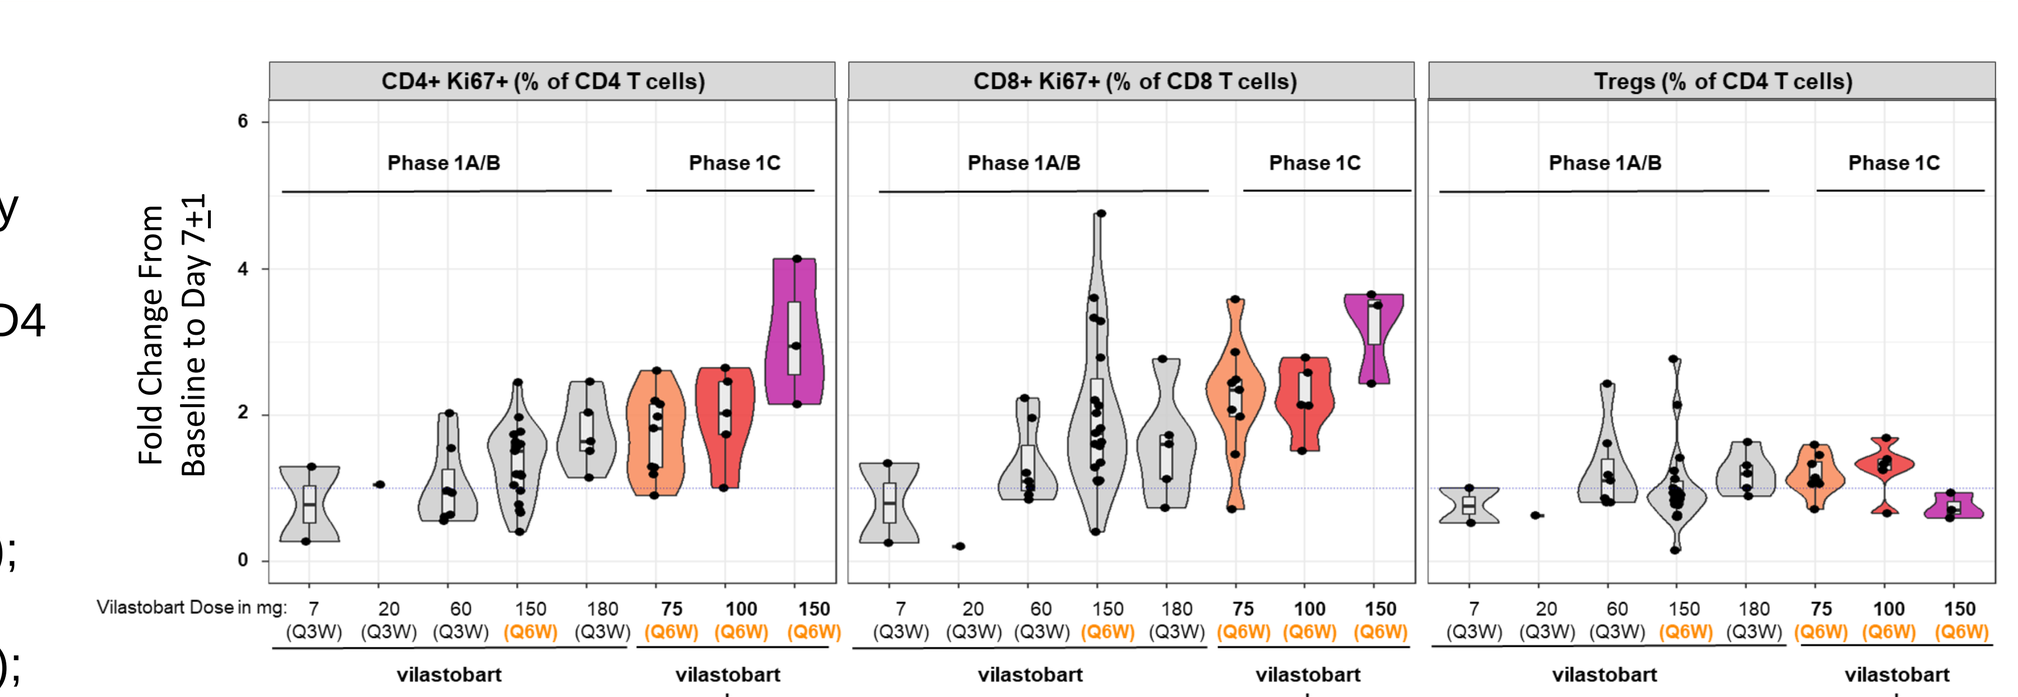
CD4+ Ki67+ (% of CD4 T cells)
CD8+ Ki67+ (% of CD8 T cells)
Tregs (% of CD4 T cells)
Phase 1A/B
Phase 1C
Fold Change From Baseline to Day 7±1
Vilastobart Dose in mg:
vilastobart
vilastobart + atezolizumab

Intratumoral Treg Depletion Observed in a Tumor Biopsy from an MSS CRC Patient, Consistent With the Intended Mechanism of Action of Vilastobart (XTX101)

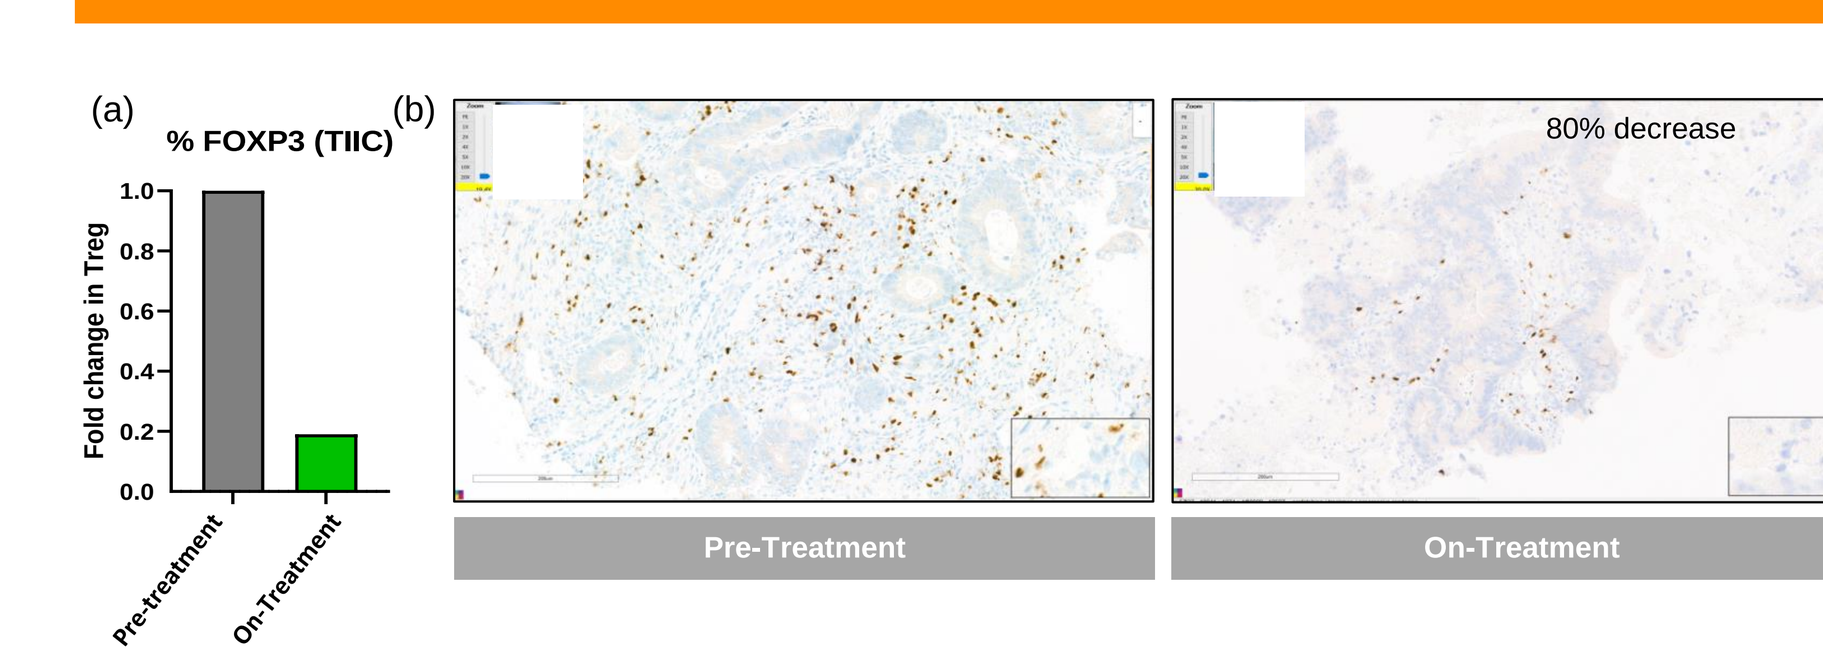
(a)
% FOXP3 (TIIC)
Fold change in Treg
Pre-treatment
On-Treatment
(b)
80% decrease
Pre-Treatment
On-Treatment

**Patient had an optional on-treatment tumor biopsy and was the only patient treated with vilastobart in combination with atezolizumab in Phase 1C for whom a tumor biopsy analysis was available as of October 7, 2024.*

FOXP3 Immunohistochemistry in an MSS CRC patient* before and after vilastobart + atezolizumab combination therapy. (a) Quantification of tumor-infiltrating FOXP3+ cells: Pre-treatment and on-treatment (Cycle 3 Day 19) tumor biopsies were analyzed. The patient received 100 mg vilastobart (Q6W) and 1200 mg atezolizumab (Q3W). The percentage of tumor-infiltrating immune cells was scored by a certified pathologist by assessing the percent positivity in the intra-tumoral stroma as a fraction of all immune cells in the intra-tumoral stroma. Change in FOXP3+ cells in the tumor takes into account the changes in intra-tumoral immune cells as assessed by H&E stain. (b) Representative FOXP3 IHC images from pre and on-treatment biopsies are shown.

CONCLUSIONS

Vilastobart (XTX101) is a novel tumor-activated, high affinity binding, Fc-enhanced anti-CTLA-4 mAb and designed to focus activity towards the tumor and minimize potential for adverse events

- Phase 1 data suggested vilastobart has a differentiated safety profile from systemically active anti-CTLA4, consistent with tumor-selective activation
- In Phase 1C combination dose-escalation, the combination of vilastobart and atezolizumab was generally well-tolerated in patients with advanced solid tumors:
 - No grade 4 or 5 TRAEs at any dose level
 - Across all dose levels, minimal irAEs, with only 1 out of 17 patients (6%) experiencing colitis
 - Dose level of vilastobart 100 mg Q6W in combination with atezolizumab 1200 mg Q3W was declared the initial RP2D
- The combination of vilastobart and atezolizumab demonstrated initial evidence of anti-tumor activity in cold tumors:
 - PR (unconfirmed) in patient with ampullary carcinoma
 - PR (unconfirmed) in patient with MSS CRC with liver metastasis, including resolution of liver metastasis
 - Additional patients with ongoing tumor reduction as stable disease
 - Pharmacodynamic changes consistent with the intended mechanism of action, including intratumoral Treg depletion
- These data supported the initiation of an ongoing Phase 2 study evaluating the combination of vilastobart (100 mg Q6W) and atezolizumab (1200 mg Q3W) in patients with MSS CRC with and without liver metastases

Data cutoff date: October 7, 2024

Abbreviations: ADCC: antibody-dependent cell-mediated cytotoxicity; AE: adverse event; ALP: alkaline phosphatase; ALT: alanine aminotransferase; AUC: area under curve; BOR: best overall response; BA: bio-analytical; CTLA-4: cytotoxic T-Lymphocyte Associated Protein 4; DL: dose level; DLT: dose limiting toxicity; ECOG PS: ECOG Performance Status; G: grade; IO: immuno-oncology; irAE: immune-related adverse event; IV: intra-venous(ly); LM: liver metastases; mAb: monoclonal antibody; MMP: matrix metalloprotease; MSI-H CRC: microsatellite instability-high; MSS CRC: microsatellite stable colorectal cancer; MTD: maximum tolerated dose; NSCLC: non-small cell lung cancer; PDL-1: programmed death receptor ligand 1; PK: pharmacokinetic; PR: partial response; PT: preferred term; Q3W: every 3 weeks; Q6W: every 6 weeks; RCC: renal cell carcinoma; RP2D: recommended phase 2 dose; SD: stable disease; TNBC: triple-negative breast cancer; TRAE: treatment related adverse event: Treg: regulatory T-cell.

References:
1. Jenkins et al., *J Immunother Cancer* 2023 Dec 12;11(12):e007785
2. Bullock et al. ESMO GI 2023
3. Davar et al. ESMO IO 2023

Contact information: medicalaffairs@xiliotx.com